Early Characterization of Research-Grade AAV6 Productions Allows for Manufacturing Alignment During Candidate Nomination

ElevateBio®, 225 Wyman Street, Waltham MA 02451, U.S.

Amira Rghei, Omar Matalka, Lisa A. Santry, Shona Robinson, James Fasano, Jai Cai, Daniel Kennedy, Theresa Dao, Azam Hassaninasab, Will Arnold, Gary Sommerville, Adriana Geldart, Debaditya Bhattacharya, Mercedes Segura, Bojiao Yin, Stacie L Seidel, Cherylene Plewa

Abstract

Adeno-associated virus (AAV) is the preferred viral vector in gene therapy, with applications to treat and potentially cure diseases caused by single-gene mutations. The recombinant AAV field has seen significant progress, with numerous active clinical trials and several of these therapies approved by regulatory authorities like the EMA and FDA. However, accurately characterizing AAV vectors, especially in terms of packaging efficiency and scalability for production, continues to be a significant hurdle prior to pre-clinical work. This challenge becomes more pronounced during the scale-up process from research and development (R&D) to process development (PD), requiring extensive and costly efforts in optimizing manufacturing, purification, and post-purification procedures. Unfortunately, the properties of the scaled-up PD product may not align with those of the initial R&D material, leading to prolonged and tedious pre-clinical development phases to reconcile these differences. There is a clear need for early and well-defined characterization of therapeutic candidates at the R&D stage that aligns, when possible, with production, purification, and characterization methods across both upstream and downstream PD platforms.

Many components that influence successful manufacturing and scaling of AAV include, but are not limited to vector design, plasmid composition including the genome size and sequence, capsid selection, production methods (including transient transfections), and purification techniques, such as chromatography and ultracentrifugation. Manufacturing standards, including GMP considerations and FDA standards, such as host cell and plasmid DNA levels, and amount of empty capsids, also need to be considered.

Here, we outline a process to streamline the selection of lead therapeutic candidates for R&D AAV6 vectors, using a high throughput approach prior to initiating scale-up activities. Five AAV6 vectors expressing therapeutically relevant transgenes, with genomes ranging in size from 2.8-4.7 kb, were packaged into AAV6 using our R&D process (shaker flask production, resin-based chromatography), or our scaled-up PD process (2L bioreactor, purified by resin-based chromatography using an AKTA - high performance liquid chromatography, pre-enriched intermediate material). Both sets of AAV6 vectors were characterized through a series of analytics, including vector genome titering (ddPCR), viral protein ratios (Maurice, automated capillary electrophoresis system), % full analysis (Mass Photometry for both, and analytical ultracentrifugation for PD material), followed by Oxford Nanopore Technology (ONT) 3rd generation sequencing.

Our results show that our R&D AAV6 vectors had crude titers >1e11 vector genomes (vg)/mL, as well as >20% full by Mass Photometry, similar to the readout of the AAV6 PD vectors. Additionally, our ONT 3rd generation sequencing analysis shows similar frequency of read alignments to AAV genome) between R&D and PD material. This early characterization of AAV6 candidates allows for earlier and more efficient testing of multiple construct designs, and cost-efficient movement of manufacturable lead therapeutic candidates from R&D to PD.

