Development and Optimization of a Scalable Affinity Purification of Adeno Associated Virus Serotype 6

Azam Hassaninasab, James Fasano, Jay Cai, Daniel Kennedy, Theresa Dao, Omar Matalka, Christine Beaudry, Miranda Williams, Lorenz Ponce, Arianna Spooner, Emily Sinclair, Mercedes Segura and Bojiao Yin

ElevateBio Basecamp, 200 Smith Street, Waltham MA 02451, US.

INTRODUCTION

The adeno-associated viral vector (AAV) provides an efficient gene therapy platform with several approved products that have marked therapeutic impact for patients. However, potency, cost, and scalability remain as bottlenecks in the manufacture and commercialization of AAV. Among all of these well-known challenges, the enrichment of high-quality AAV particles through the downstream process has been recognized as one of the most impactful. Chromatographic methods show the potential to enable the downstream process with scalability, GMP-friendly and lower cost. However, the chromatography process often requires extensive efforts of optimization to fit the specificity to each serotype. Affinity chromatography (AC) has been used in large-scale bioprocessing for almost 40 years and is considered the preferred method for primary capture in downstream processing of various types of biopharmaceuticals. The key factors for an effective AAV purification process are high overall yields and maximized impurity reduction.

In this study, a full size therapeutic AAV construct (~4.7kb) has been used to establish and further optimize the affinity chromatography process for AAV6 with improved recovery and product quality. After cell lysis and endonuclease treatment from upstream, the crude harvest is clarified and prepared for the capture step. The optimal affinity binding conditions were different for various serotypes. Here we used the design of experiment (DoE) methodology to model load adjustment and percent recovery. As data shows, the recovery of AAV6 in affinity purification step was significantly improved by load adjustment of the material to higher pH and conductivity. Changes in residence times during column loading step did not affect the recovery, however, increase of load volume (CV) was significantly improved the step recovery. Moreover, DoE study of the formulation of elution buffers demonstrated the reduction of aggregation and improvement in product quality with no effect on recovery. Overall, the recovery and vector genome titer of small scale and 2L scale runs were comparable without statistically significant difference, suggesting that the scalability of these optimized chromatography parameters.

AAV UPSTREAM AND DOWNSTREAM PROCESS OVERVIEW

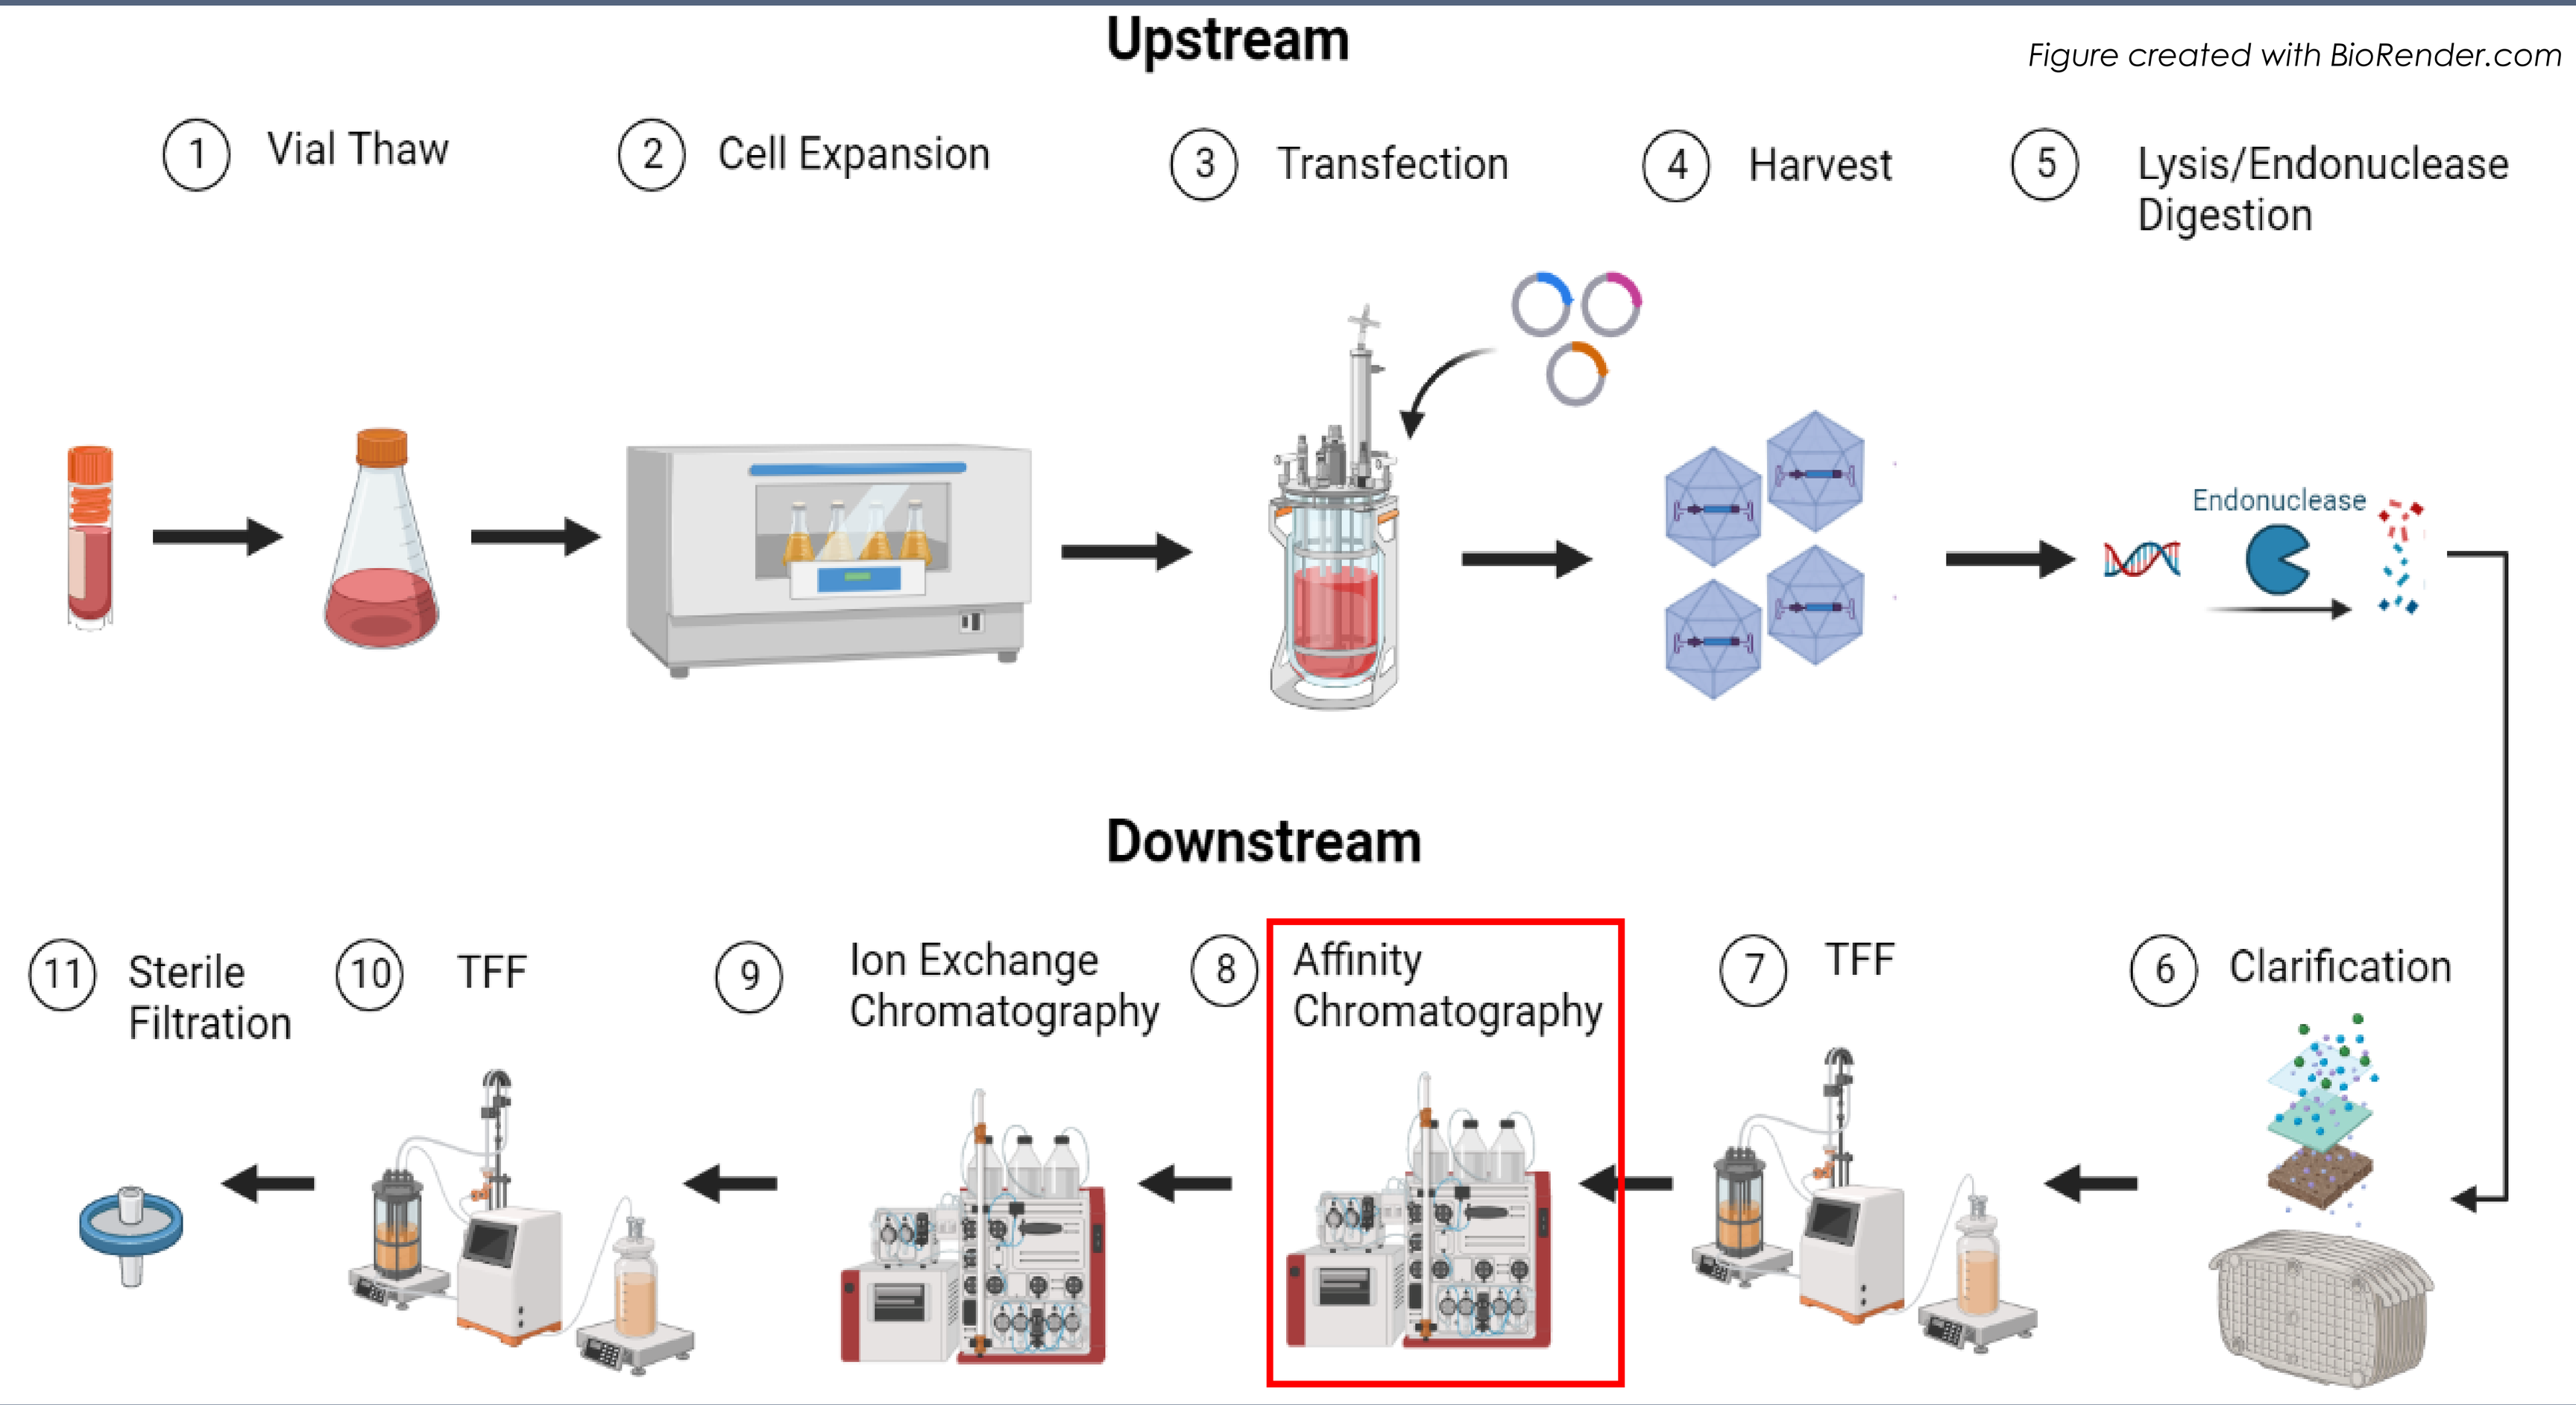
Upstream
Figure created with BioRender.com
1 Vial Thaw
2 Cell Expansion
3 Transfection
4 Harvest
5 Lysis/Endonuclease Digestion
Endonuclease
Downstream
6 Clarification
7 TFF
8 Affinity Chromatography
9 Ion Exchange Chromatography
10 TFF
11 Sterile Filtration

