Design and Production of Circular RNAs (circRNAs) with Validated Functional Activity in Cell Models

ElevateBio®, 225 Wyman St, Waltham MA 02451, U.S.

Isabella Darke, Patrick Magunga, Meemanage De Zoysa, Nicholas Dreisbach, Miranda Yang, Dana Cairns, Stacie L. Seidel, Austin Thiel, Melissa Carpenter, Cherylene Plewa

Abstract

The COVID-19 pandemic has highlighted the therapeutic potential of Ribonucleic Acid (RNA) after messenger RNA (mRNA) was successfully used in vaccines developed to combat the virus. Despite this success, mRNA technology has challenges including reactogenicity, poor stability, and high cost of manufacturing. Circular RNA (circRNA), which is a covalently closed RNA, is being developed to overcome the shortcomings of mRNA to achieve longer therapeutic expression. However, generating circRNAs with low impurities at a large scale has proven to be a challenge in the RNA field.

At ElevateBio, we developed a robust and scalable method to produce high quality circRNAs using a Permuted Intron-Exon (PIE)-based construct design. A set of characterization assays was also developed and implemented to ensure the quality of final circRNA products. Initial testing validates the circRNA platform and demonstrates that ElevateBio circRNAs have low toxicity in cells, express robust levels of transgene products and show functional activity in cell models. The current circRNA design and production method are undergoing further development to be used in a range of therapeutic applications both *ex vivo* and *in vivo*.

Background

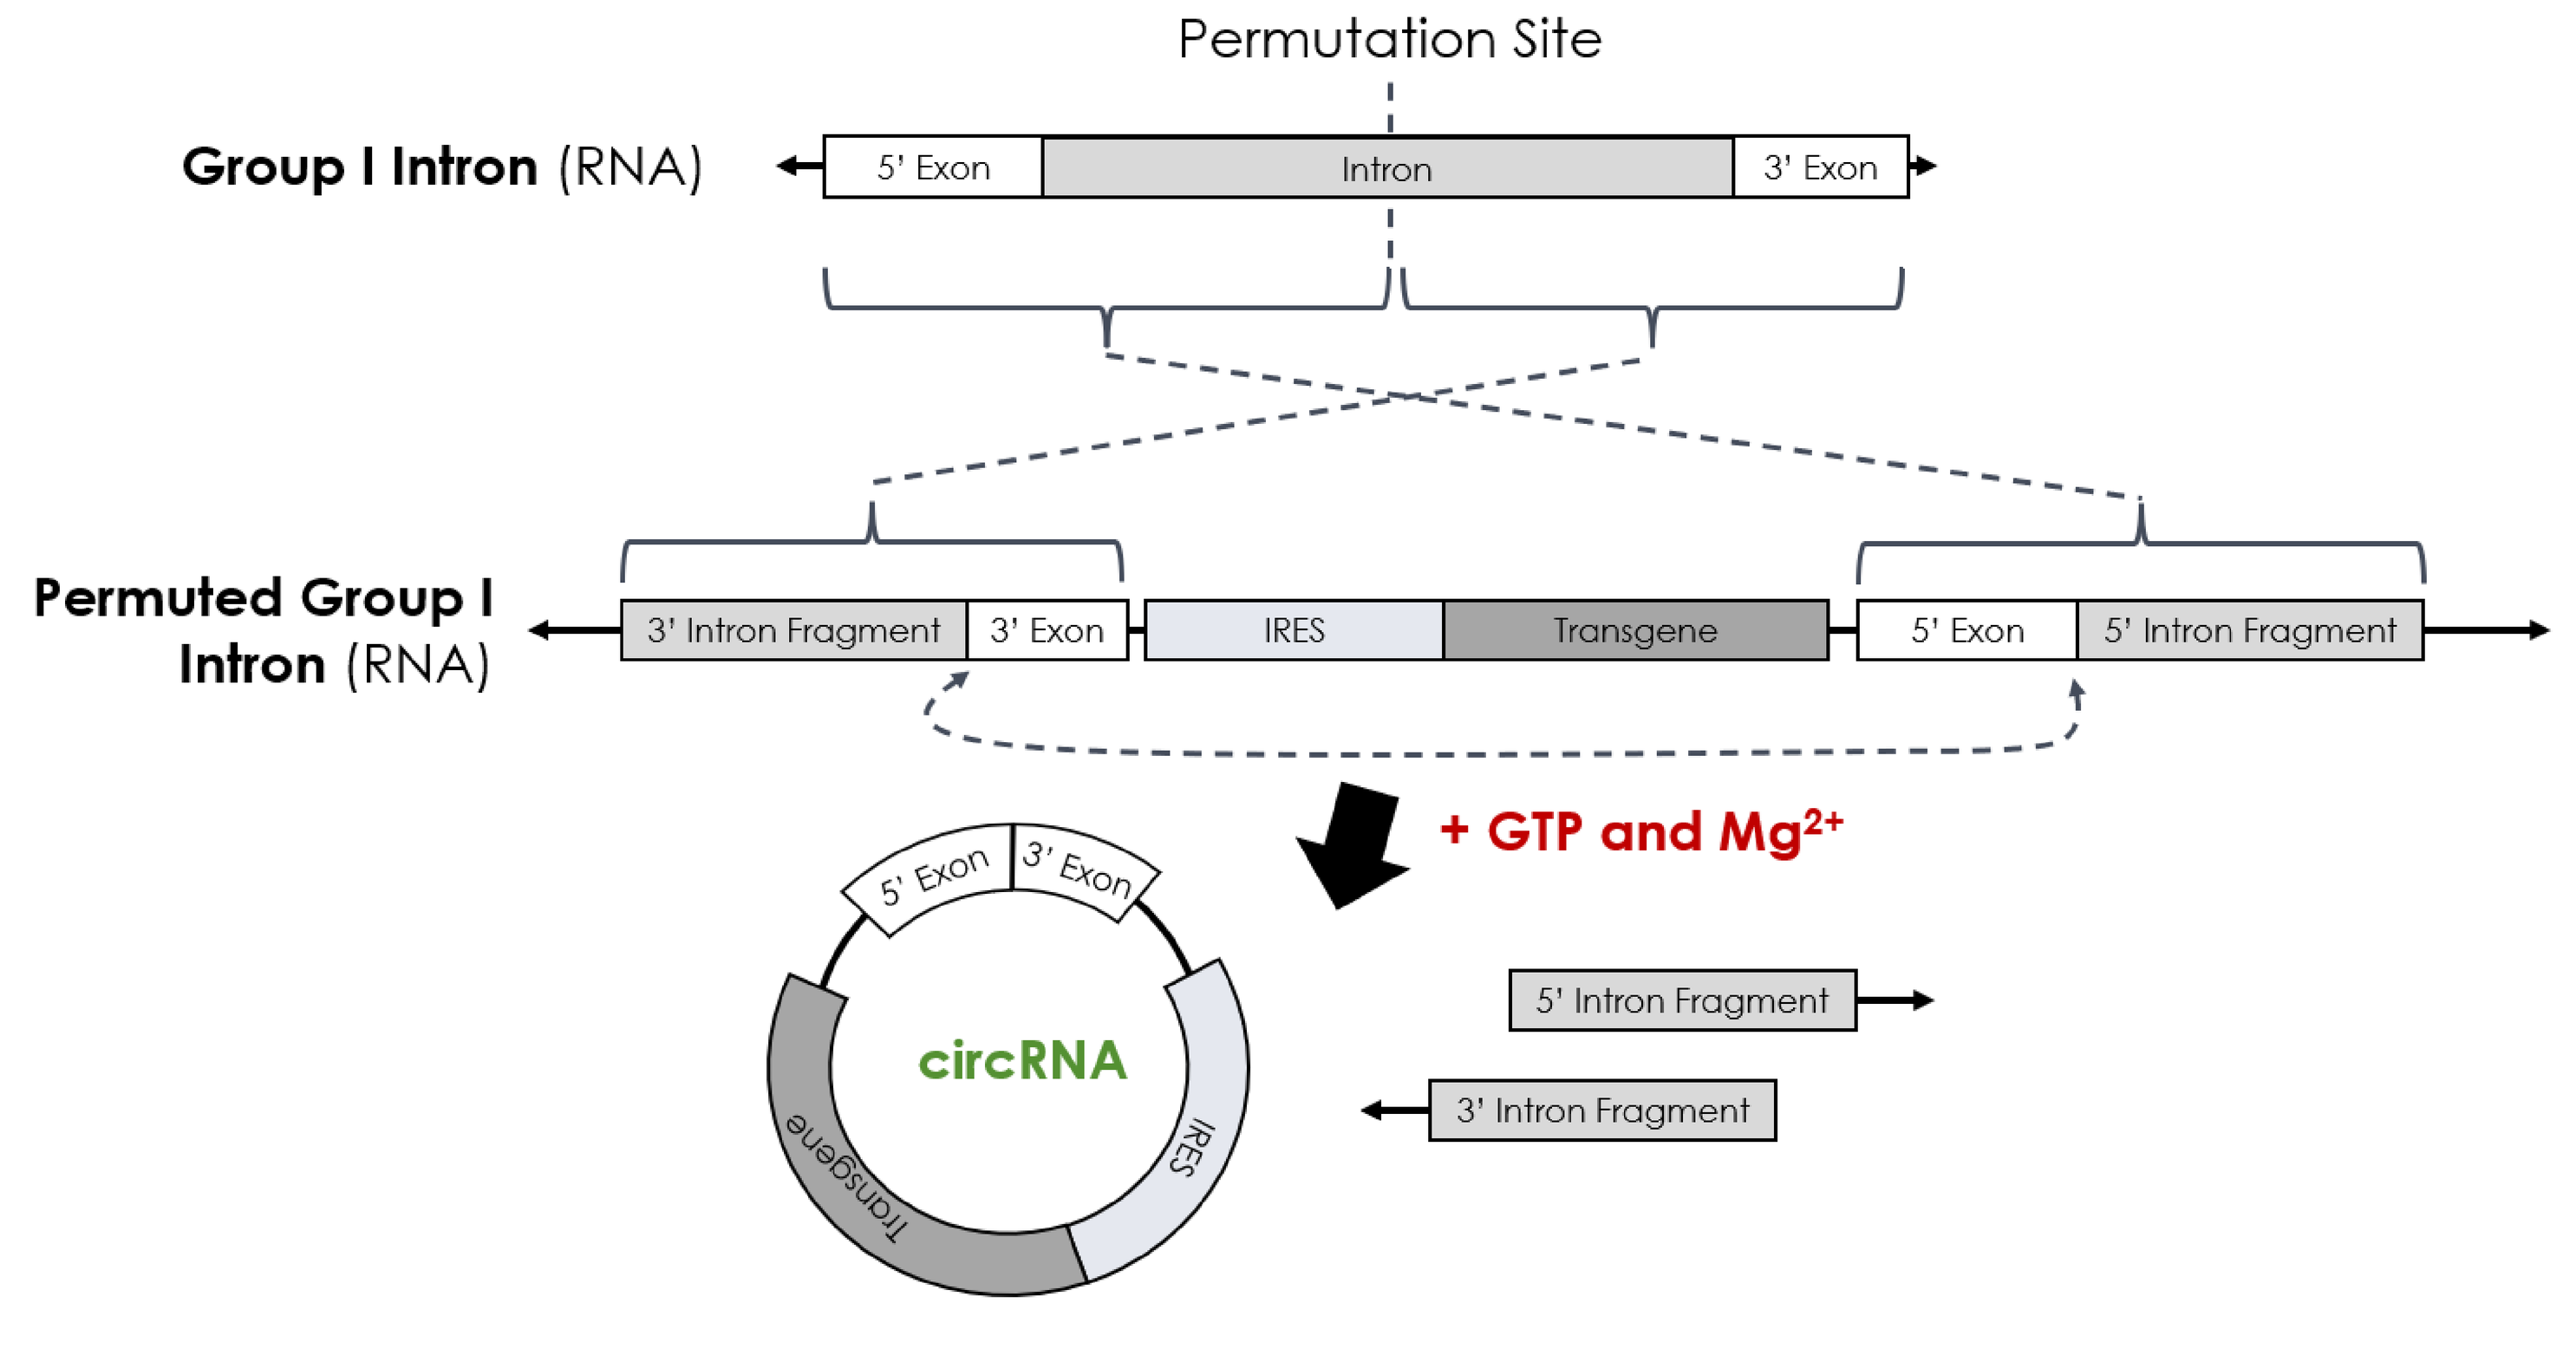
Permutation Site
Group I Intron (RNA)
5' Exon
Intron
3' Exon
Permuted Group I Intron (RNA)
3' Intron Fragment
3' Exon
IRES
Transgene
5' Exon
5' Intron Fragment
+ GTP and Mg2+
circRNA
5' Intron Fragment
3' Intron Fragment