Production & Analytical Methods

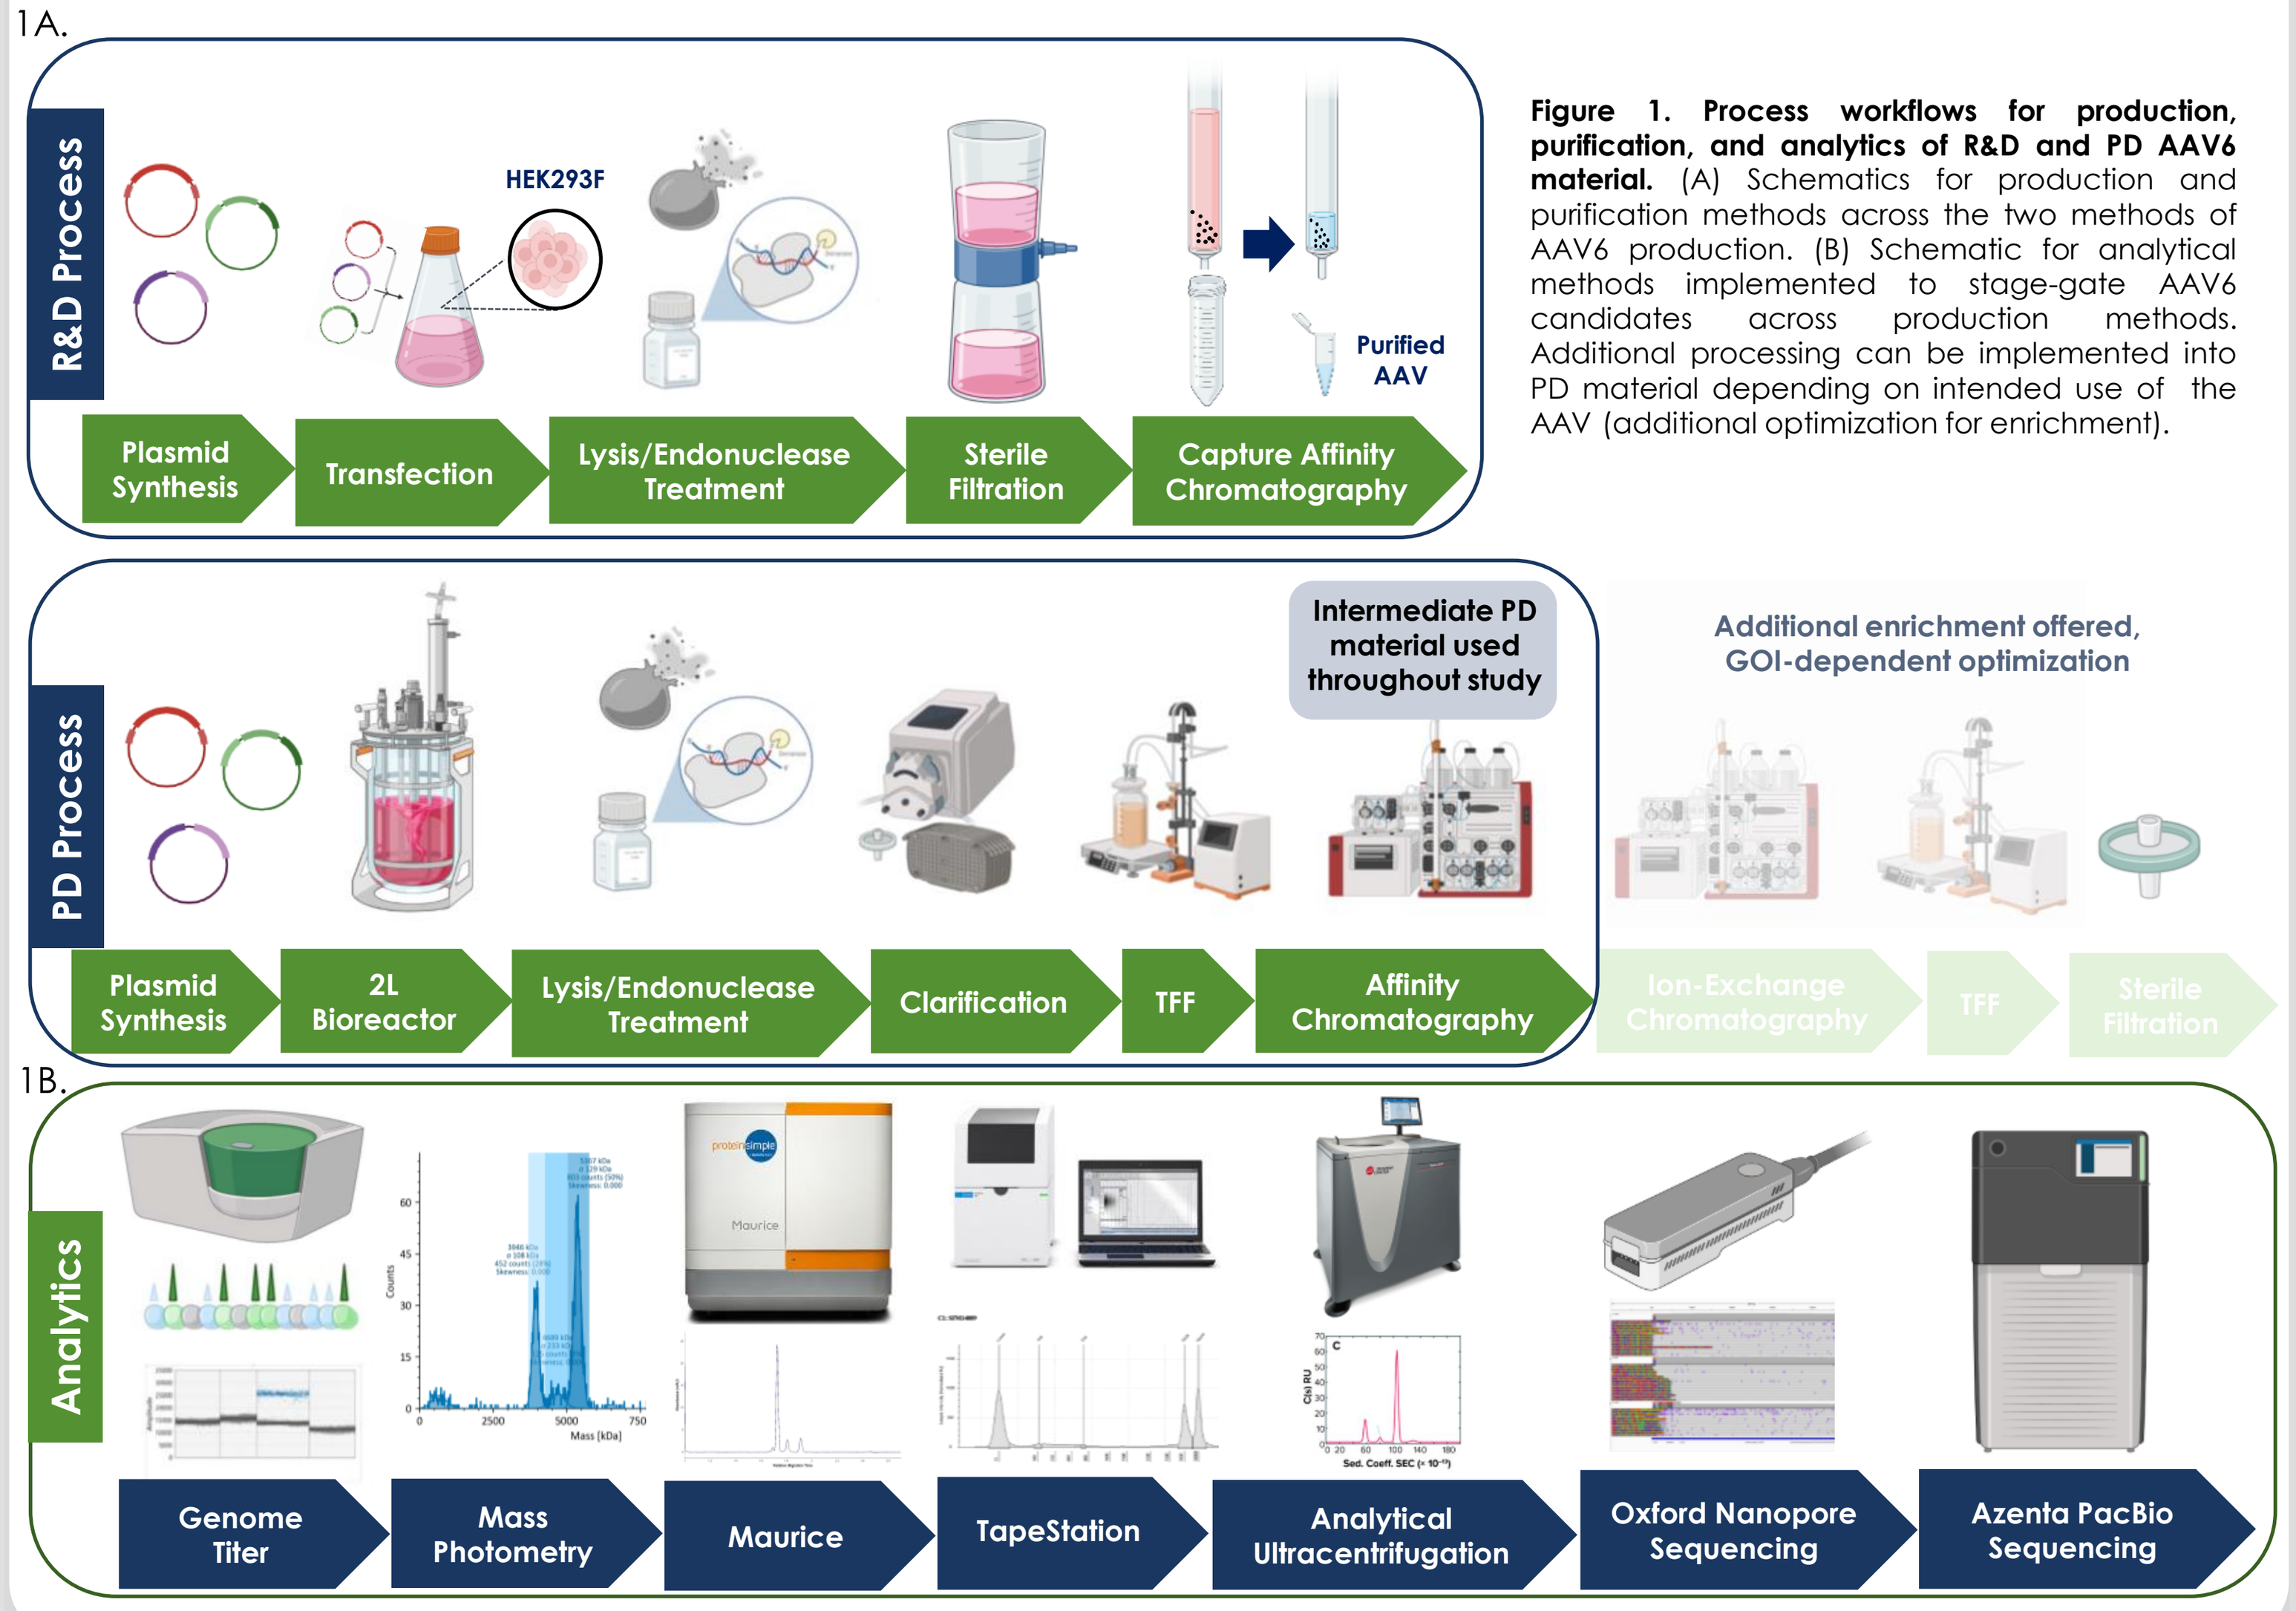
1A.
R&D Process
HEK293F
Purified AAV
Plasmid Synthesis
Transfection
Lysis/Endonuclease Treatment
Sterile Filtration
Capture Affinity Chromatography
PD Process
Intermediate PD material used throughout study
Additional enrichment offered, GOI-dependent optimization
Plasmid Synthesis
2L Bioreactor
Lysis/Endonuclease Treatment
Clarification
TFF
Affinity Chromatography
Ion-Exchange Chromatography
TFF
Sterile Filtration
1B.
Analytics
Genome Titer
Mass Photometry
Maurice
TapeStation
Analytical Ultracentrifugation
Oxford Nanopore Sequencing
Azenta PacBio Sequencing

Figure 1. Process workflows for production, purification, and analytics of R&D and PD AAV6 material. (A) Schematics for production and purification methods across the two methods of AAV6 production. (B) Schematic for analytical methods implemented to stage-gate AAV6 candidates across production methods. Additional processing can be implemented into PD material depending on intended use of the AAV (additional optimization for enrichment).