SCHEMATIC MAP of GOI PLASMID

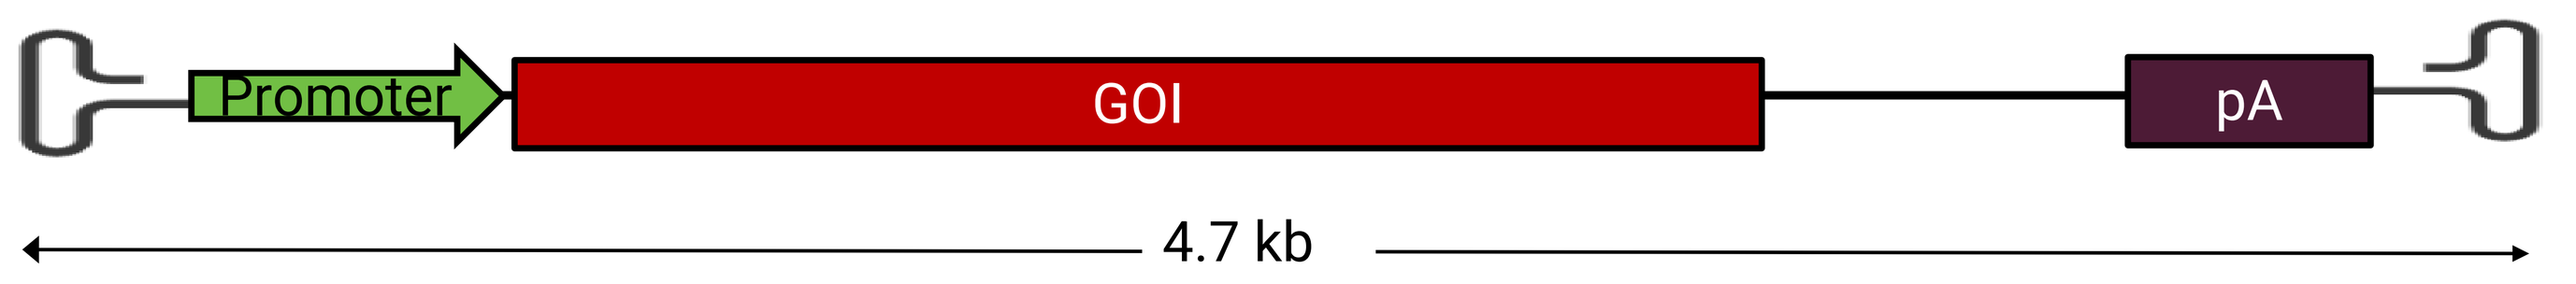
Promoter
GOI
pA
4.7 kb

RESULTS

DoE1: LOAD CONDITIONING

Fig 1A.

DOE Parameters
Salt (150-400 mM)
pH (7.4-8.0)