Schematic of RNA Circularization. CircRNA constructs are covalently closed molecules without 5' caps and 3' tails, and they are formed by a ribozymatic back splicing mechanism. The circRNA constructs contain an internal ribosomal entry site (IRES) and transgene inserted into a Permuted Group I intron design. GTP and Magnesium are added as additional cofactors to facilitate circularization via intron splicing. Each component of the circRNA construct can be optimized to enhance circularization and expression.

Process Schematic

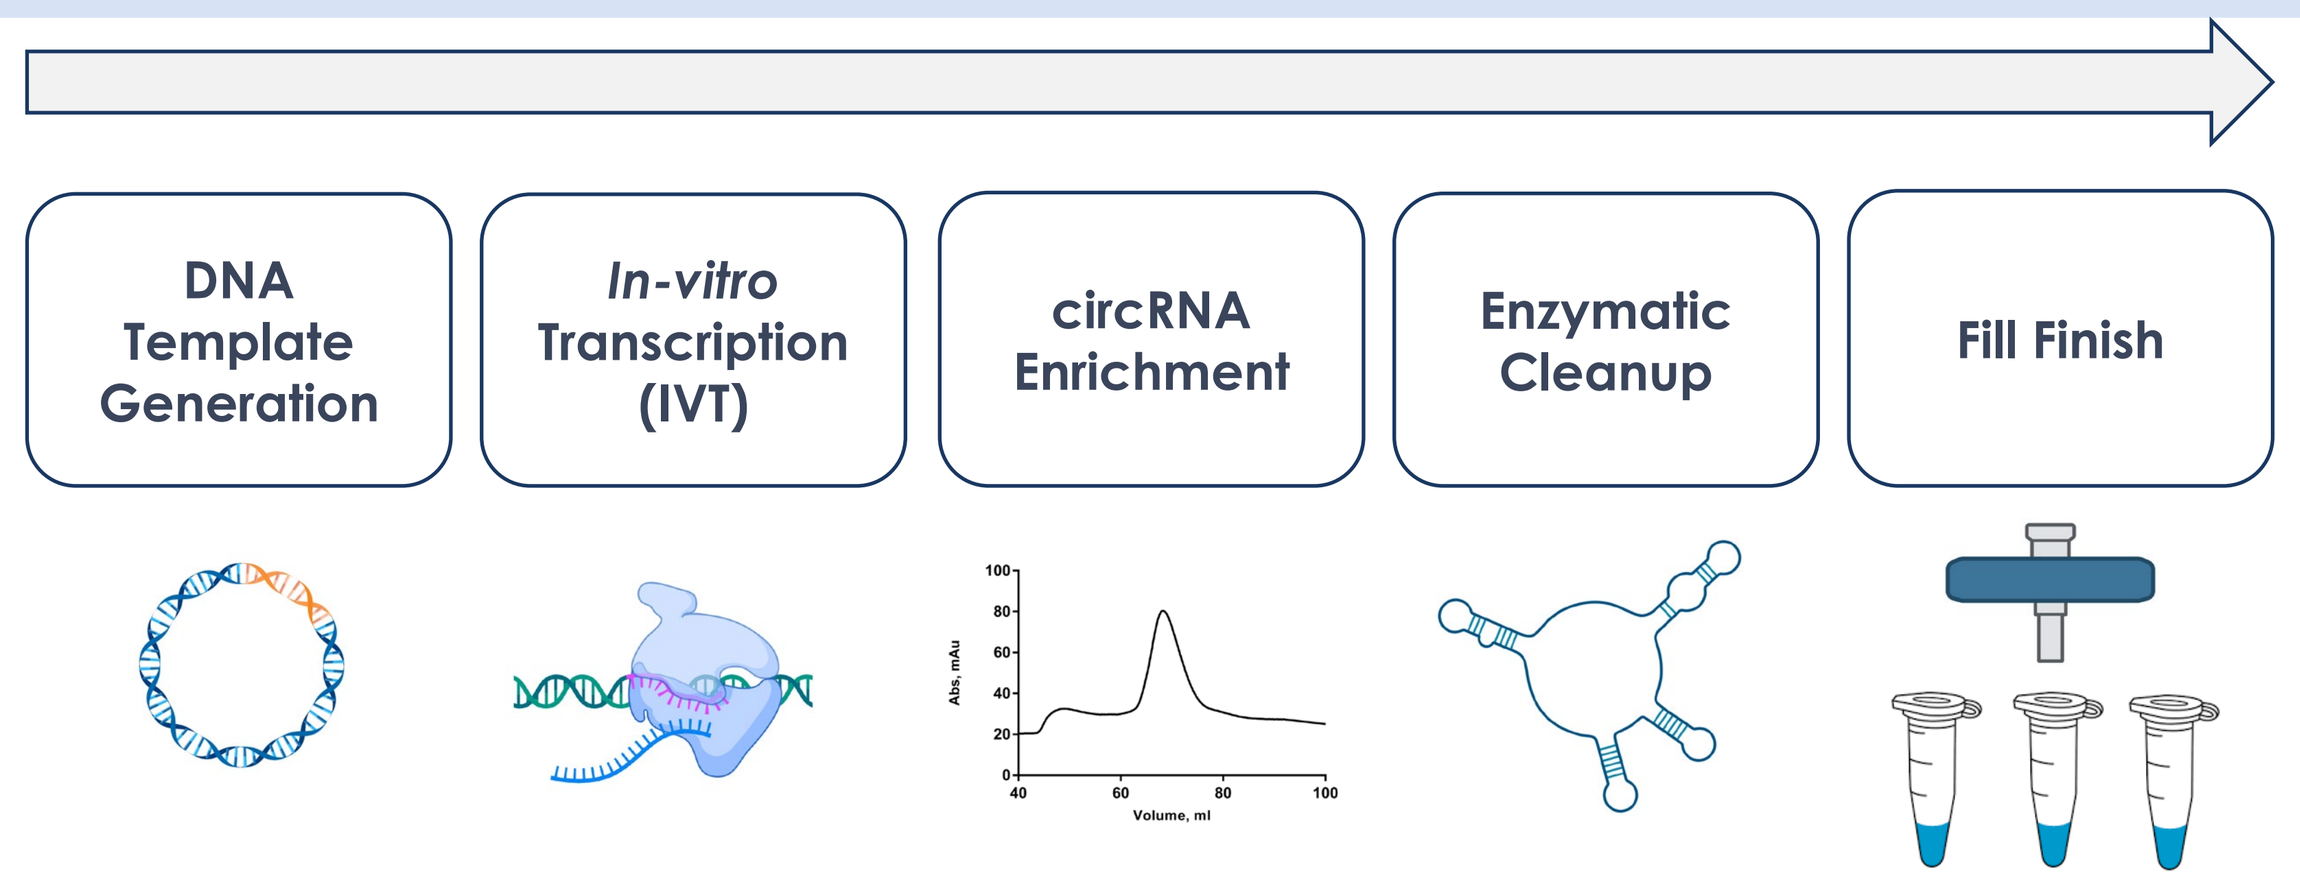
DNA Template Generation
In-vitro Transcription (IVT)
circRNA Enrichment
Enzymatic Cleanup
Fill Finish

circRNA Production Process: circRNA is generated from precursor RNA which is transcribed *in vitro* from a linearized plasmid DNA template. The sample is then sequentially enriched for circRNA through a combination of chromatography-based purification and enzymatic cleanup. Final material is sterile filtered prior to downstream analysis. To generate clinic-ready circRNA material, we developed a Controlled Laboratory Operations (CLO) process which involved robust documentation practices, tracking and quality control of all raw materials, and extensive operator training on locked protocol execution. Figure created with BioRender.com