Results

Similar AAV Genome Titers Can Be Seen In R&D and PD Produced AAV6

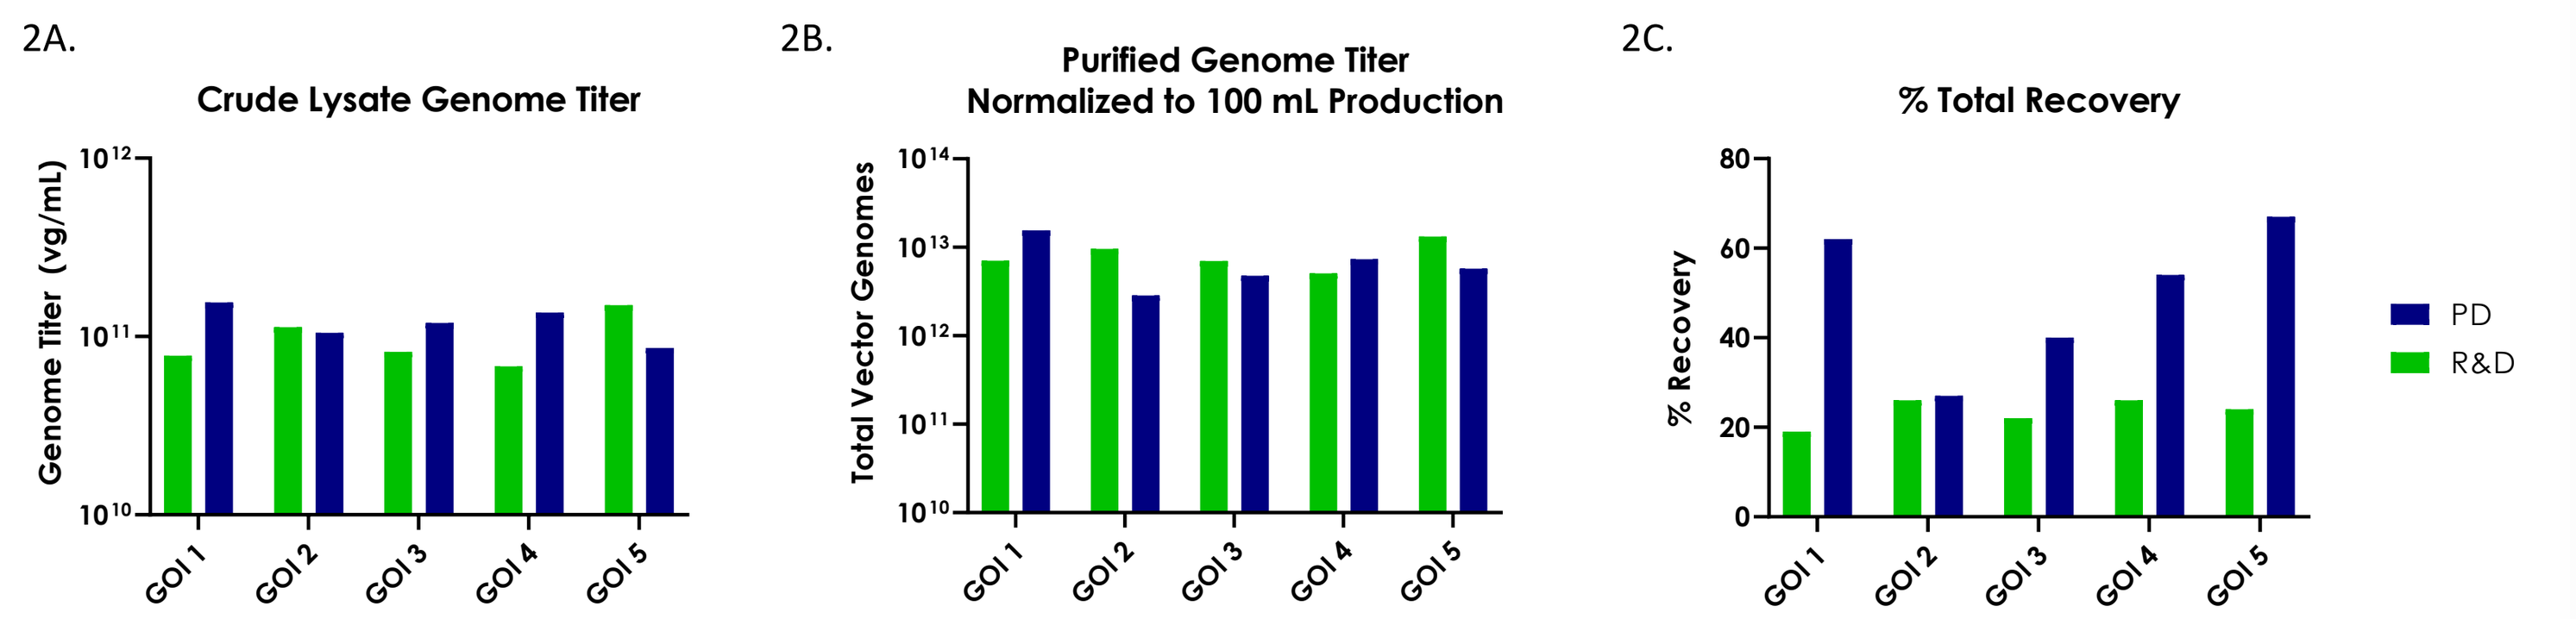
2A.
Crude Lysate Genome Titer
Genome Titer (vg/mL)
2B.
Purified Genome Titer
Normalized to 100 mL Production
Total Vector Genomes
2C.
% Total Recovery
% Recovery
PD
R&D
GOI 1
GOI 2
GOI 3
GOI 4
GOI 5

Figure 2. Genome titer comparisons between R&D and PD AAV6 material. (A) Shaker flasks (R&D material) and 2L bioreactor PD intermediate material containing serum-free suspension cells were transfected using a triple plasmid system. Crude lysates were harvested and genome titers, vector genomes (vg)/mL, were quantified using ddPCR (Bio-Rad). (B) R&D and PD crude lysates was purified with either column-based capture affinity chromatography (R&D) or AKTA (FPLC), and ddPCR was used to determine vg/mL. Genome titers were normalized to 100 mL loading volume. (C) Recovery of AAV material for AAV6 products expressed as a percentage of total vg remaining after purification.