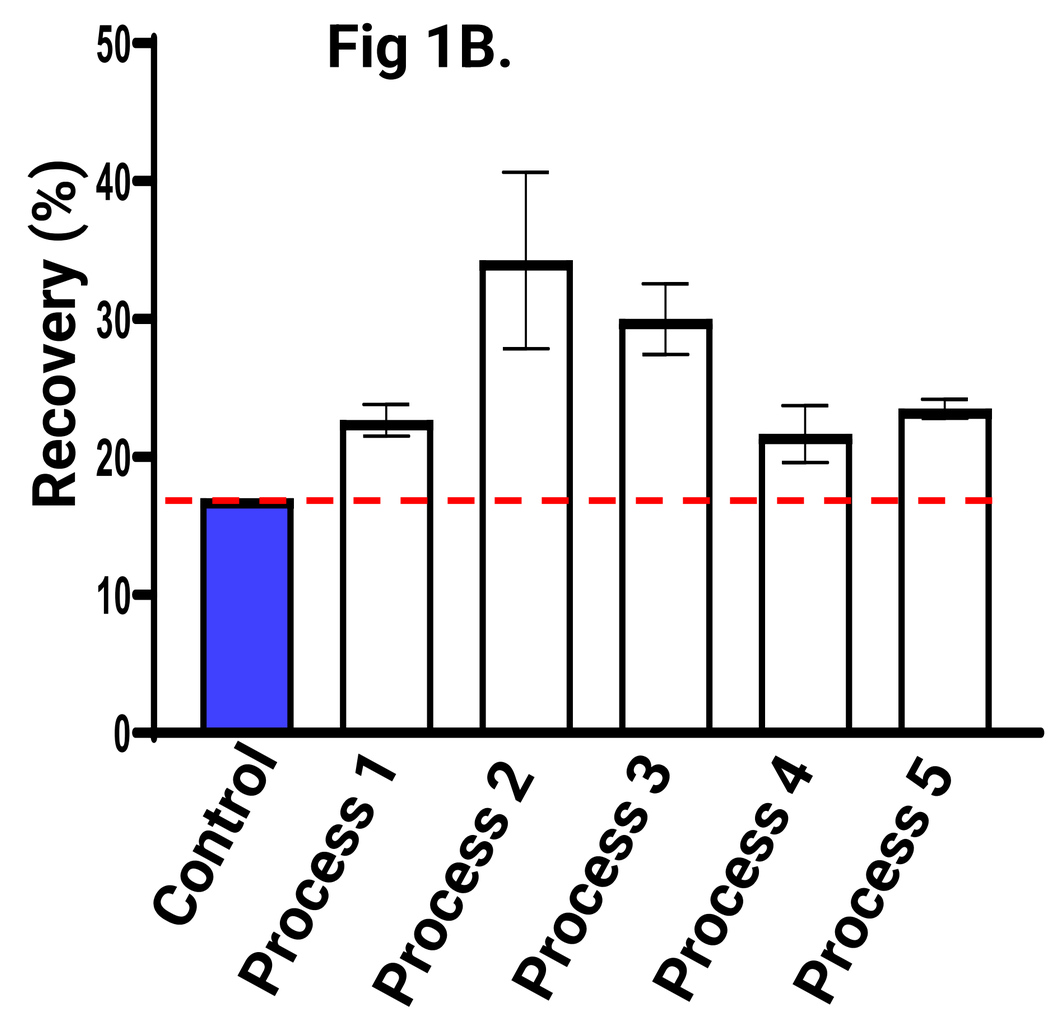
Fig 1B.
Recovery (%)
0
10
20
30
40
50
Control
Process 1
Process 2
Process 3
Process 4
Process 5

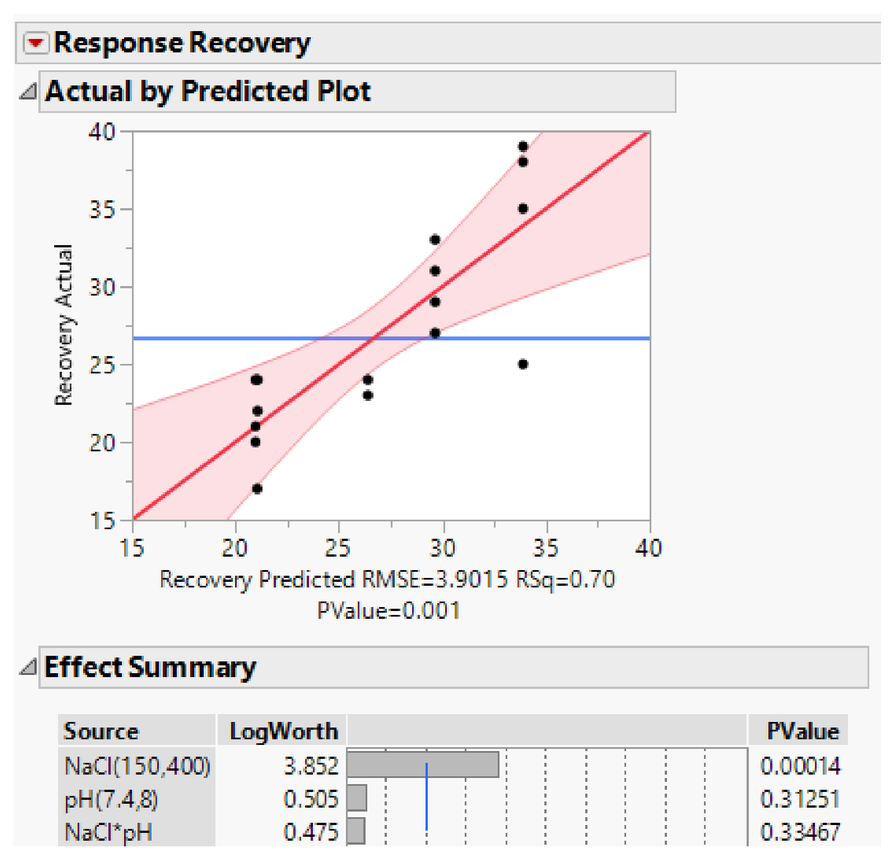
Fig 1C.
Response Recovery
Actual by Predicted Plot
Recovery Actual
Recovery Predicted RMSE=3.9015 RSq=0.70
PValue=0.001
Effect Summary
Source LogWorth PValue
NaCl(150,400) 3.852 0.00014
pH(7.4,8) 0.505 0.31251
NaCl*pH 0.475 0.33467

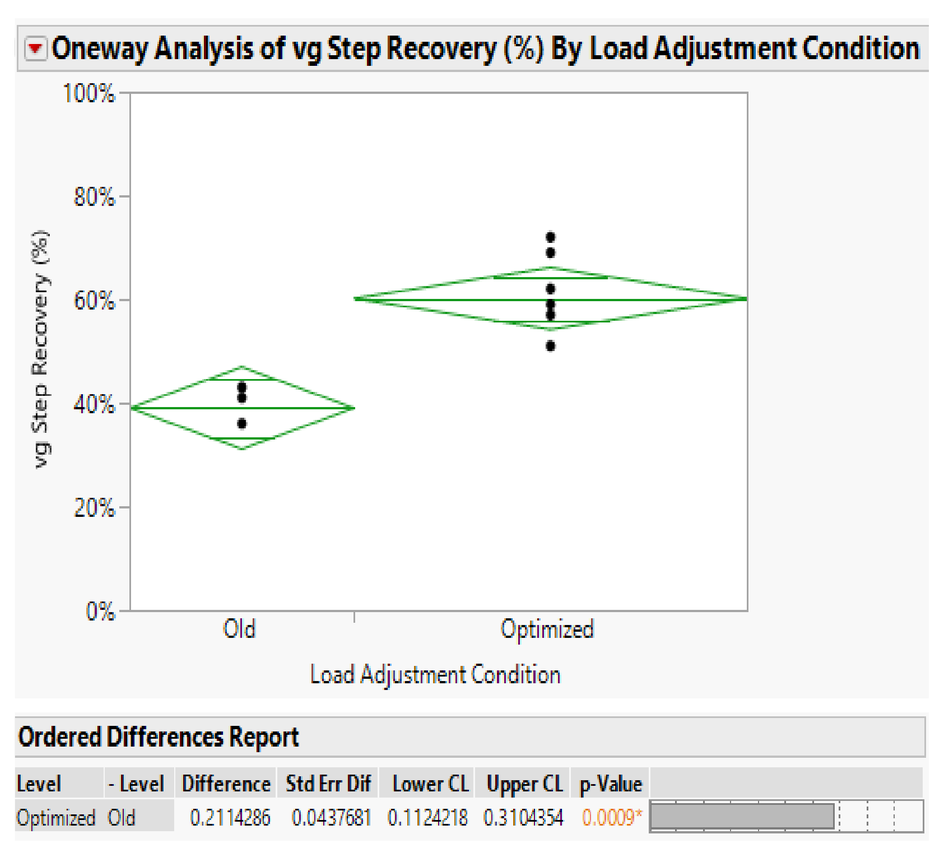
Fig 1D.
Oneway Analysis of vg Step Recovery (%) By Load Adjustment Condition
vg Step Recovery (%)
Old
Optimized
Load Adjustment Condition
Ordered Differences Report
Level - Level Difference Std Err Dif Lower CL Upper CL p-Value
Optimized Old 0.2114286 0.0437681 0.1124218 0.3104354 0.0009*

Fig 1. Load Adjustment Optimization. (A) 2 factor DoE performed in affinity batch purification looking at load adjustment conditions (B) Average of recovery percentage (C) Model from load adjustment optimization DoE. (D) The statistical analyses of control and optimized run by load conditioning showed the significant improvement (p-value 0.0009).