ElevateBio Process Enriches for circRNA

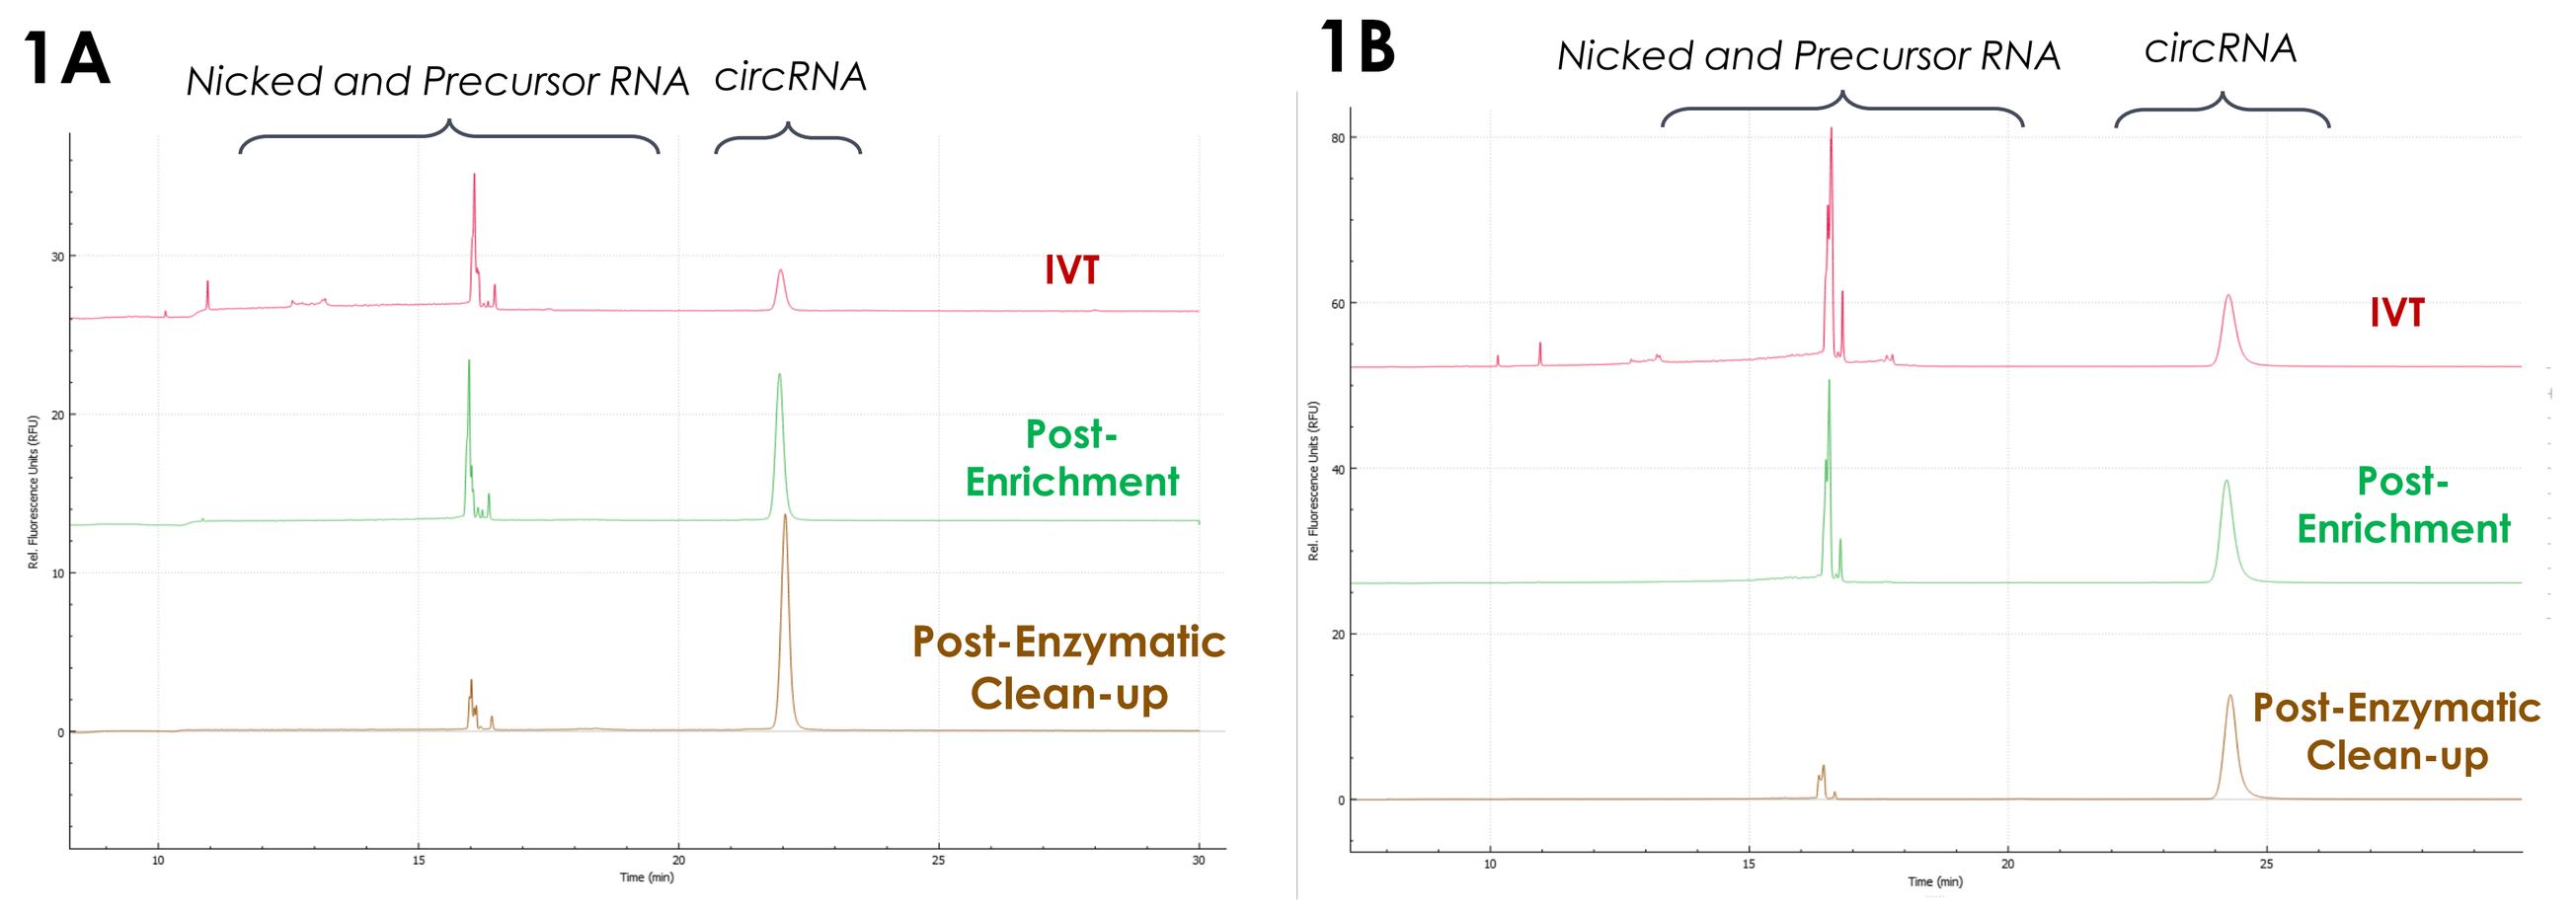
1A
Nicked and Precursor RNA
circRNA
IVT
Post-Enrichment
Post-Enzymatic Clean-up
1B
Nicked and Precursor RNA
circRNA
IVT
Post-Enrichment
Post-Enzymatic Clean-up

Figure 1: Step Comparison of CGE Data. Sequential purification steps significantly reduce the quantity of high and low molecular weight non-circular RNA impurities, including nicked and unspliced (precursor) RNA. Relative amounts of each species in circRNA-1 and circRNA-2 samples (Figure 1A and 1B, respectively) are quantified by capillary gel electrophoresis (CGE). In both samples, circRNA is enriched from a starting purity of 20-40% post-*in vitro* transcription (IVT) to a final purity of over 80%.