Mass Photometry Highlights Similar Percentage Of Full AAV Virion

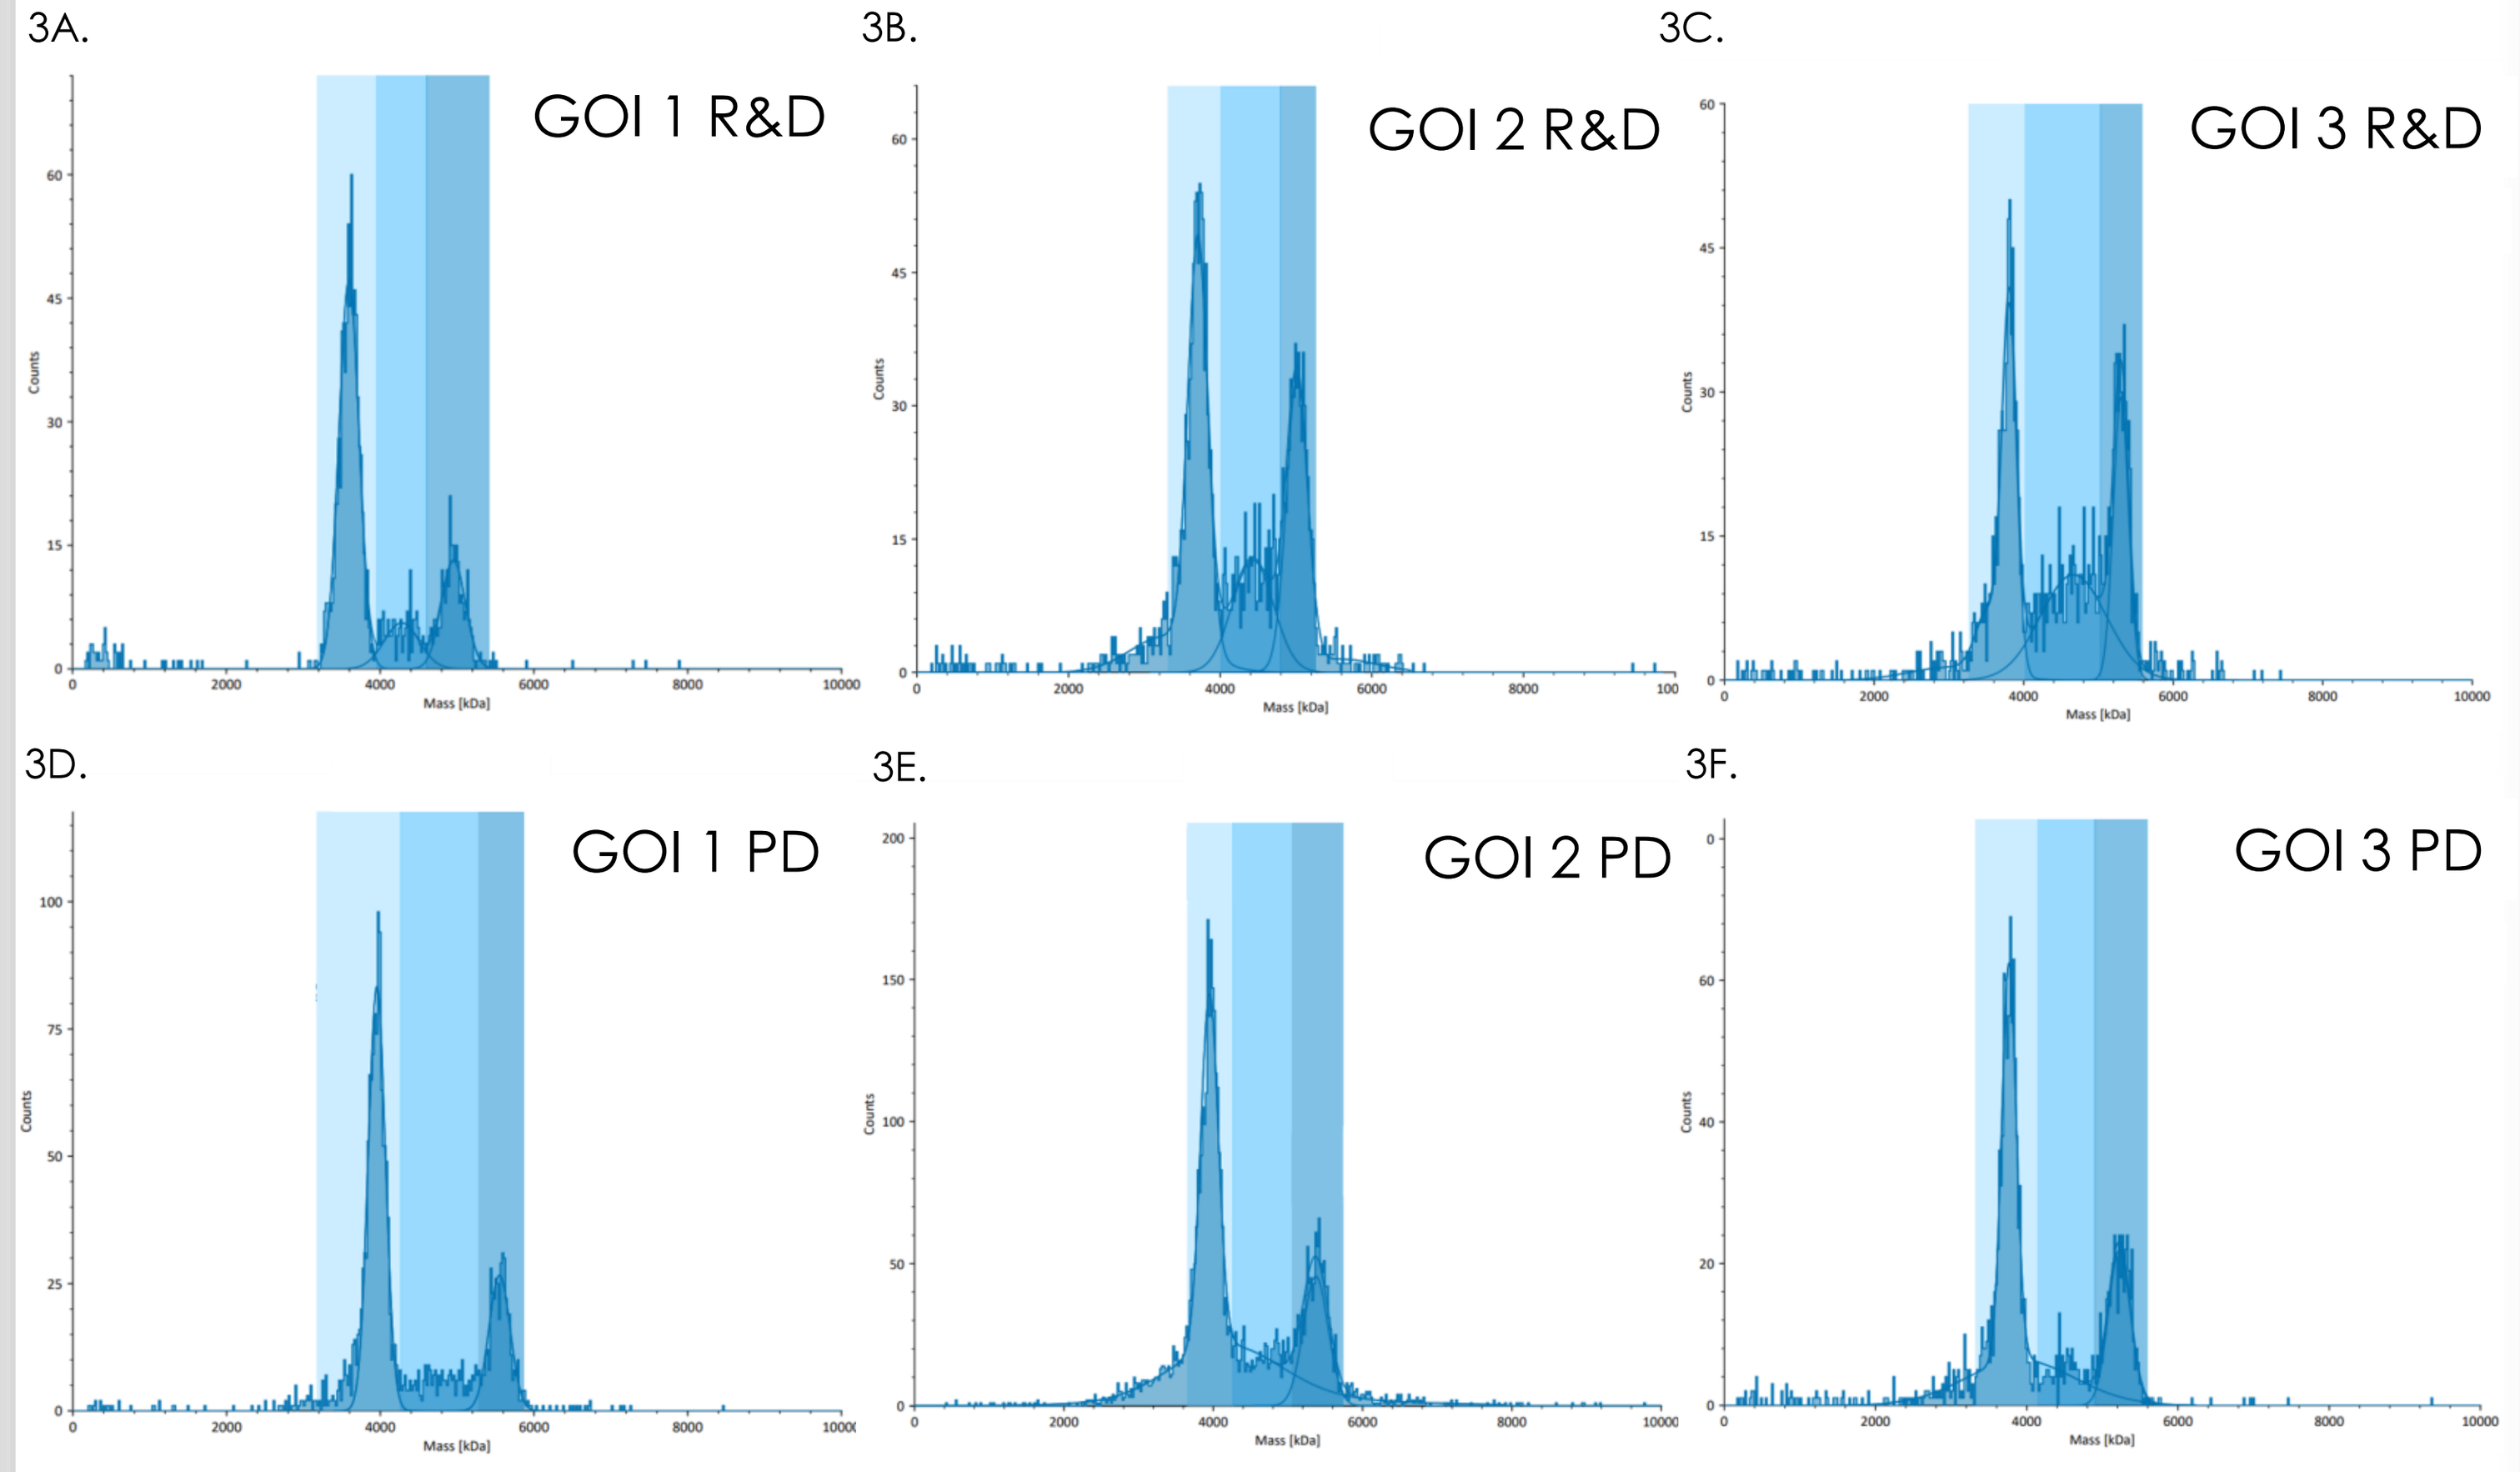
3A.
GOI 1 R&D
3B.
GOI 2 R&D
3C.
GOI 3 R&D
3D.
GOI 1 PD
3E.
GOI 2 PD
3F.
GOI 3 PD
Mass [kDa]