COLUMN DYNAMIC BINDING CAPACITY OVER RESIDENCE TIME

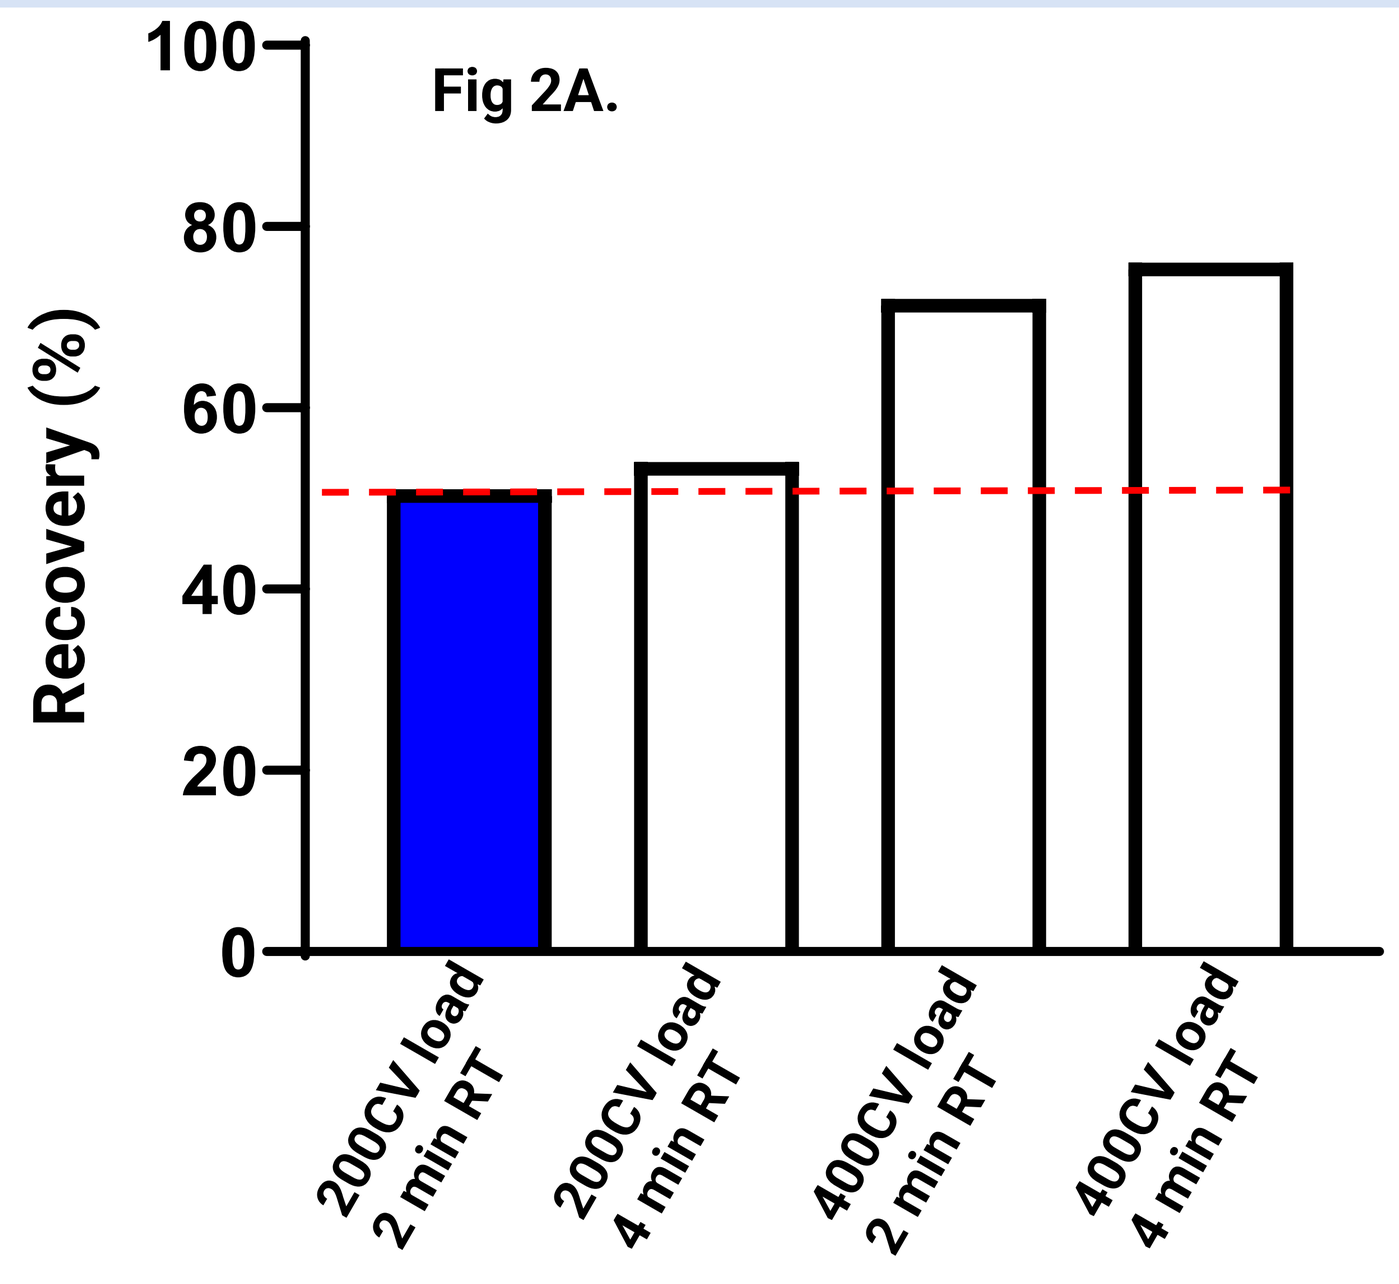
Fig 2A.
Recovery (%)
0
20
40
60
80
100
200CV load 2 min RT
200CV load 4 min RT
400CV load 2 min RT
400CV load 4 min RT

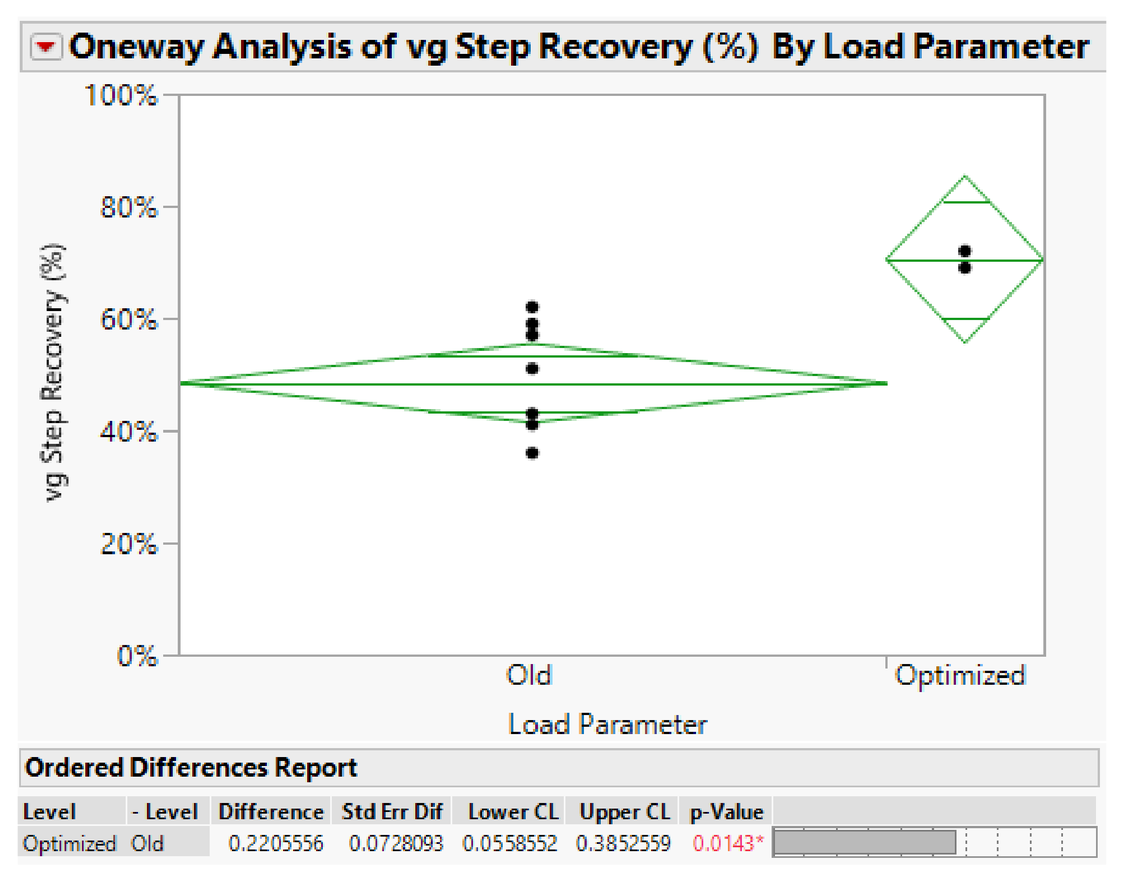
Fig 2B.
Oneway Analysis of vg Step Recovery (%) By Load Parameter
vg Step Recovery (%)
Old
Optimized
Load Parameter
Ordered Differences Report
Level - Level Difference Std Err Dif Lower CL Upper CL p-Value
Optimized Old 0.2205556 0.0728093 0.0558552 0.3852559 0.0143*

Fig 2. Column Dynamic Binding Capacity Over Residence Time Study. (A) Effect of column challenge over different residence time on recovery. (B) The statistical analyses of control and optimized run by load parameters including RT and load volume showed the significant improvement (p-value 0.0143).