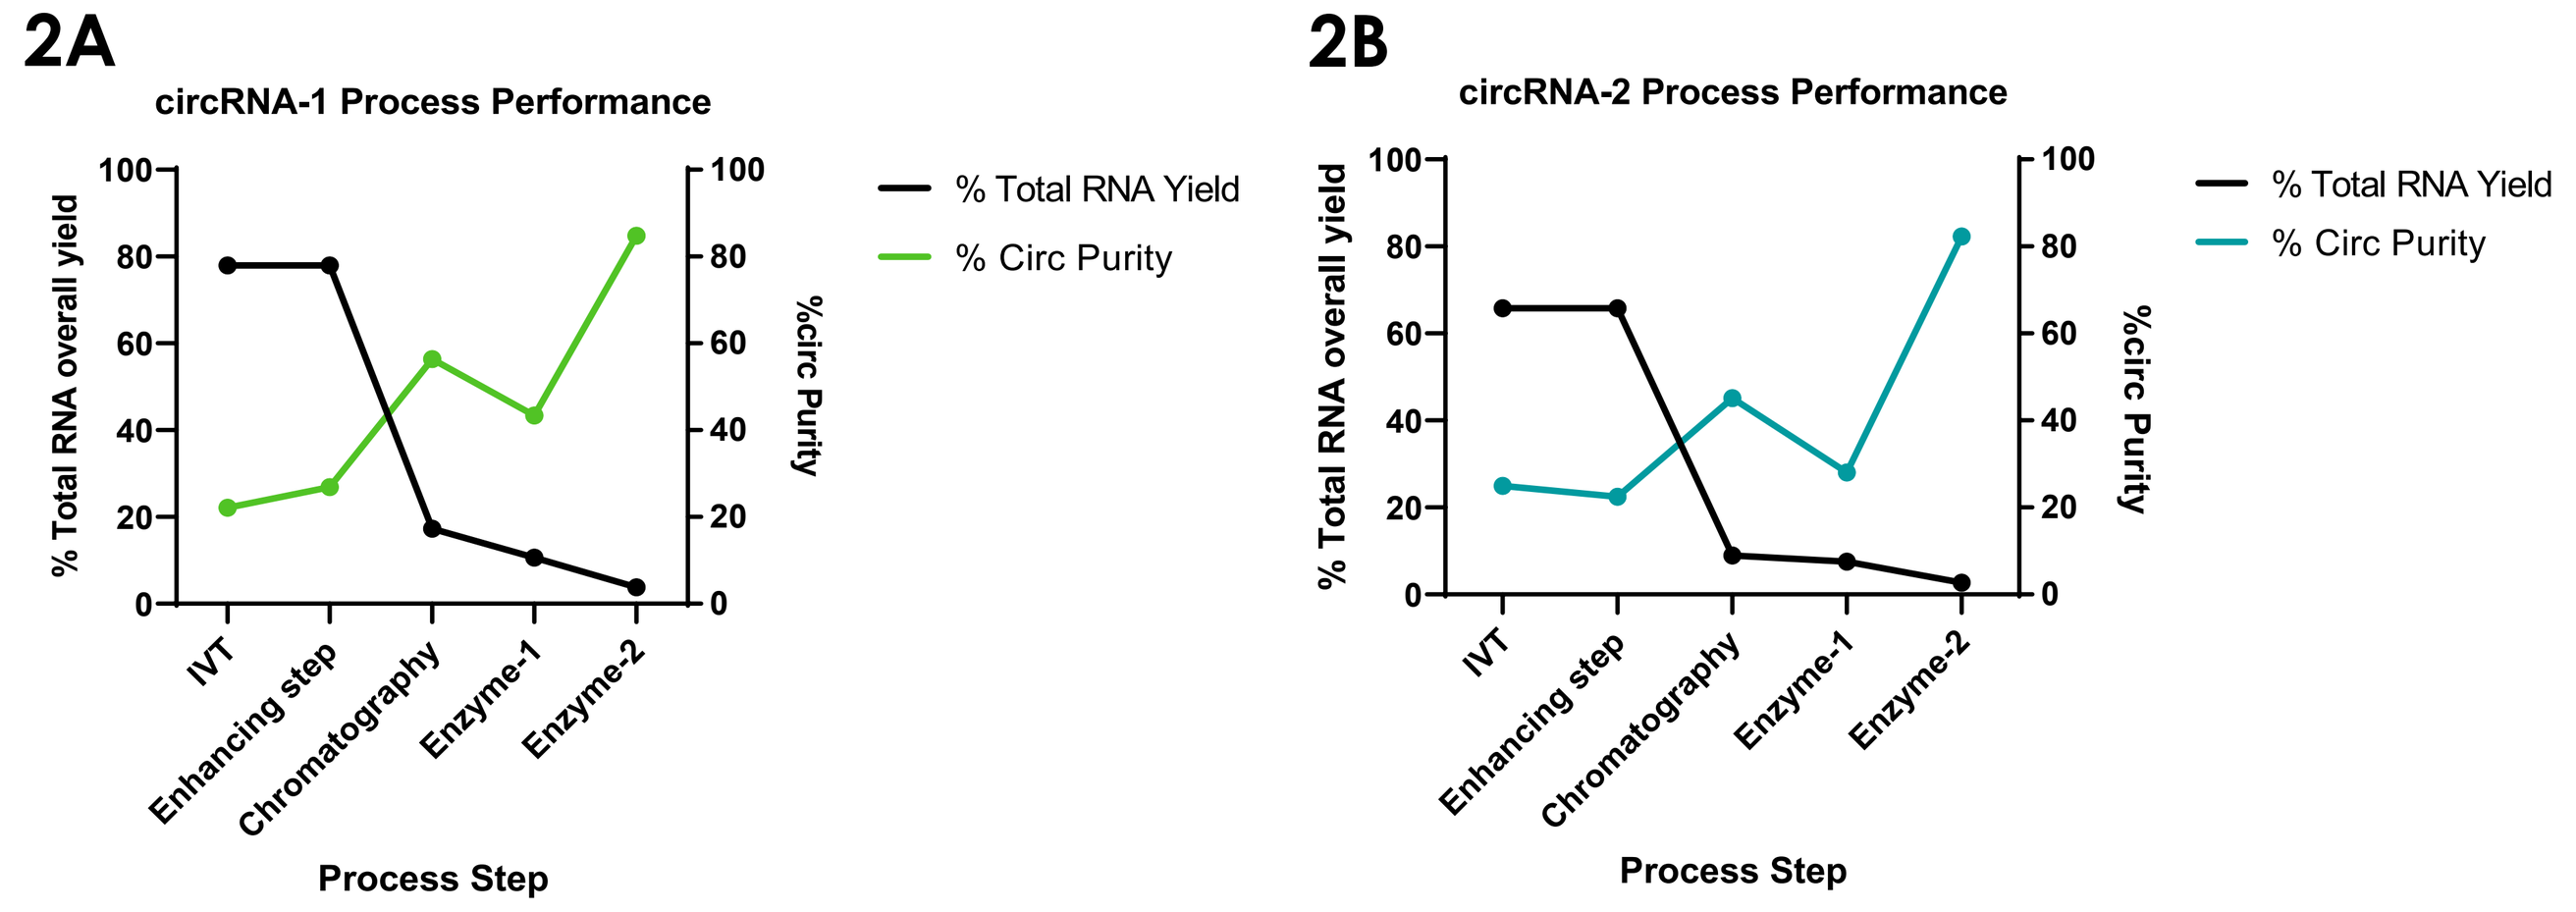
2A
circRNA-1 Process Performance
% Total RNA overall yield
%circ Purity
% Total RNA Yield
% Circ Purity
IVT
Enhancing step
Chromatography
Enzyme-1
Enzyme-2
Process Step
2B
circRNA-2 Process Performance
% Total RNA overall yield
%circ Purity
% Total RNA Yield
% Circ Purity
IVT
Enhancing step
Chromatography
Enzyme-1
Enzyme-2
Process Step

Figure 2: Performance of circRNA Enrichment Process Developed at ElevateBio. Two key steps are responsible for the enrichment of circRNA to achieve final circRNA purity >80%. The process shows robustness across different circRNA constructs (Figure 2A and 2B) based on overall yield and purity.