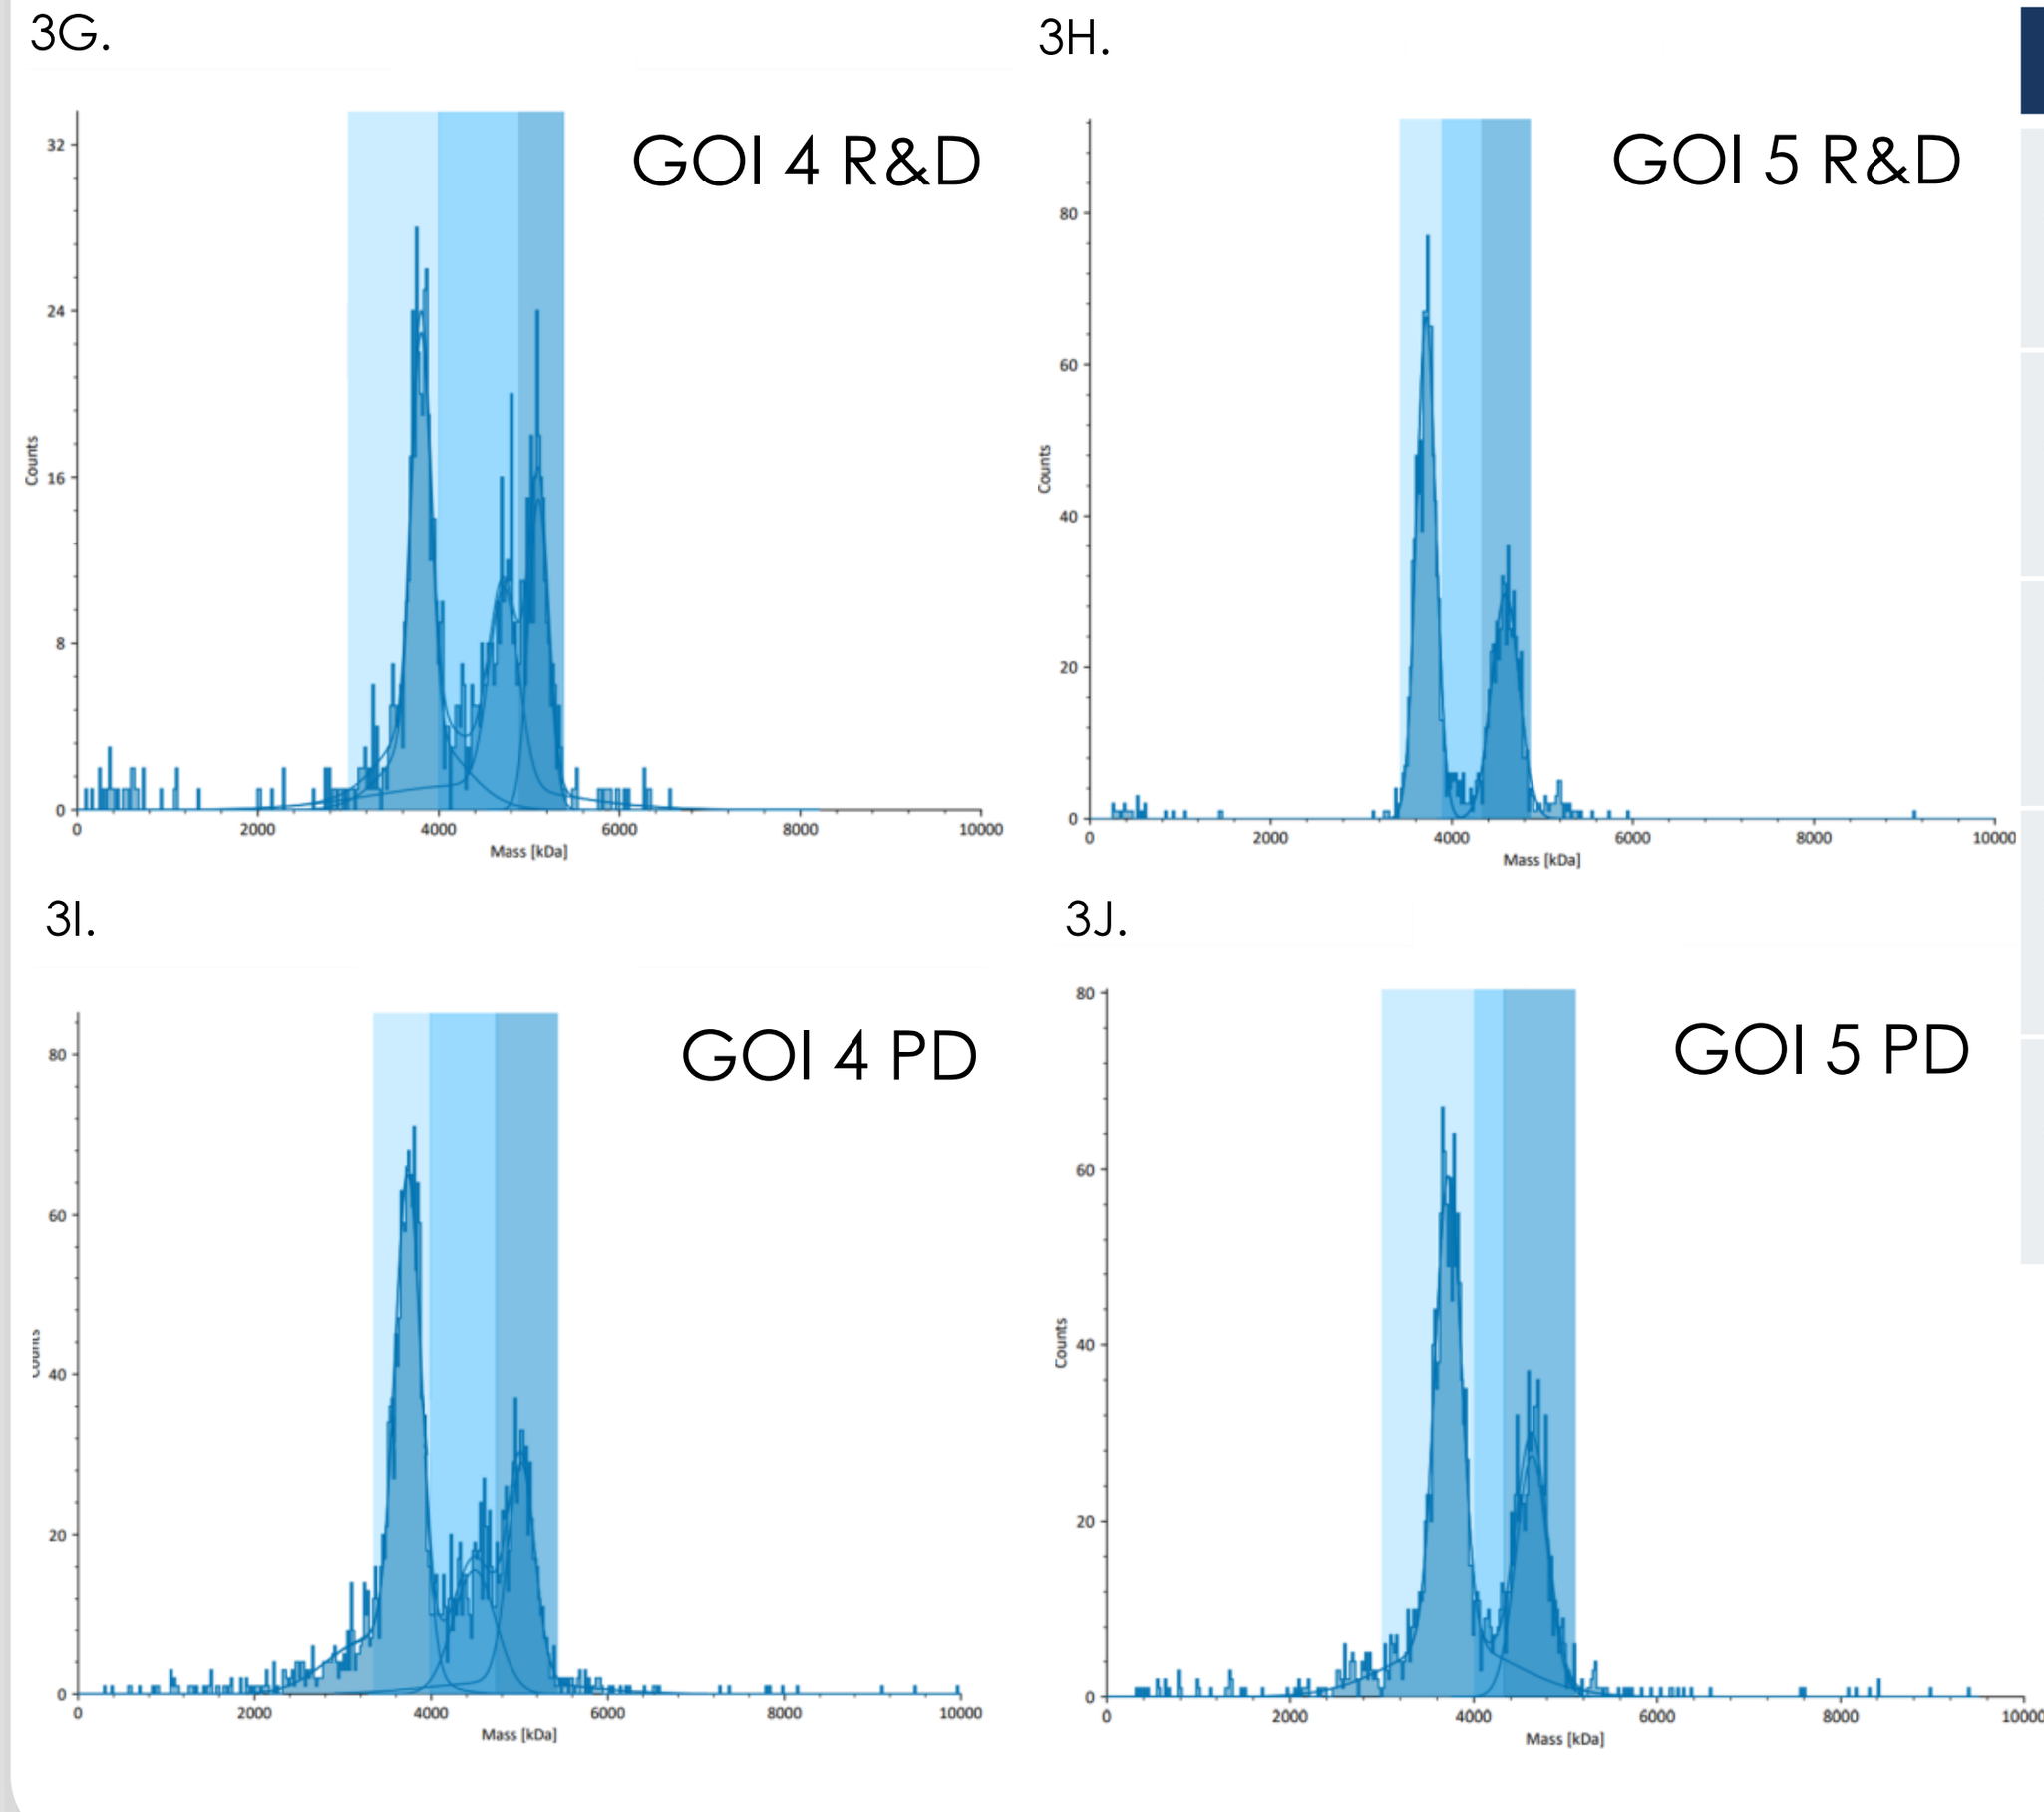
3G.
GOI 4 R&D
3H.
GOI 5 R&D
3I.
GOI 4 PD
3J.
GOI 5 PD
Mass [kDa]