DoE2: ELUTION BUFFER FORMULATION

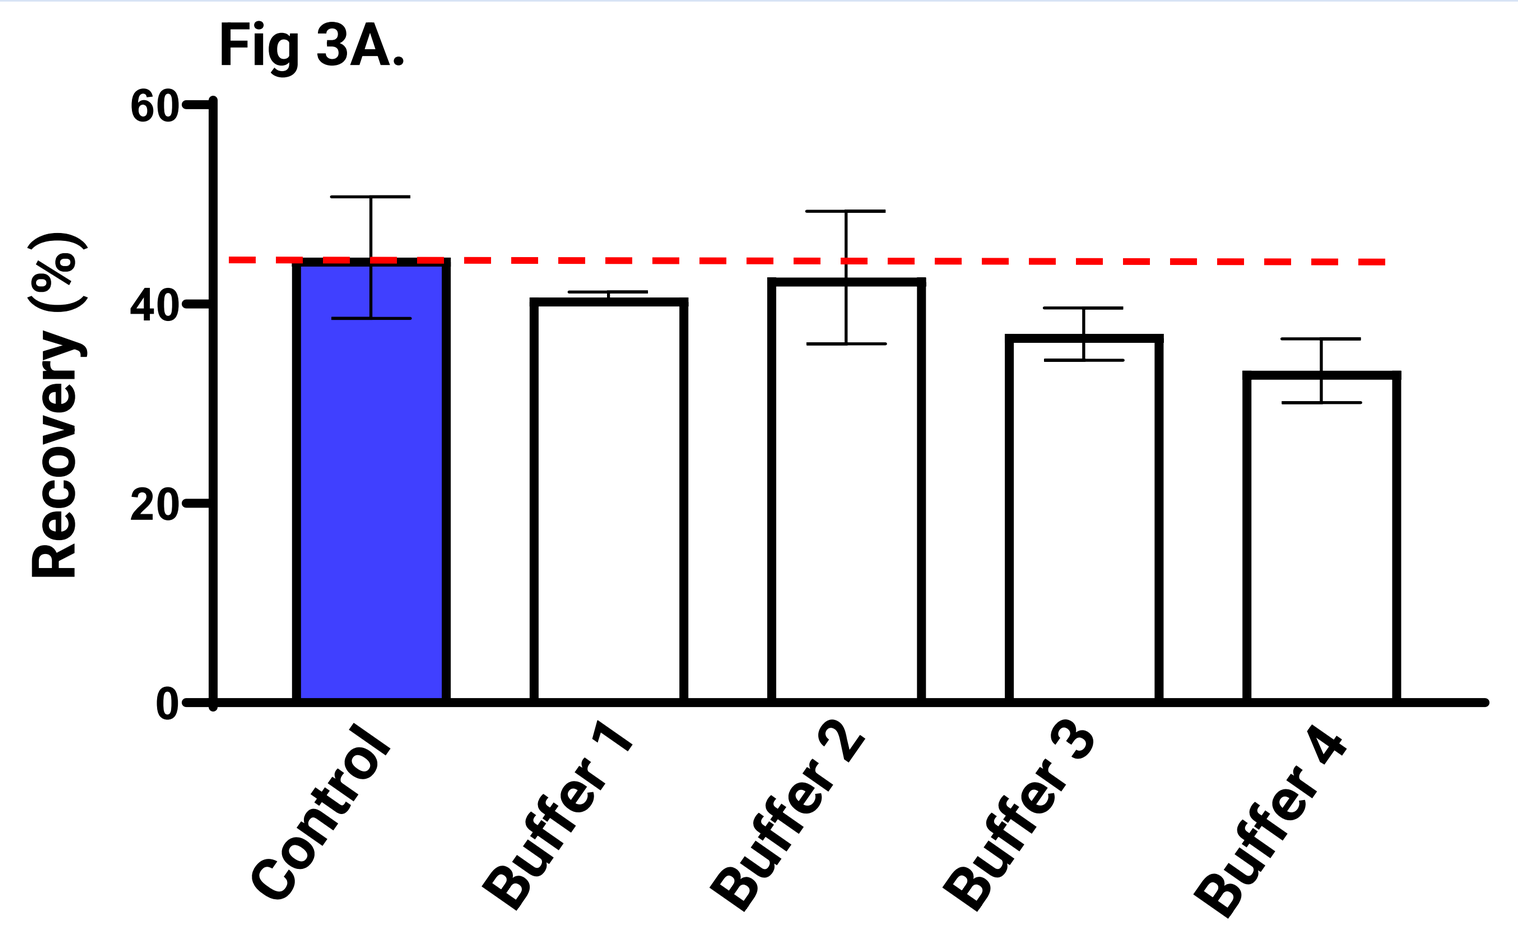
Fig 3A.
Recovery (%)
0
20
40
60
Control
Buffer 1
Buffer 2
Buffer 3
Buffer 4

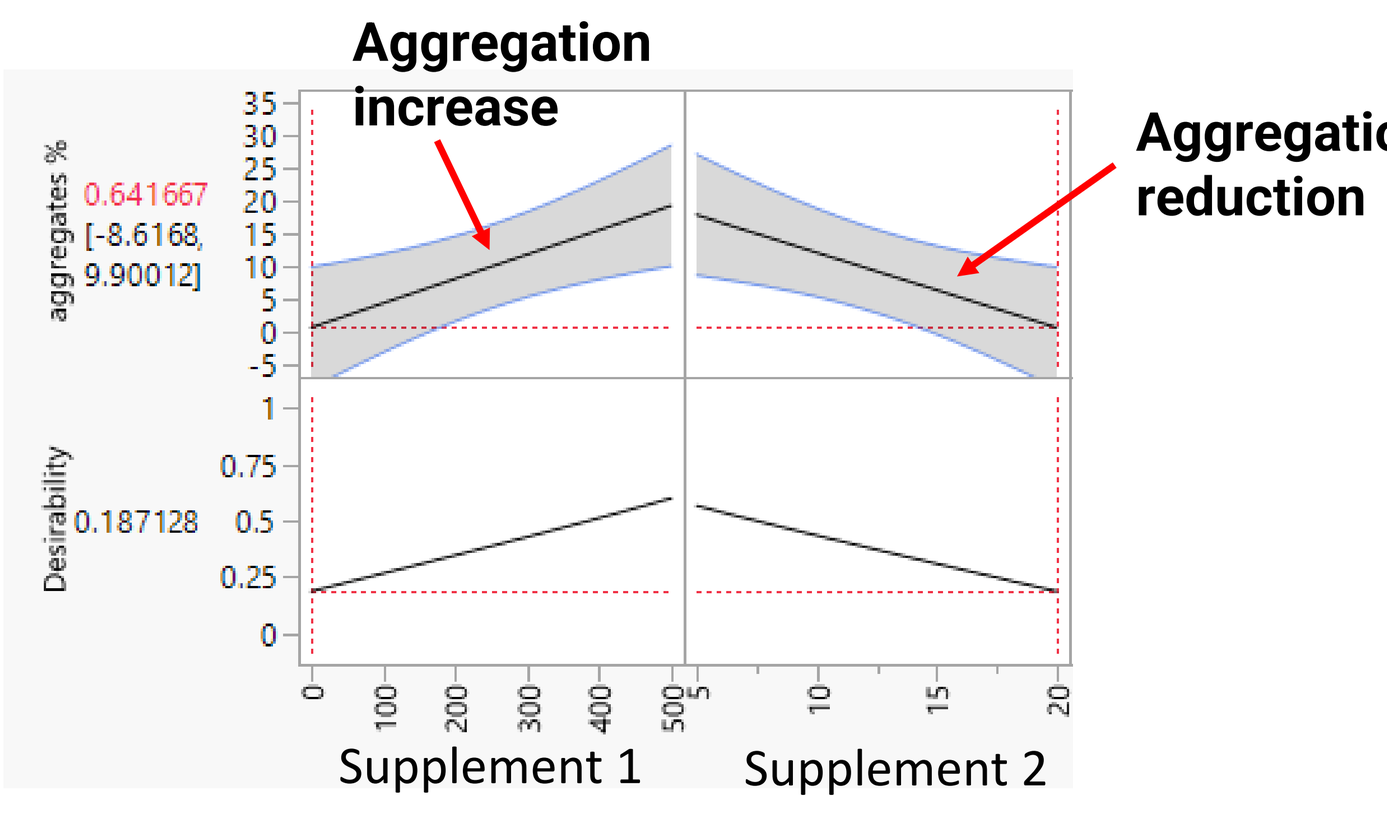
Fig 3B.
Aggregation increase
Aggregation reduction
aggregates %
0.641667
[-8.6168, 9.90012]
Desirability
0.187128
Supplement 1
Supplement 2

Fig 3. Elution Buffer Optimization. (A) 2 factor DoE performed in affinity batch purification looking at different elution buffers (B) The effect of addition of two different supplements in the elution buffer on eluate aggregation; data showed the increasing of supplement 1 concentration led to increase aggregation and the increasing of supplement 2 concentration led to aggregation reduction.