Scaled-up Process Meets High %circ Threshold

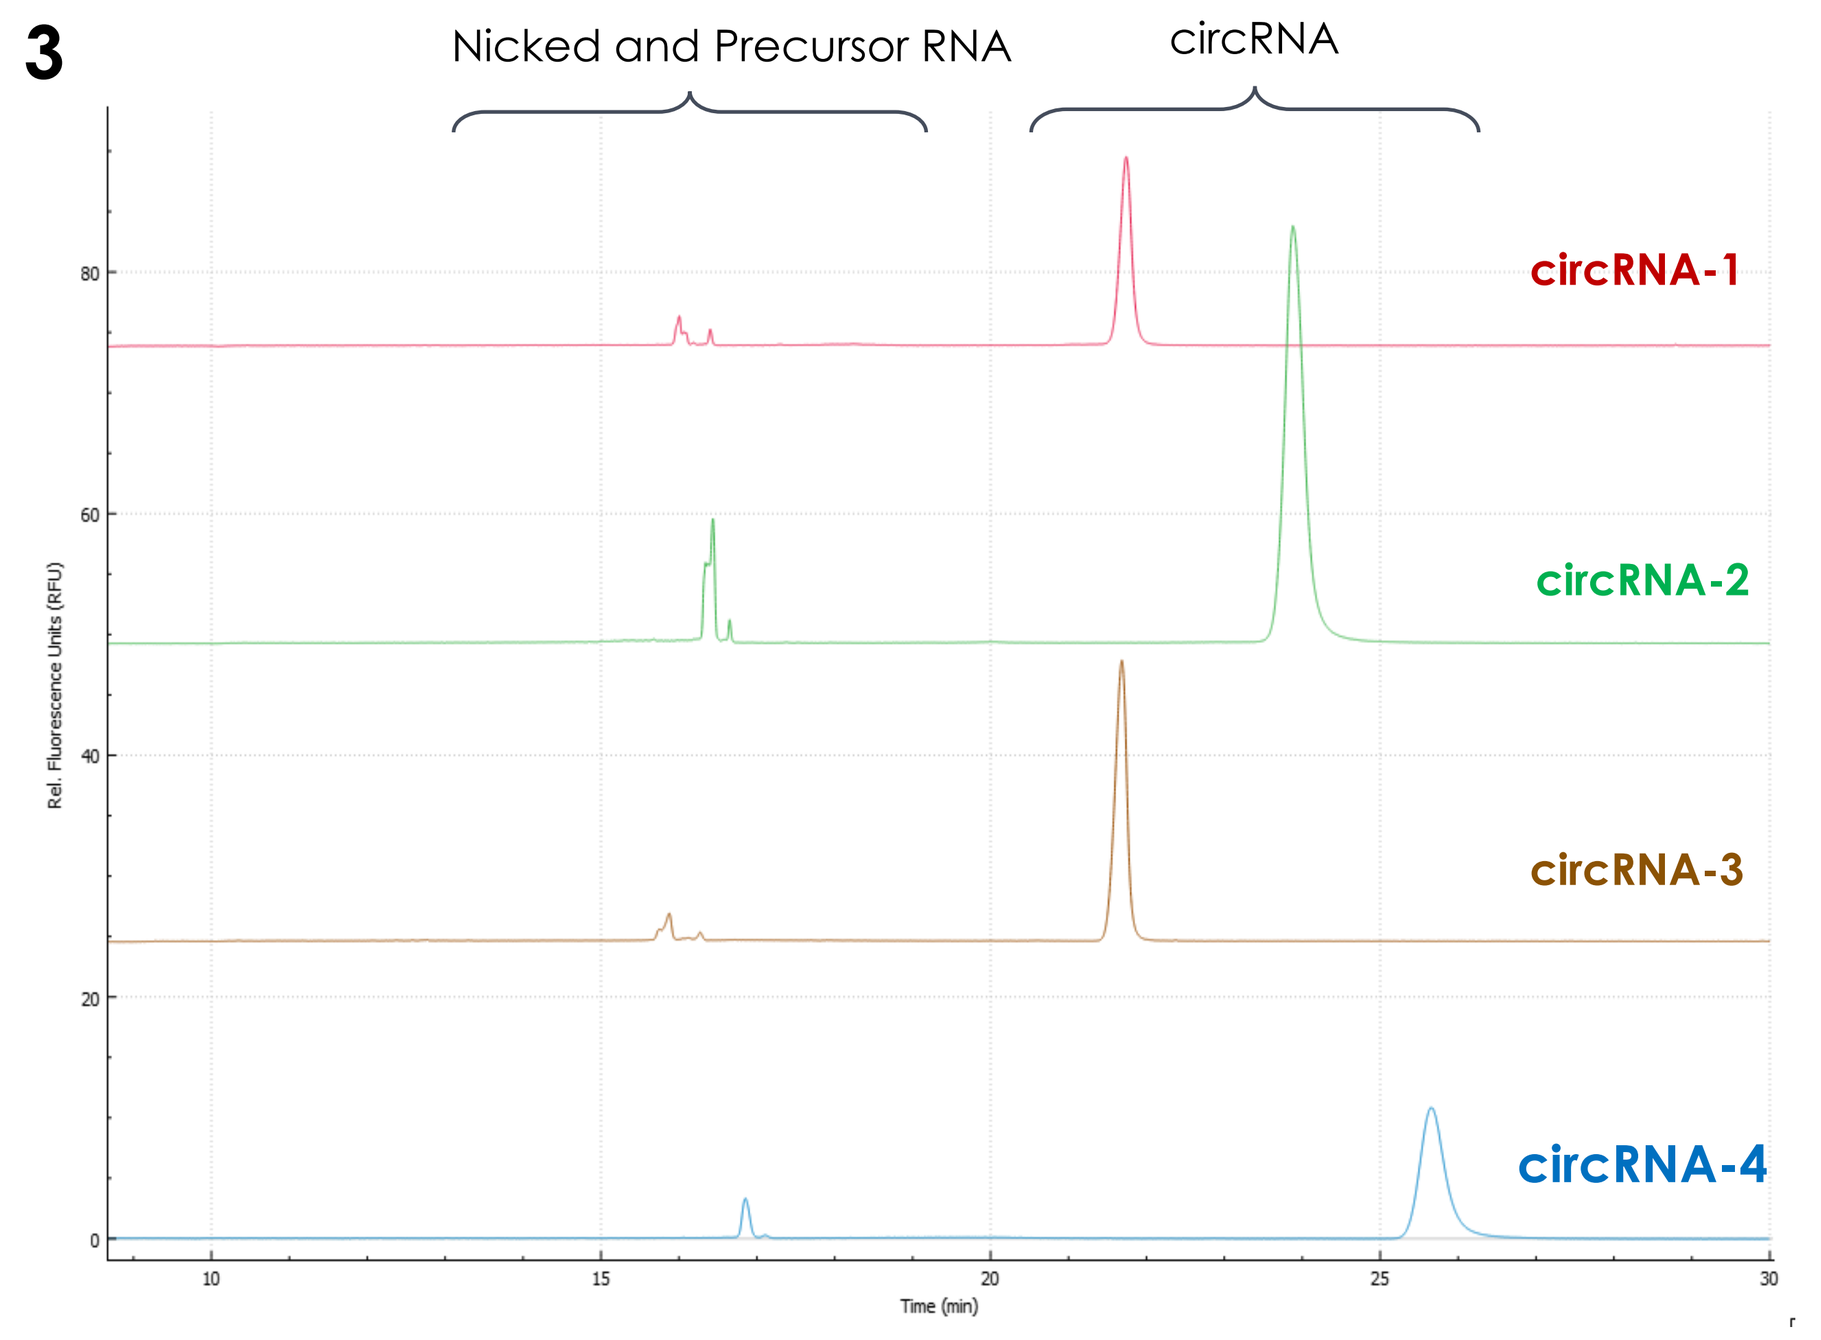
3
Nicked and Precursor RNA
circRNA
circRNA-1
circRNA-2
circRNA-3
circRNA-4

Figure 3: Final Purity of Representative Constructs. The final circRNA products show circRNA purity assessed by CGE analysis. This representative sample set shows variable transgene sizes which affect the elution time of all RNA species.

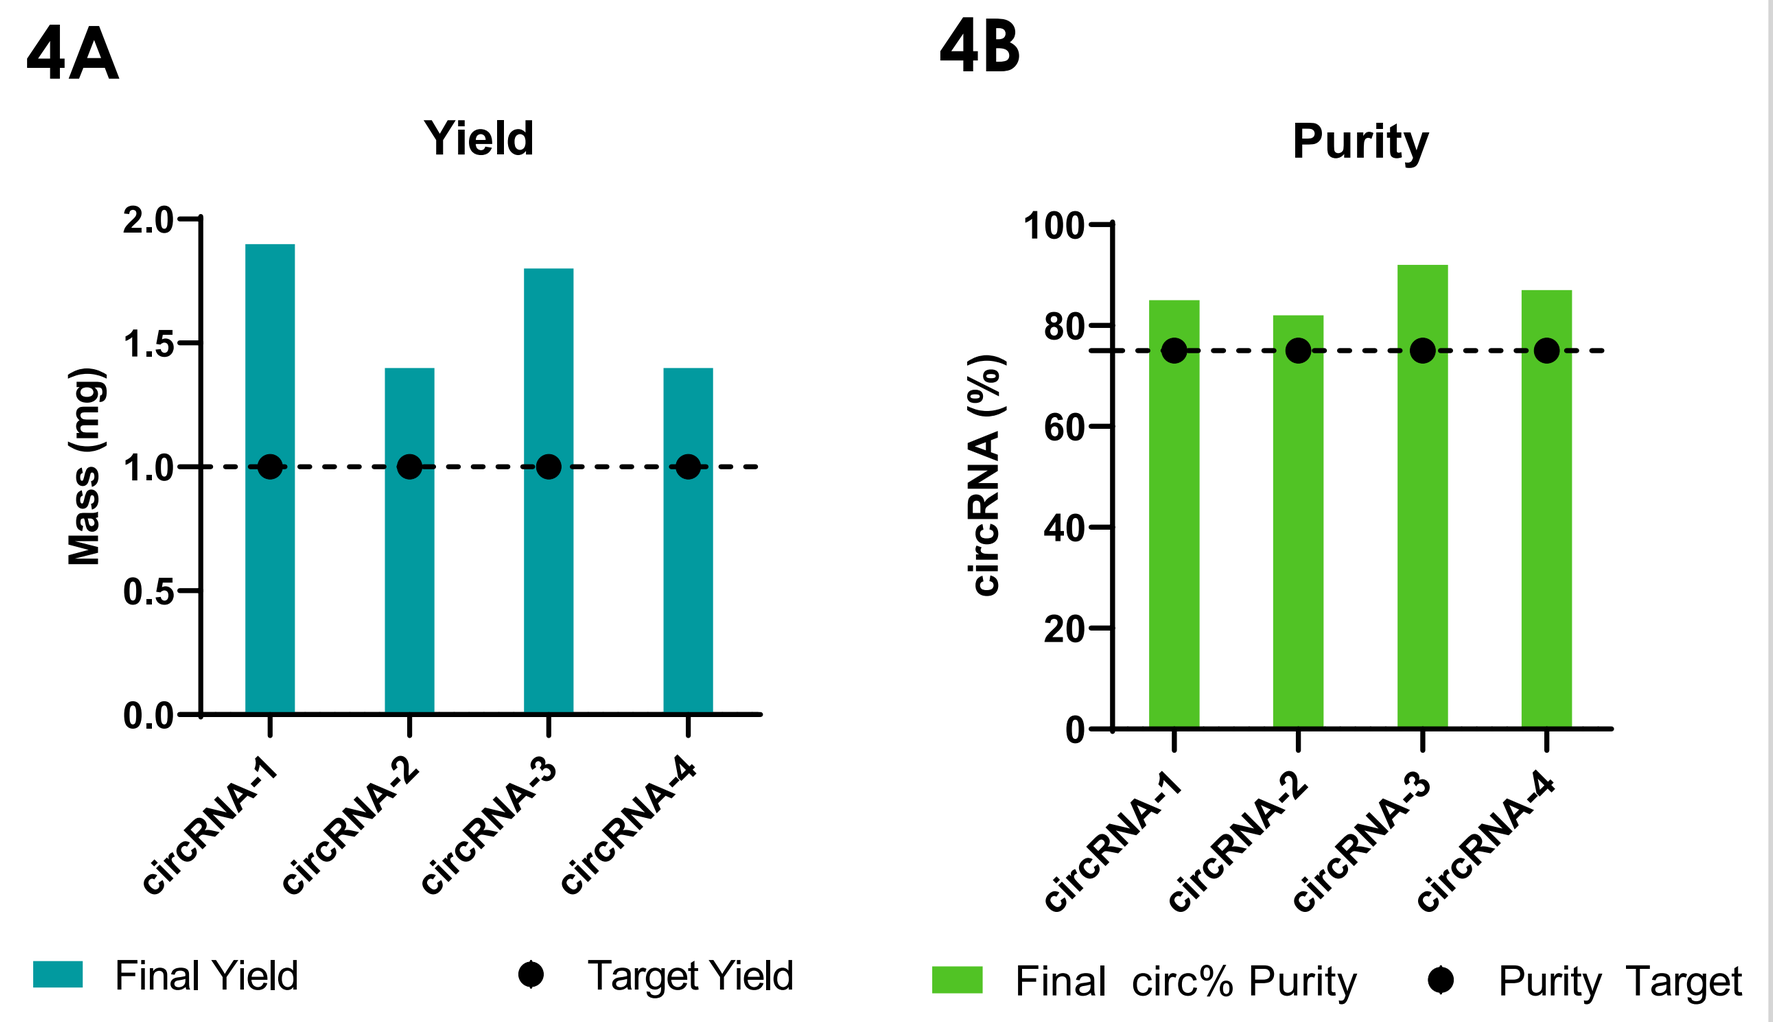
4A
Yield
Mass (mg)
circRNA-1
circRNA-2
circRNA-3
circRNA-4
Final Yield
Target Yield
4B
Purity
circRNA (%)
circRNA-1
circRNA-2
circRNA-3
circRNA-4
Final circ% Purity
Purity Target