	Process	Empty (%)	Partial (%)	Full (%)	Variance for % Full
GOI 1	R&D	65	12.6	22.4	0.7
	PD	63.8	14.5	21.7	
GOI 2	R&D	46.4	23	30.6	4.0
	PD	57	16.4	26.6	
GOI 3	R&D	39.3	29.2	31.5	3.5
	PD	60.4	11.6	28	
GOI 4	R&D	42.5	31.1	26.4	0.5
	PD	52.2	21.9	25.9	
GOI 5	R&D	57.8	6.9	35.3	2.1
	PD	59.6	7.2	33.2	

Figure 3. Mass distribution of AAV6 material. R&D and PD intermediate AAV6 material were analyzed using the SamuxMP Mass Photometer (Refeyn) to quantify empty, partial and full virions. A single standard molecular species is set to 3700 kDa as a measure of empty AAV6 particles.

Viral Protein Analysis Reveals Minimal Difference In Protein Ratios

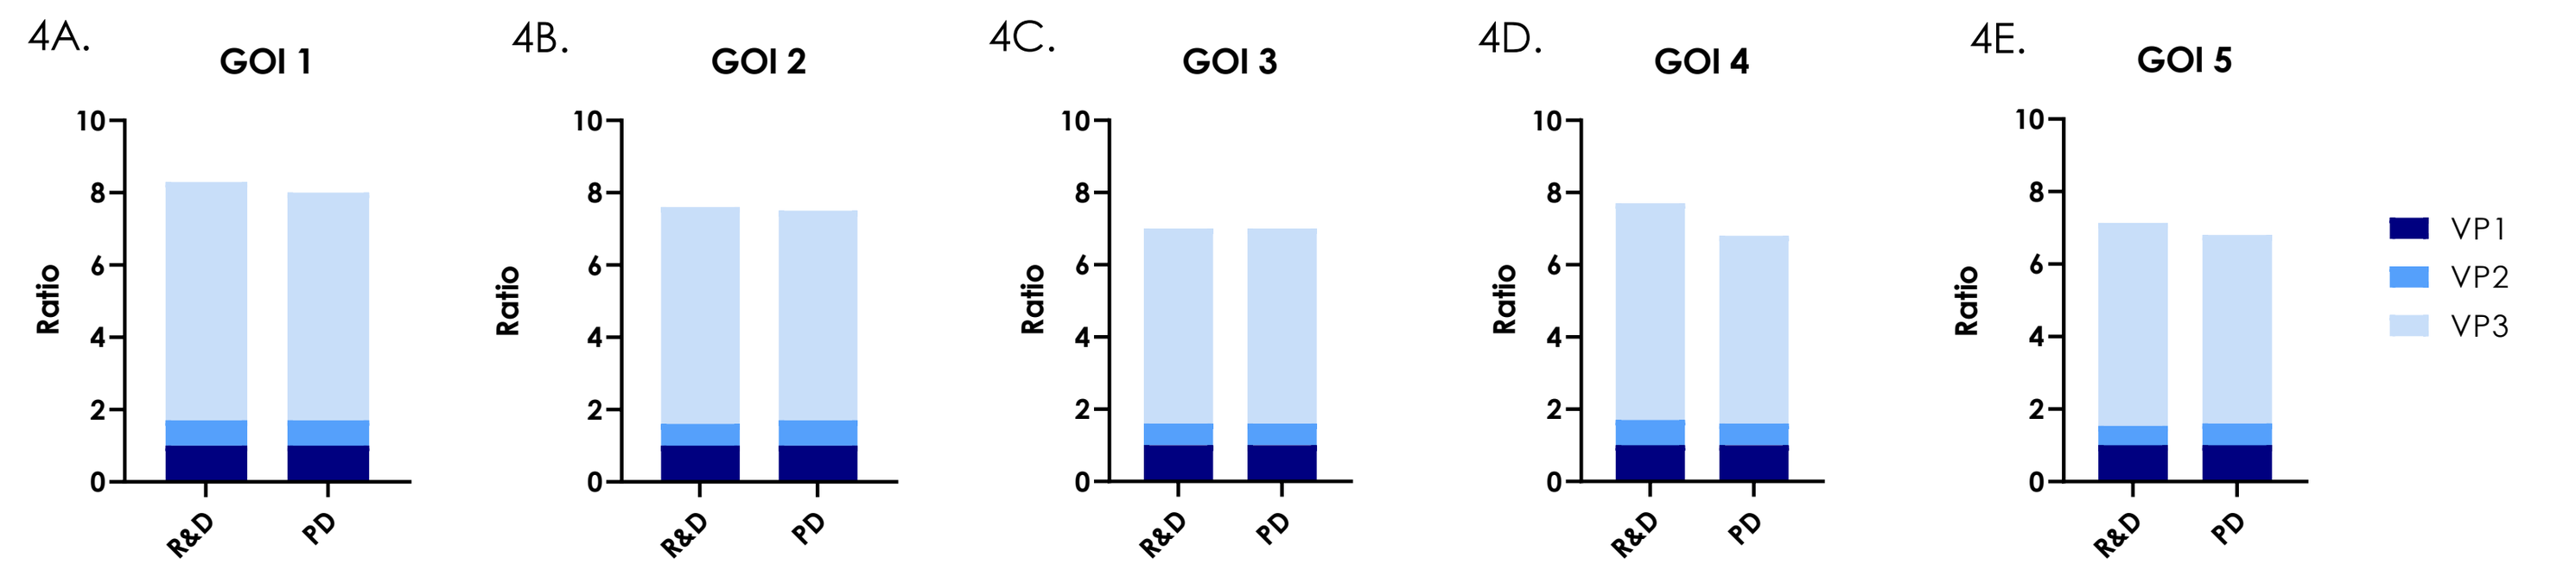
4A.
GOI 1
4B.
GOI 2
4C.
GOI 3
4D.
GOI 4
4E.
GOI 5
Ratio
R&D
PD
VP1
VP2
VP3

Figure 4. Viral capsid protein ratios of AAV6. Viral protein ratios were determined by Maurice (ProteinSimple), an automated SDS-PAGE. All VP proteins are normalized to VP1 (1).

DNA Fragment Analysis Shows Similar DNA Populations

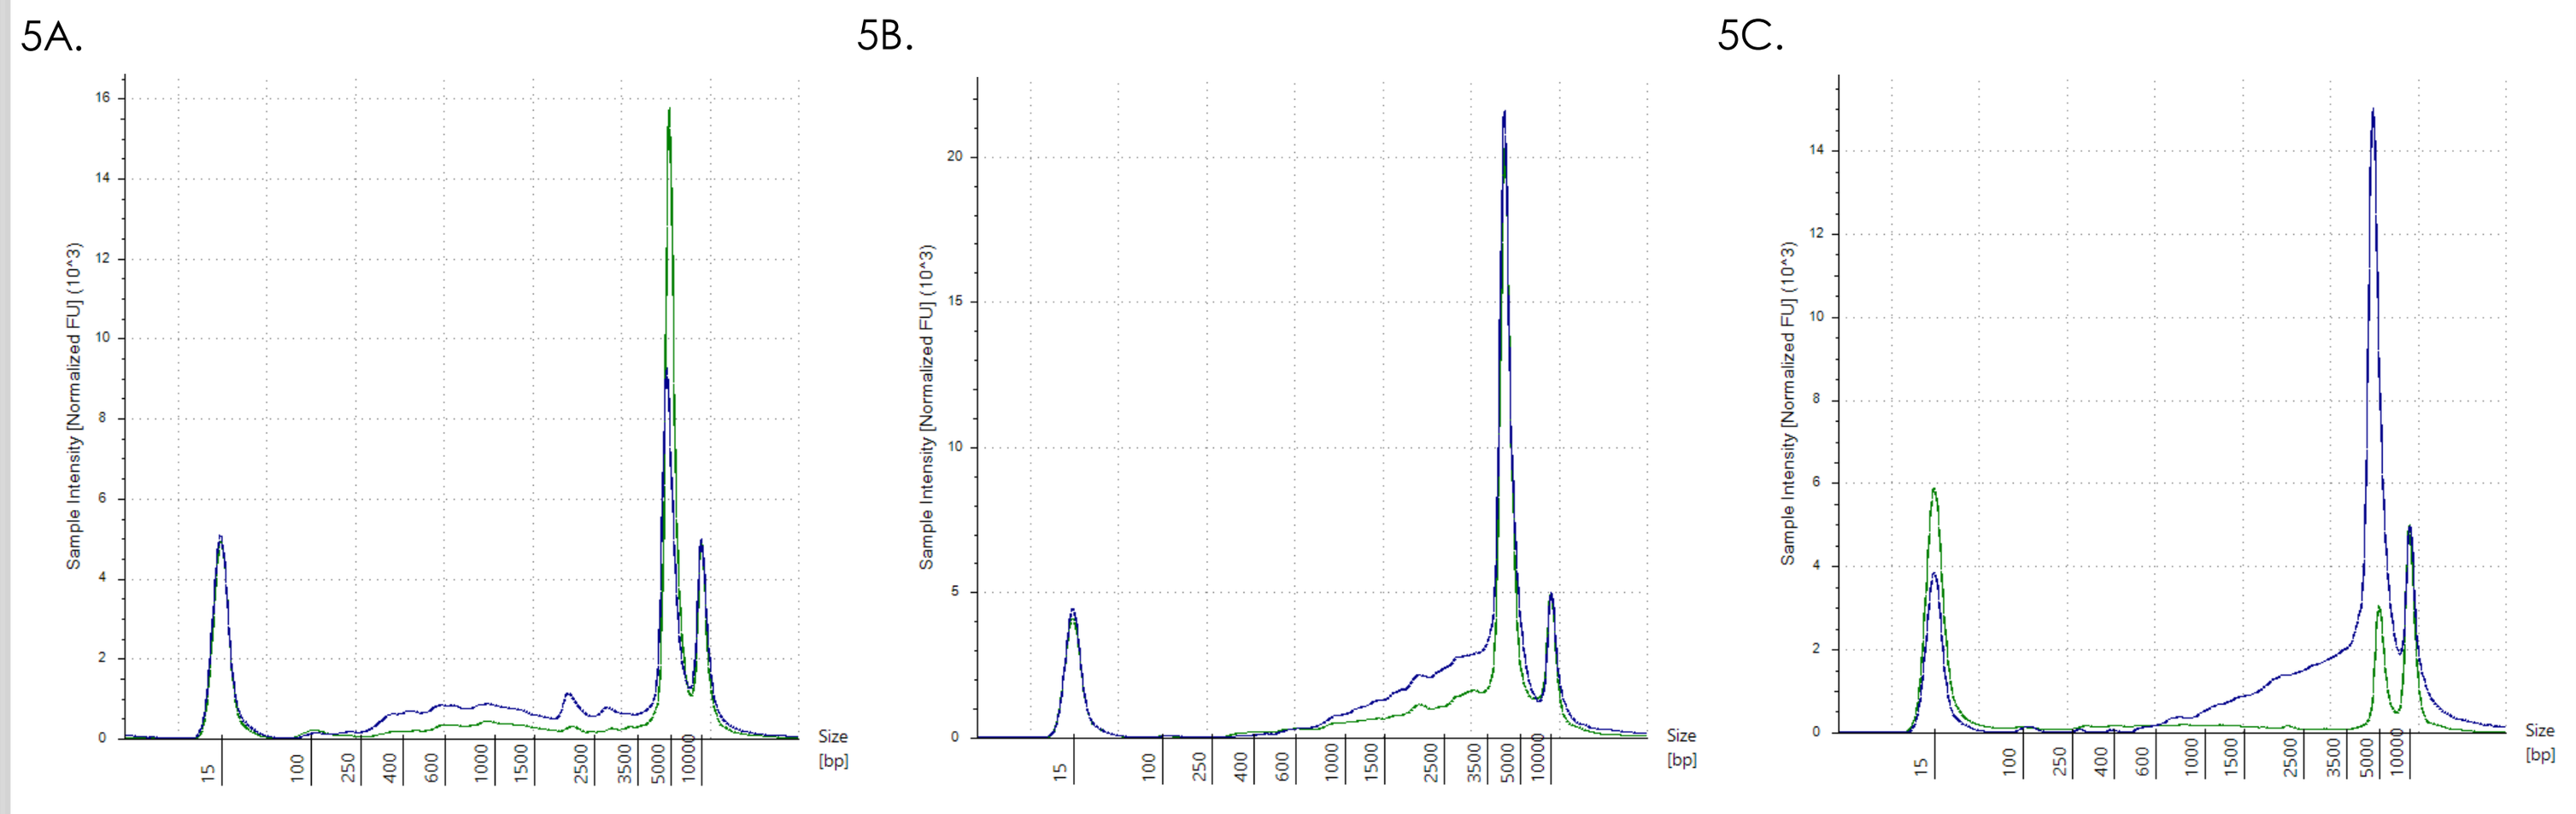
5A.
5B.
5C.
Size [bp]