SCABILITY of THE OPTIMIZED AFFINITY CHROMATOGRAPHY

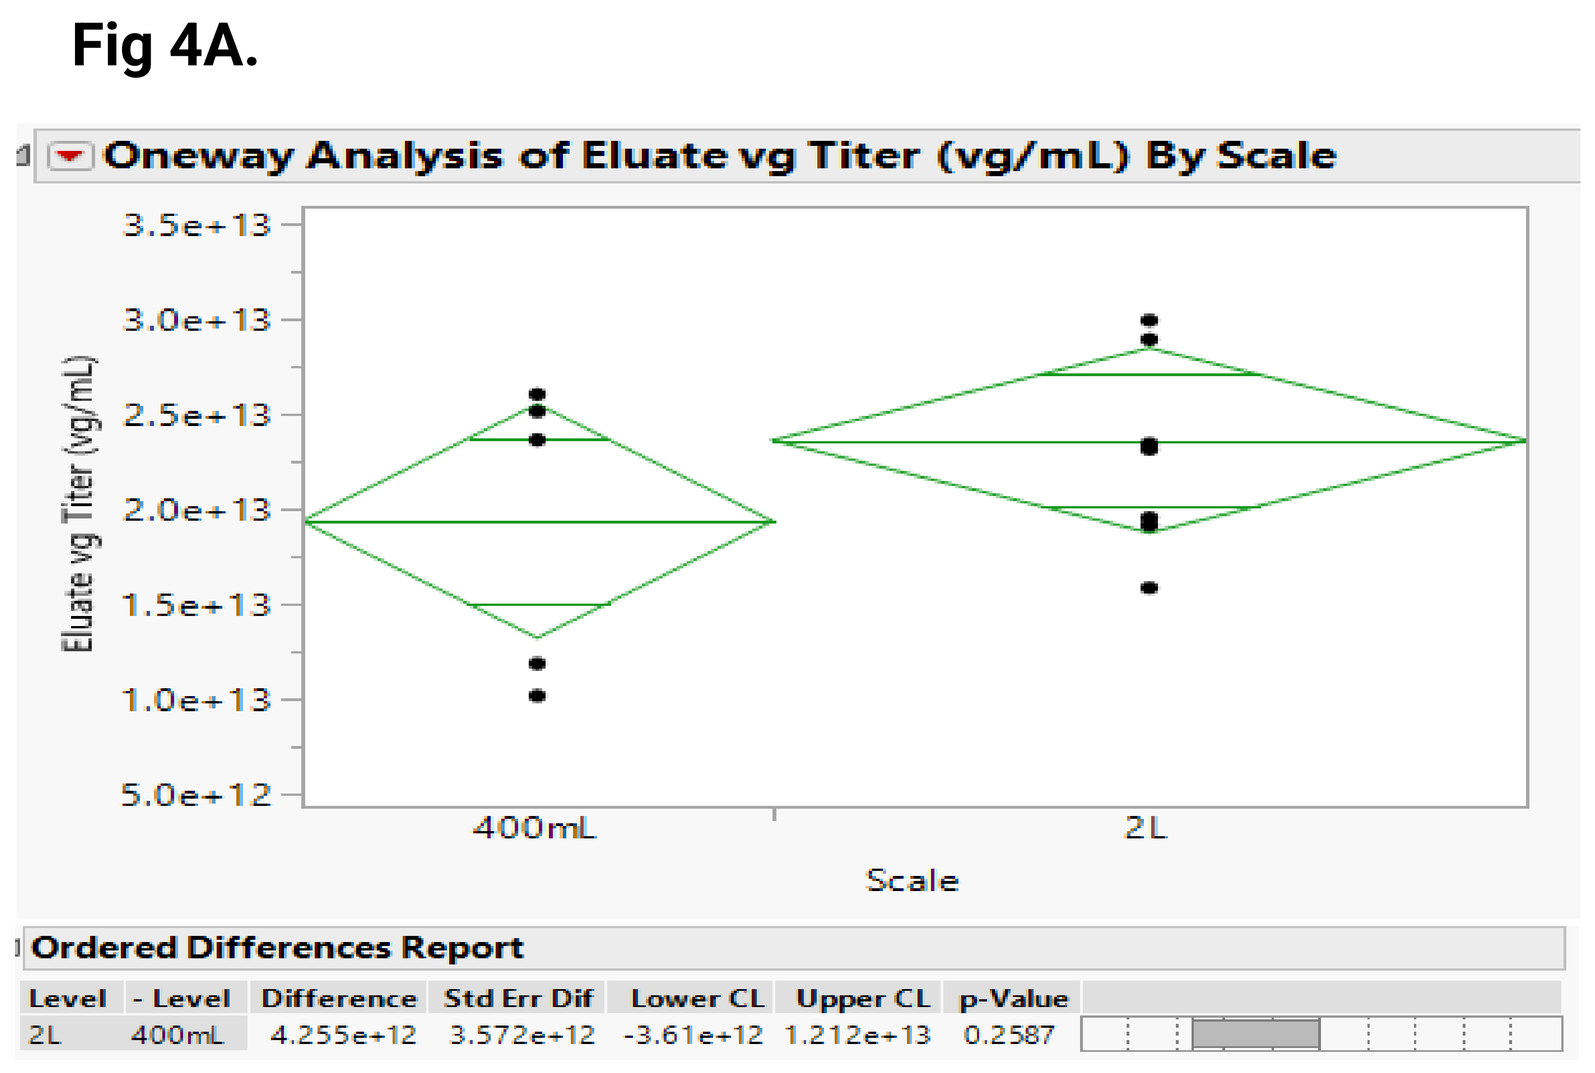
Fig 4A.
Oneway Analysis of Eluate vg Titer (vg/mL) By Scale
Eluate vg Titer (vg/mL)
400mL
2L
Scale
Ordered Differences Report
Level - Level Difference Std Err Dif Lower CL Upper CL p-Value
2L 400mL 4.255e+12 3.572e+12 -3.61e+12 1.212e+13 0.2587

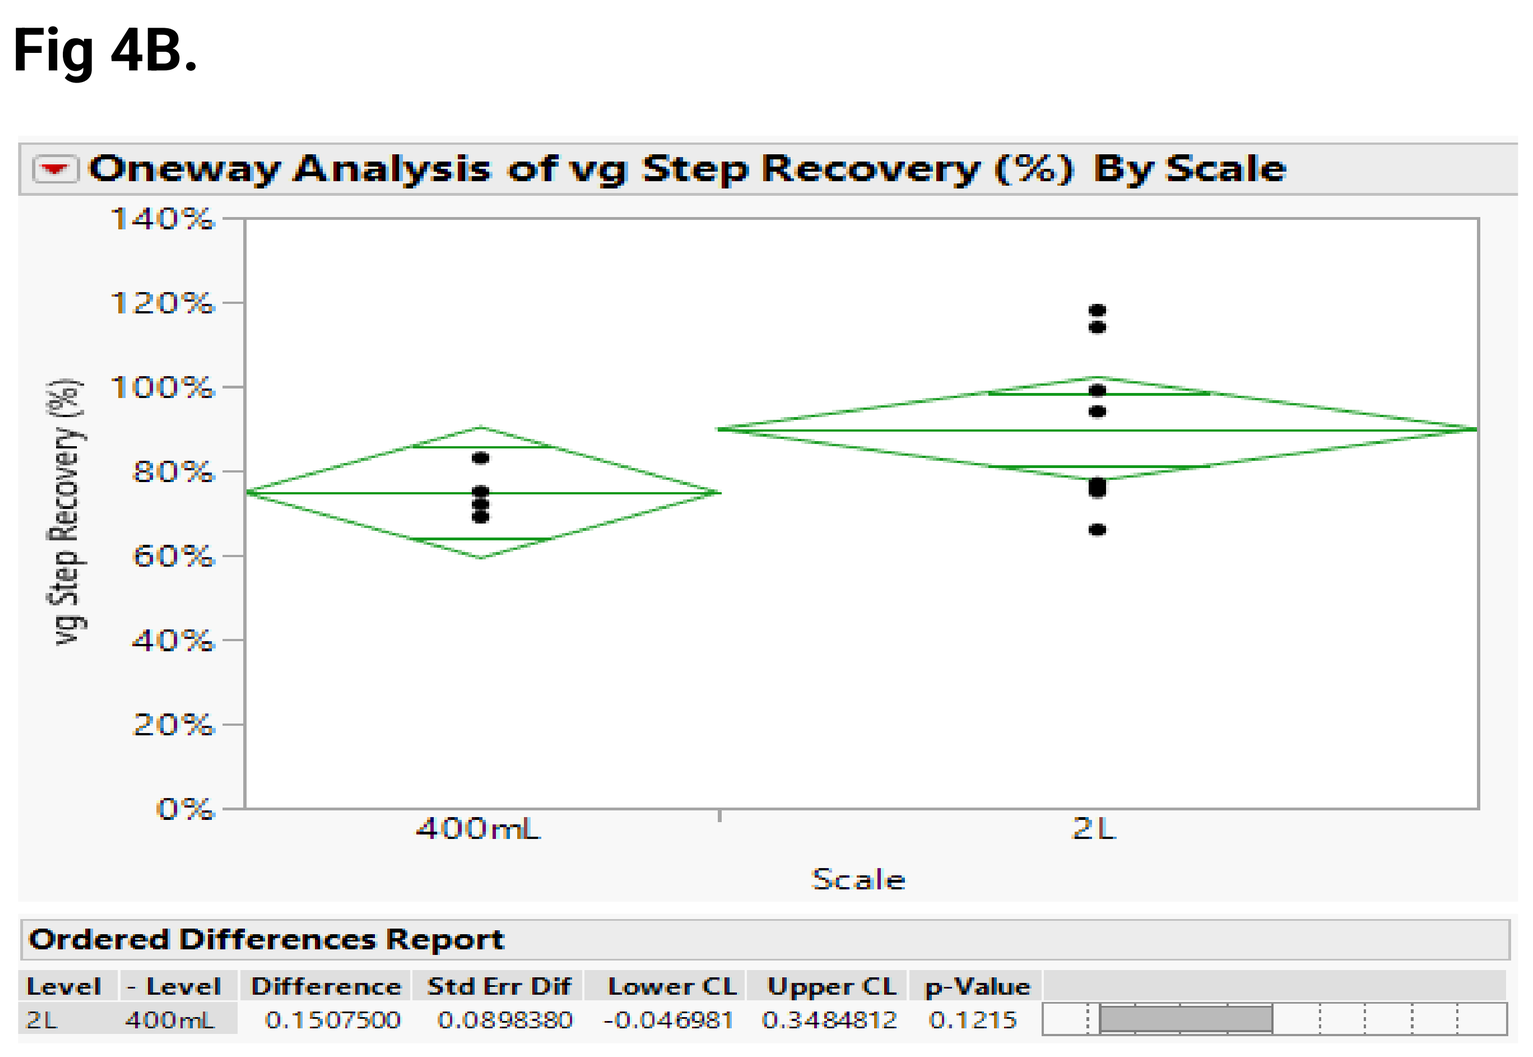
Fig 4B.
Oneway Analysis of vg Step Recovery (%) By Scale
vg Step Recovery (%)
400mL
2L
Scale
Ordered Differences Report
Level - Level Difference Std Err Dif Lower CL Upper CL p-Value
2L 400mL 0.1507500 0.0898380 -0.046981 0.3484812 0.1215