Figure 4: Production Metrics. Final circRNA batches meet and exceed target metrics. **4A**. Scaled-up production process results in >1 mg final material. **4B**. Final circRNA purity (as determined by CGE analysis) exceeds 75% cutoff for batch acceptance and downstream use.

Final circRNAs Pass Robust Analytical Panel

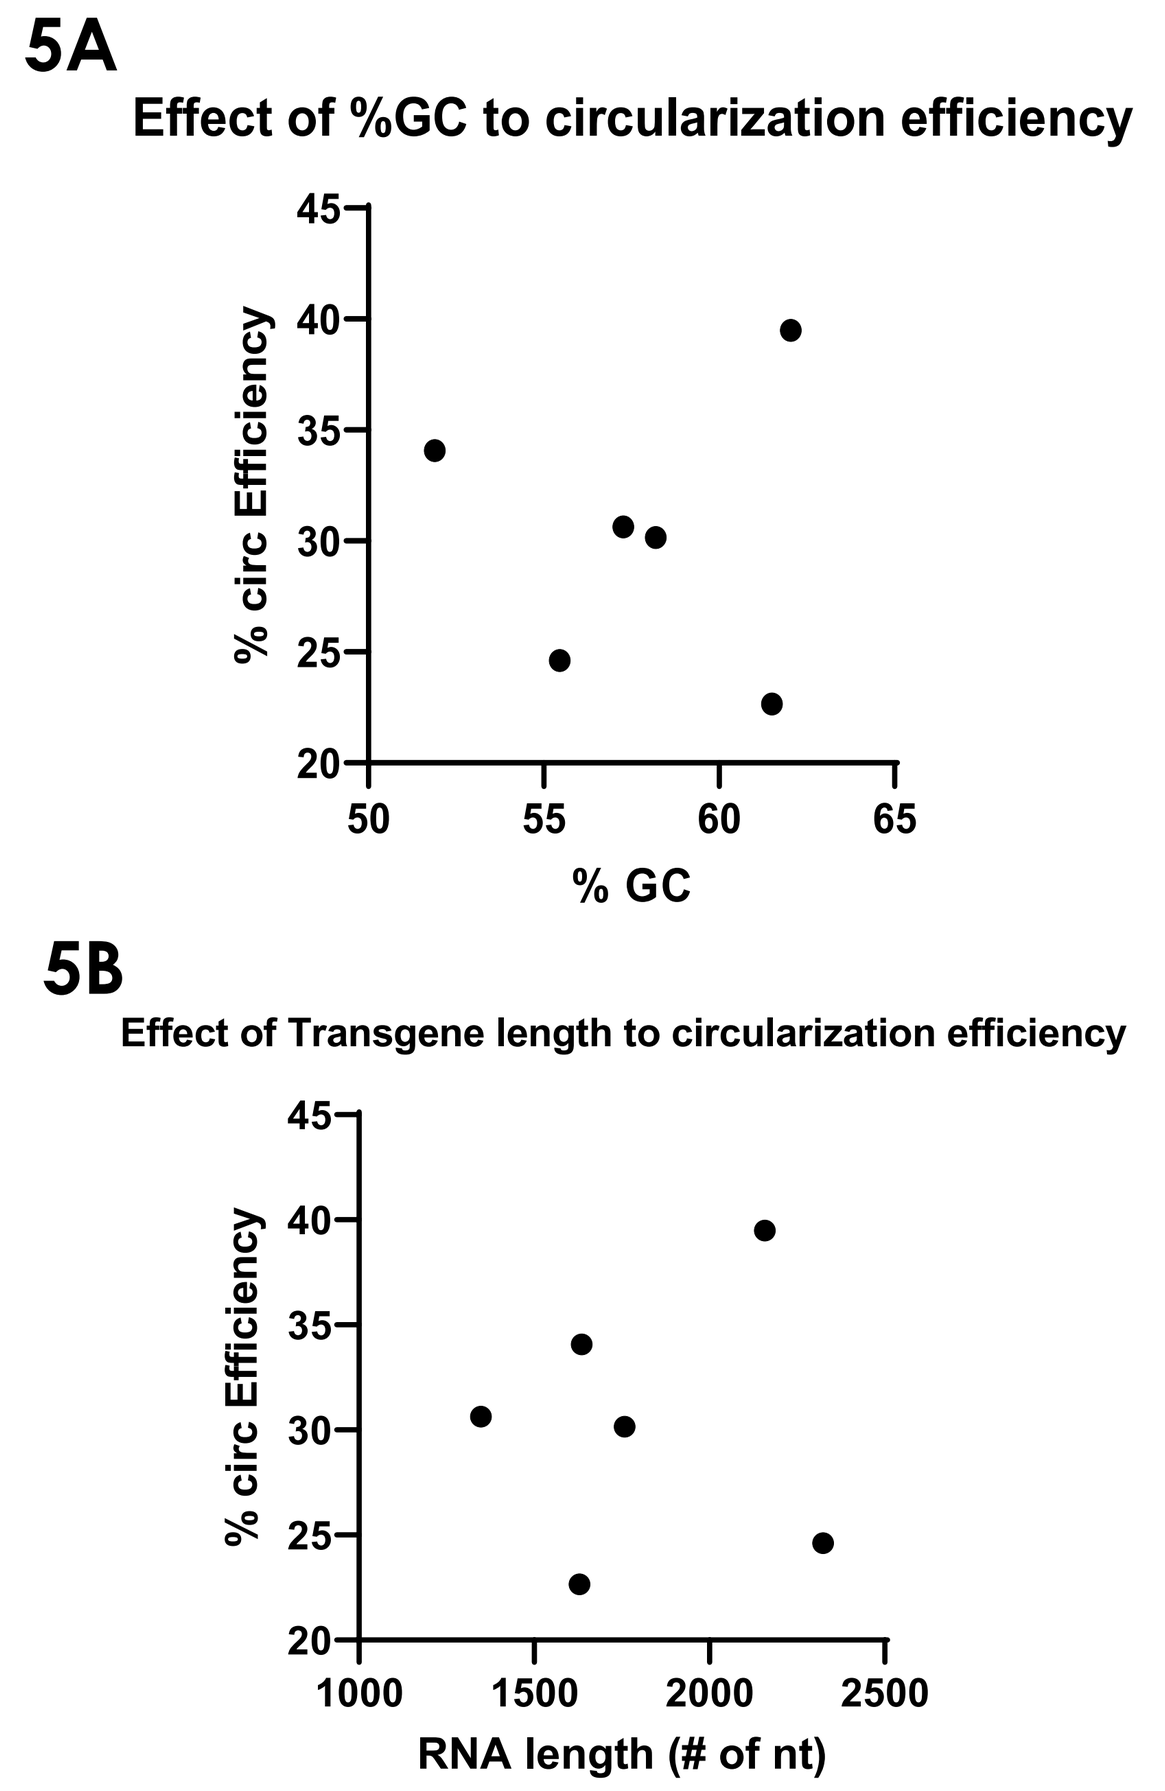
5A
Effect of %GC to circularization efficiency
% circ Efficiency
% GC
5B
Effect of Transgene length to circularization efficiency
% circ Efficiency
RNA length (# of nt)

Figure 5: Impact of %GC Content and Transgene Length on Circularization Efficiency. Circularization efficiency (conversion of precursor intermediate into circRNA) was determined by assessing circRNA purity using CGE immediately after *in vitro* transcription.