Fig 4. Scalability of the optimized affinity Chromatography. (A) The vg titer concentration of affinity eluate from 400 ml versus 2L scales showed no statistically significant difference between two scales (p-Value 0.2587) (B) Affinity step recovery data for the 400 ml small scale and 2 L scale-up runs showed no statistically significant difference between two scales (p-value 0.1215)

FULL AND EMPTY CAPSIDS PERCENTAGE

Mass-photometry and AUC Data (Affinity)

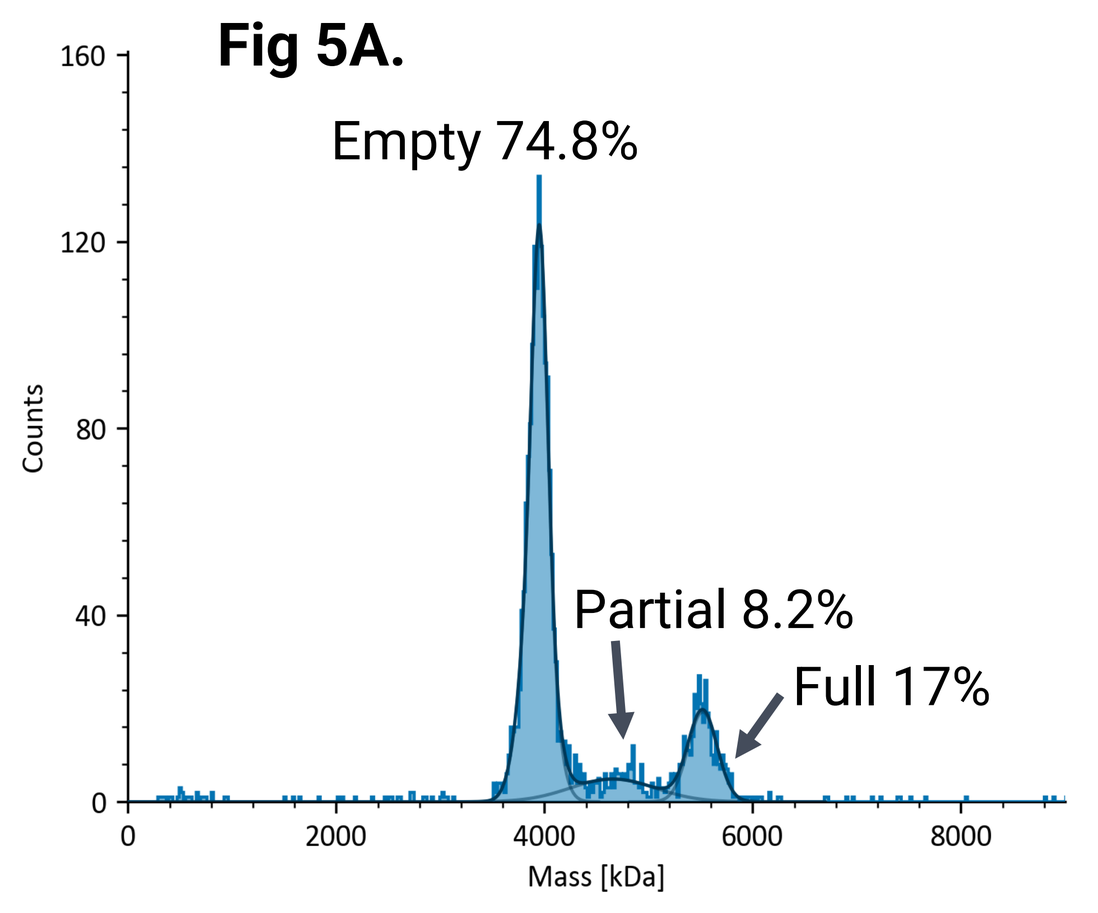
Fig 5A.
Empty 74.8%
Partial 8.2%
Full 17%
Counts
Mass [kDa]

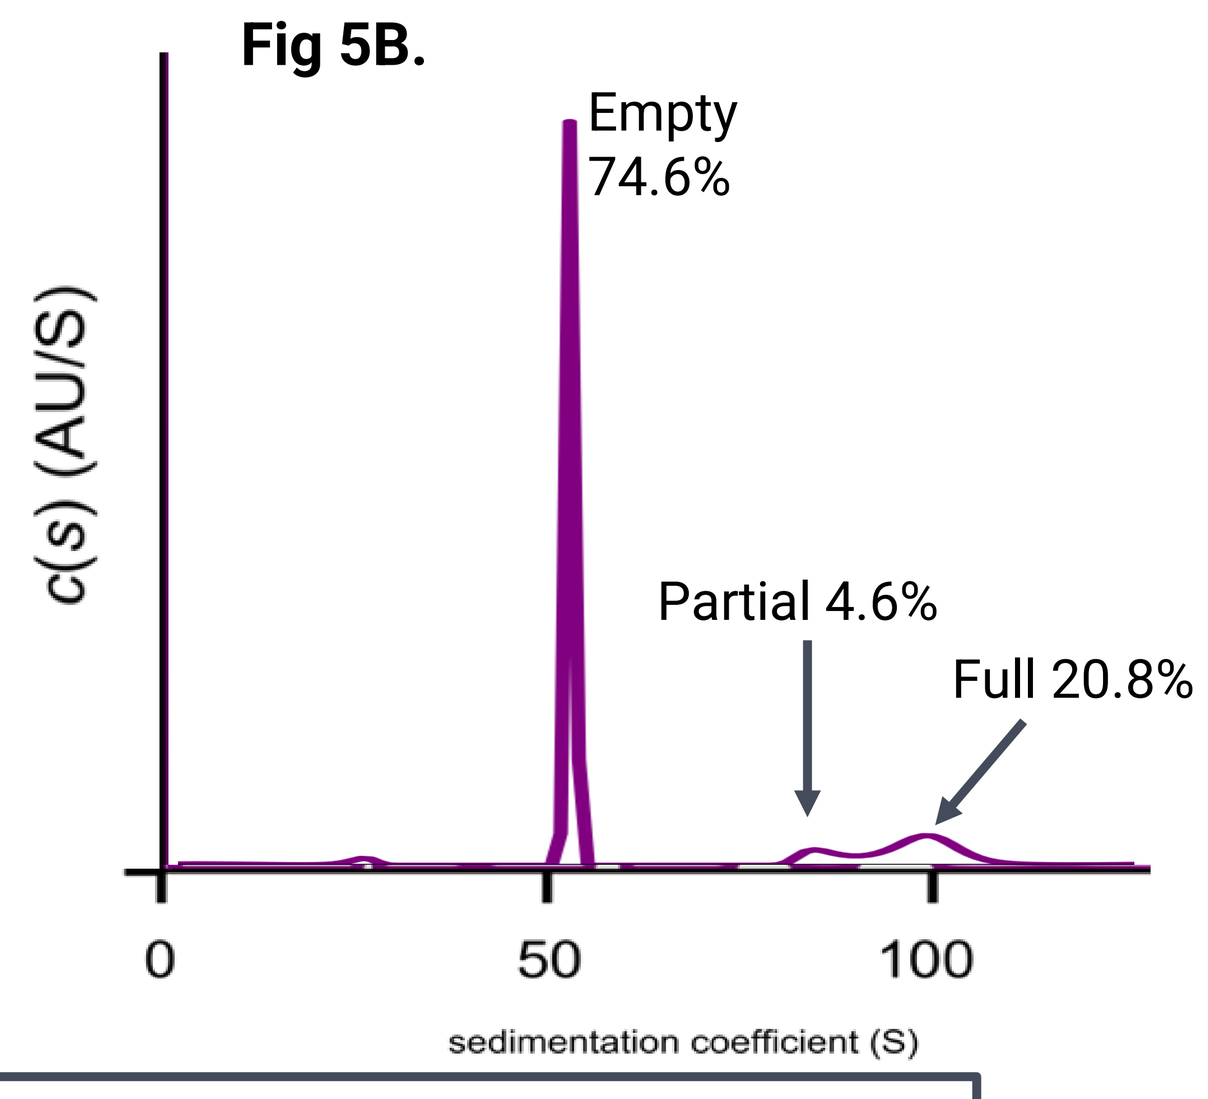
Fig 5B.
Empty 74.6%
Partial 4.6%
Full 20.8%
c(s) (AU/S)
sedimentation coefficient (S)