Table 1

Assay	Metric	Result
Nanodrop	A260/280	Pass
	A260/230	Pass
	Concentration (mg/mL)	Pass
CGE	circ%	Pass
Next Generation Sequencing	Sequence Identity	Confirmed
Residual Protein - Qubit	ug/mL	Pass
Residual Plasmid - ddPCR	Copies/mL KanR	Pass
Endotoxin - Chromogenic LAL	EU/mL	Pass

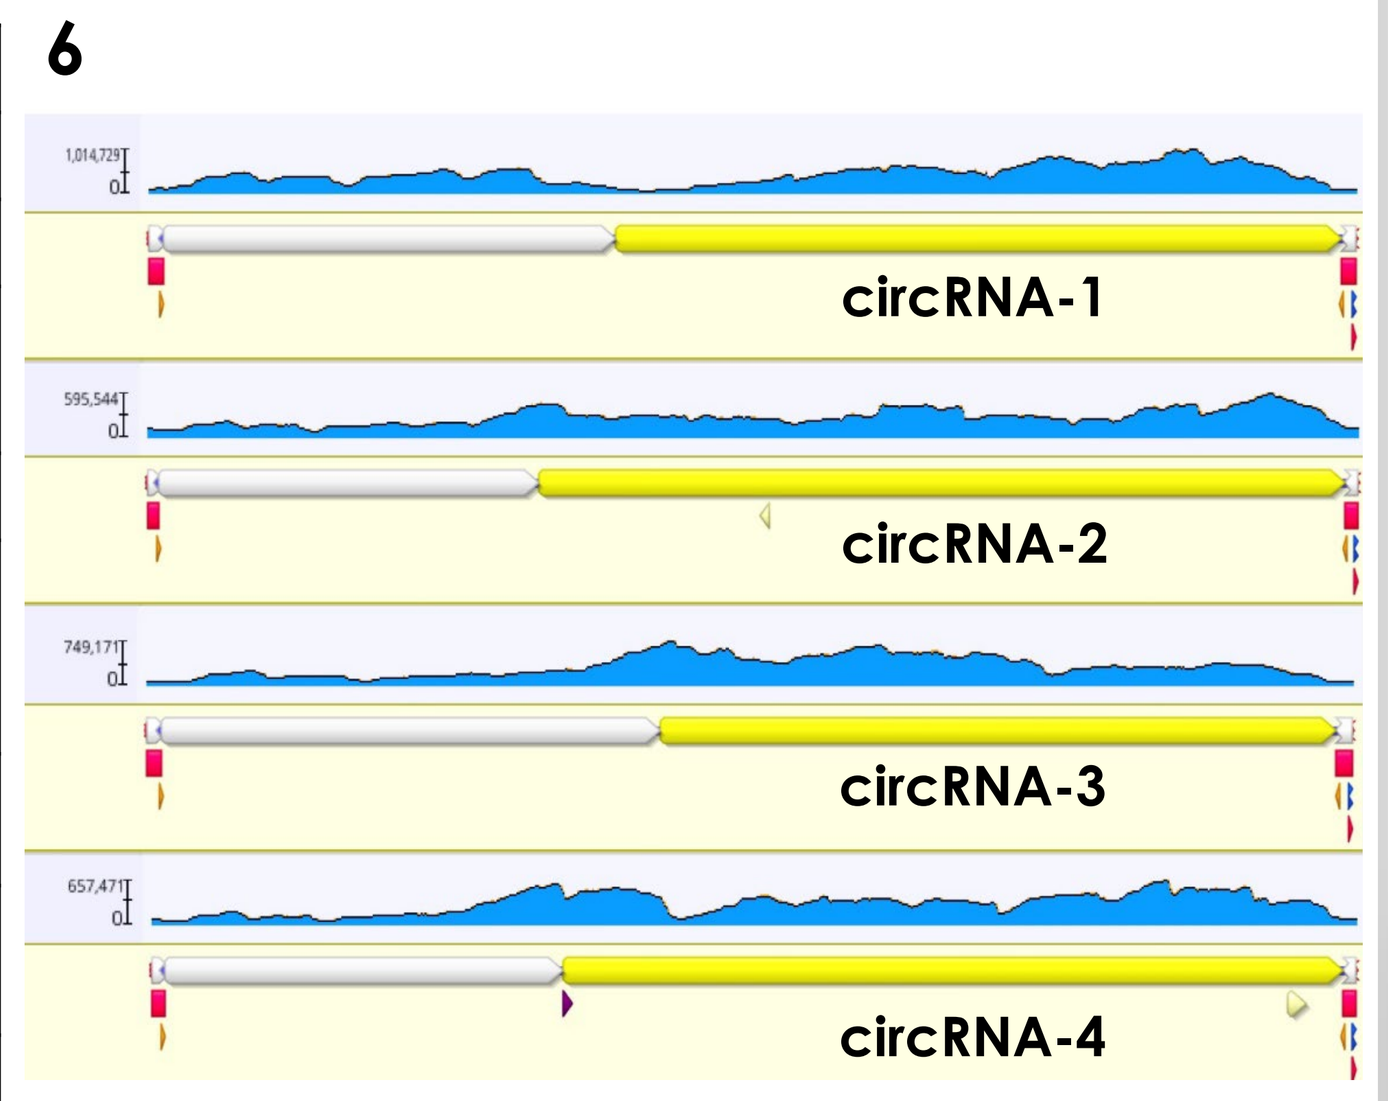
6
circRNA-1
circRNA-2
circRNA-3
circRNA-4

Table 1: Analytical Panel Results. The final circRNA preparations underwent thorough characterization, including assessment of concentration, purity, sequence identity, and presence of contaminants. All materials met the internally defined acceptance criteria before proceeding to downstream functional processes.

Figure 6: Next Generation Sequencing of circRNA Constructs Confirmed Circularization and Sequence identity. Sample read coverage and consensus view is shown mapped to circular reference. All four circRNA samples have coverage over unique GOI relative to circular reference confirming the sequence identity. 100% of established metrics (six pass/fail metrics for the workflow and 11 metrics for sequence identity) were passed for all circRNA samples.

circRNA Outperforms mRNA *In Vitro*

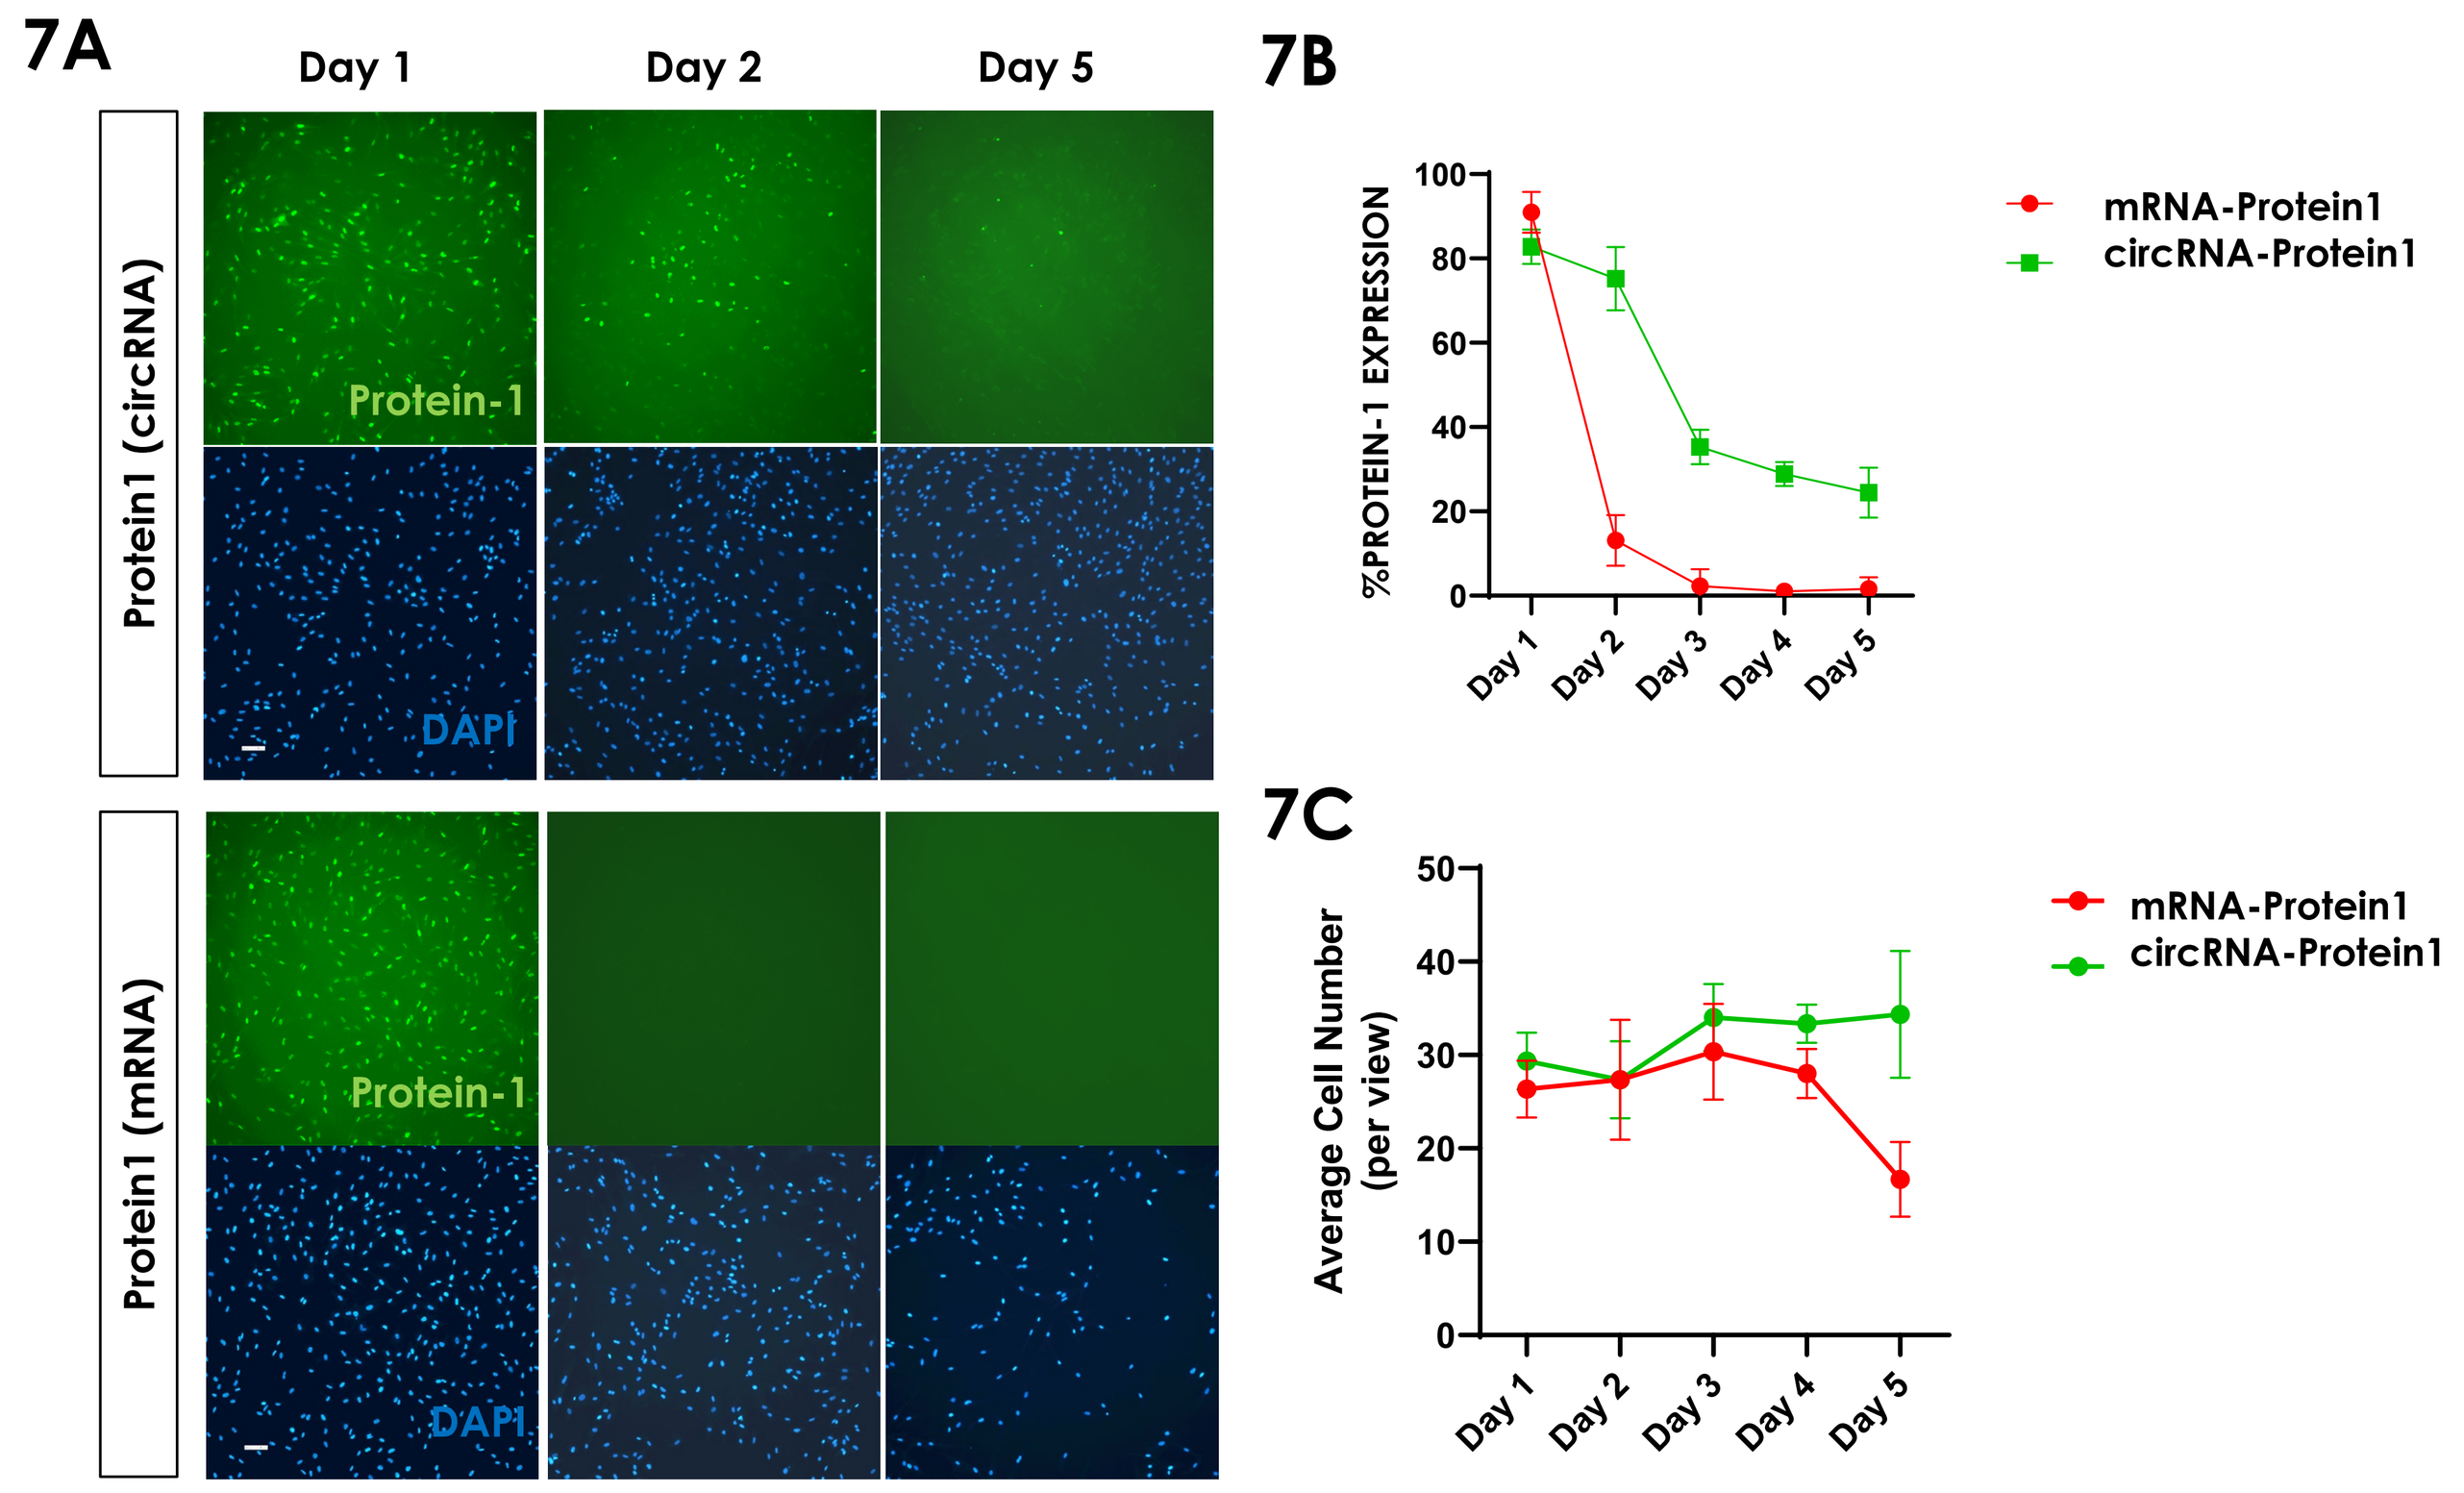
7A
Day 1
Day 2
Day 5
Protein1 (circRNA)
Protein1 (mRNA)
Protein-1
DAPI
7B
%PROTEIN-1 EXPRESSION
mRNA-Protein1
circRNA-Protein1
7C
Average Cell Number (per view)
mRNA-Protein1
circRNA-Protein1

Figure 7: circRNA vs mRNA Expression in Cells. 7A. Fibroblasts were transfected with circRNA or commercial mRNA using RNAiMAX transfection reagent and then fixed and immunostained each day for 5 days to analyze expression of Protein 1 constructs over time (Scale bar = 100 um). **7B.** Quantification of Protein 1 expression using mRNA or circRNA delivery. **7C.** Quantification of cell number over time in response to mRNA or circRNA transfection. Cells were fixed and immunostained for circRNA-1 expression every 24 hours for 5 days.

At Day 1 post-transfection, both circRNA and mRNA treated cells resulted in over 80% expressing cells (Figure 7B). Interestingly, Protein 1 expression was sustained with over 60% of cells expressing by Day 2 with circRNA, while mRNA was almost undetectable. Protein 1 expression sustained a higher expression from circRNA than mRNA up to Day 5. Further, mRNA transfection caused a decrease in cell number by Day 5, suggesting that mRNA may be cytotoxic to cells over time (Figure 7C).

CONCLUSION

ElevateBio has developed an efficient, scaled-up process for producing high purity circRNA with functional *in vitro* expression. Our proprietary Permuted Intron-Exon (PIE) system allows for a circularizing backbone into which different IRES and transgene elements can be inserted. RNA is transcribed, circularized, and enriched through a combination of separation chromatography and enzymatic cleanup steps.

- We developed a **comprehensive analytical panel** to standardize acceptance criteria for samples before downstream functional testing.
- Our circRNAs demonstrate **low cytotoxicity and stable, long-lasting expression** compared to mRNA.
- We are continuing to develop in-house analytics for assessing the **dsRNA content and immunogenicity** of our final circRNA product, which will inform both our design and process optimization efforts.
- Our modular circRNA platform allows for **further screening of IRES and transgene targets** relevant to a wide range of therapeutic applications.
- Further functional cell-based testing is also in progress.

Contact Info: Tess Kitchener, ElevateBio, 914-471-1931, tkitchener@elevate.bio