Full Capsids 20%

Mass-photometry and AUC Data (IEX Enrichment)

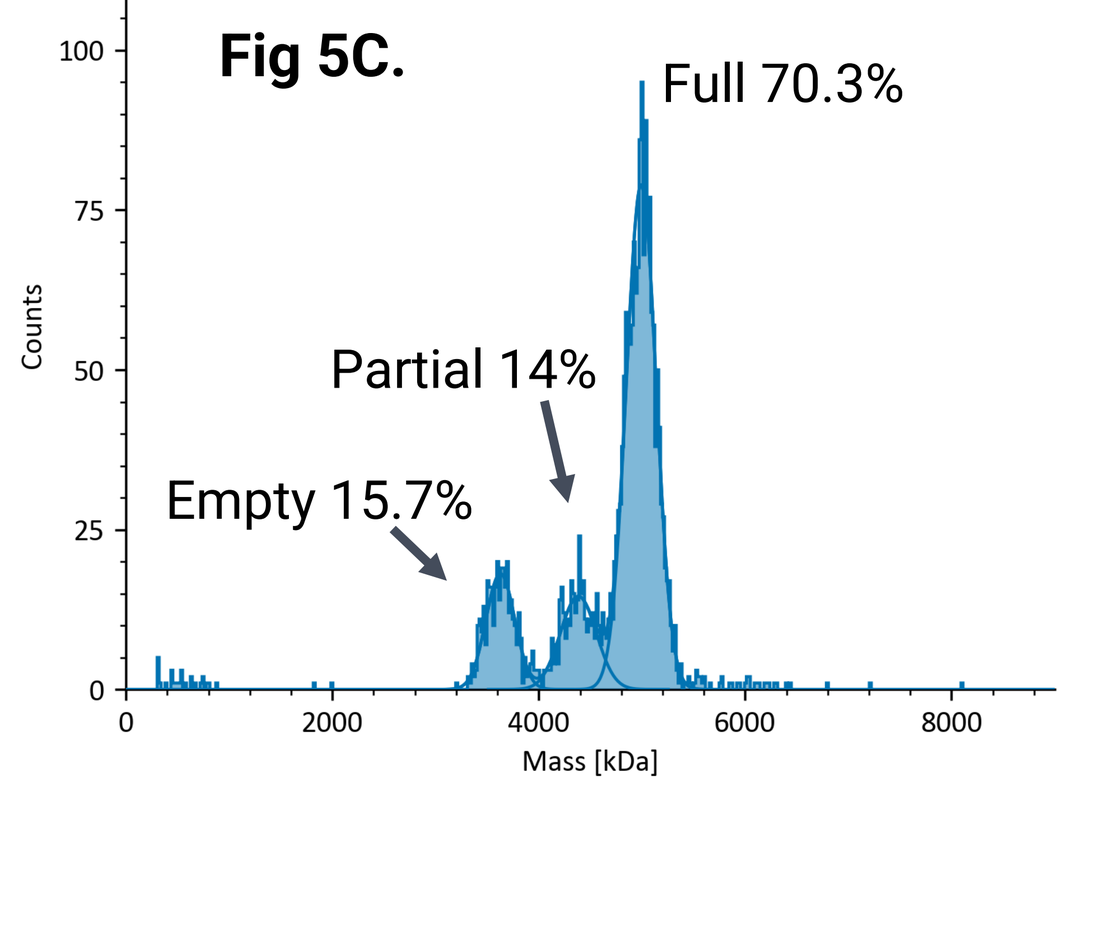
Fig 5C.
Full 70.3%
Partial 14%
Empty 15.7%
Counts
Mass [kDa]

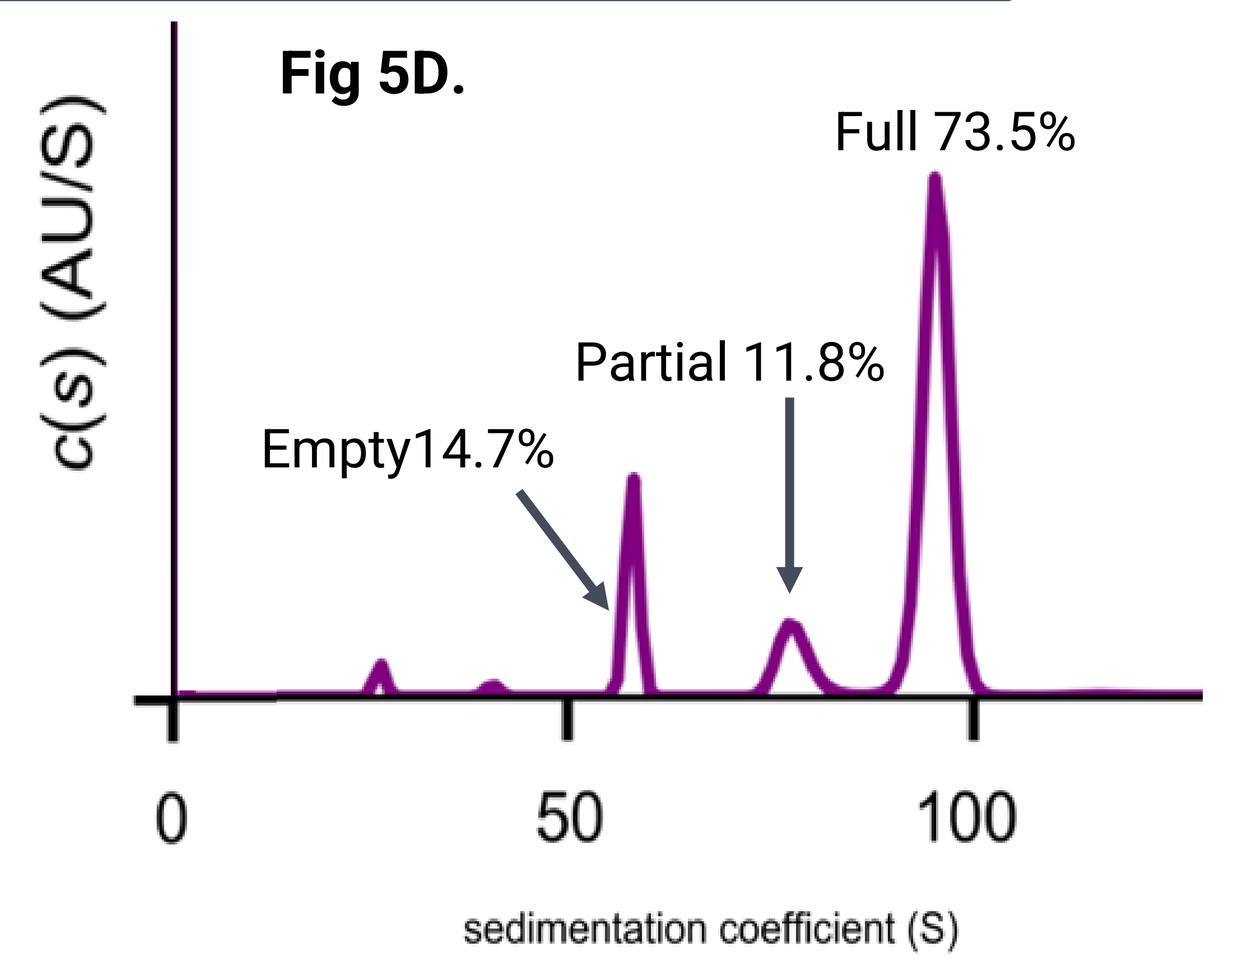
Fig 5D.
Full 73.5%
Partial 11.8%
Empty14.7%
c(s) (AU/S)
sedimentation coefficient (S)

Full Capsids 73%

Fig 5. Full and empty capsids percentage. (A) Mass photometry of affinity eluate from 2L scale purification (B) AUC data of affinity eluate from 2L scale purification (C) Mass photometry of IEX eluate from 2L scale purification and (D) AUC data of IEX eluate from 2L scale purification

CONCLUSION

Our data demonstrates the optimized and scalable AAV6 affinity purification.

- The AAV recovery by affinity chromatography was improved about two times, from 15% to >35%, as a result of the DoE study evaluating the load conditioning parameters.
- The AAV recovery was improved to >70% by optimization of load volume over time.
- The amount of capsid aggregation could be reduced by optimization of buffer used for AAV elution from the affinity chromatography column.
- The optimized affinity purification showed to be scalable from shake flask to 2L reactor.
- The percent of full capsids was enriched from 20% in the affinity eluate to >70% after IEX purification.

Contact Information

Tess Kitchener

914-471-1931

tkitchener@elevate.bio