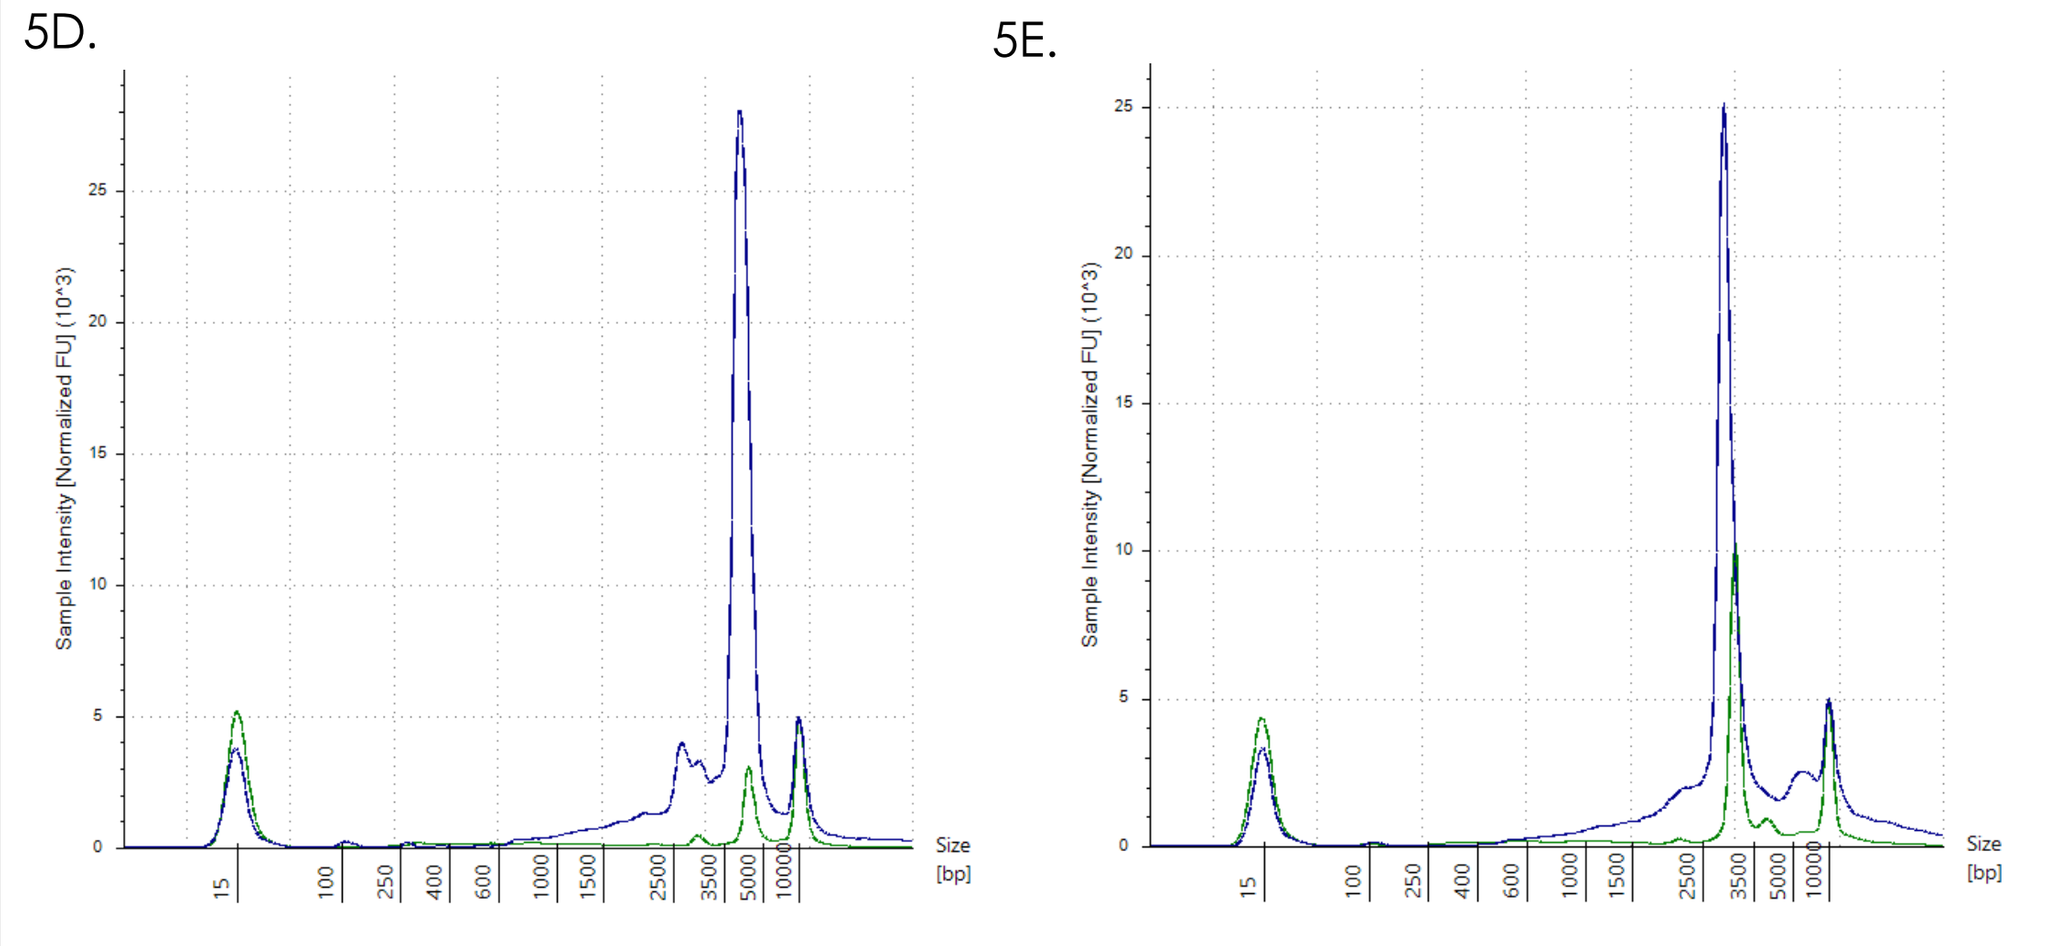
5D.
5E.
Size [bp]

Figure 5. Electropherogram patterns of DNA isolated for AAV virions. R&D and PD intermediate AAV6 material underwent a Dnasel treatment, Proteinase K treatment, and clean-up prior to DNA fragment analysis to visualize packaged DNA only. (A-E) 50 µL of purified vector was used as input for each sample for DNA extraction; 1 µL of extraction was run on Agilent TapeStation 4200, D5000 screentape. R&D material is denoted in green, and PD material is denoted in blue.

Analytical Ultracentrifugation Aligns With Mass Photometry Analysis

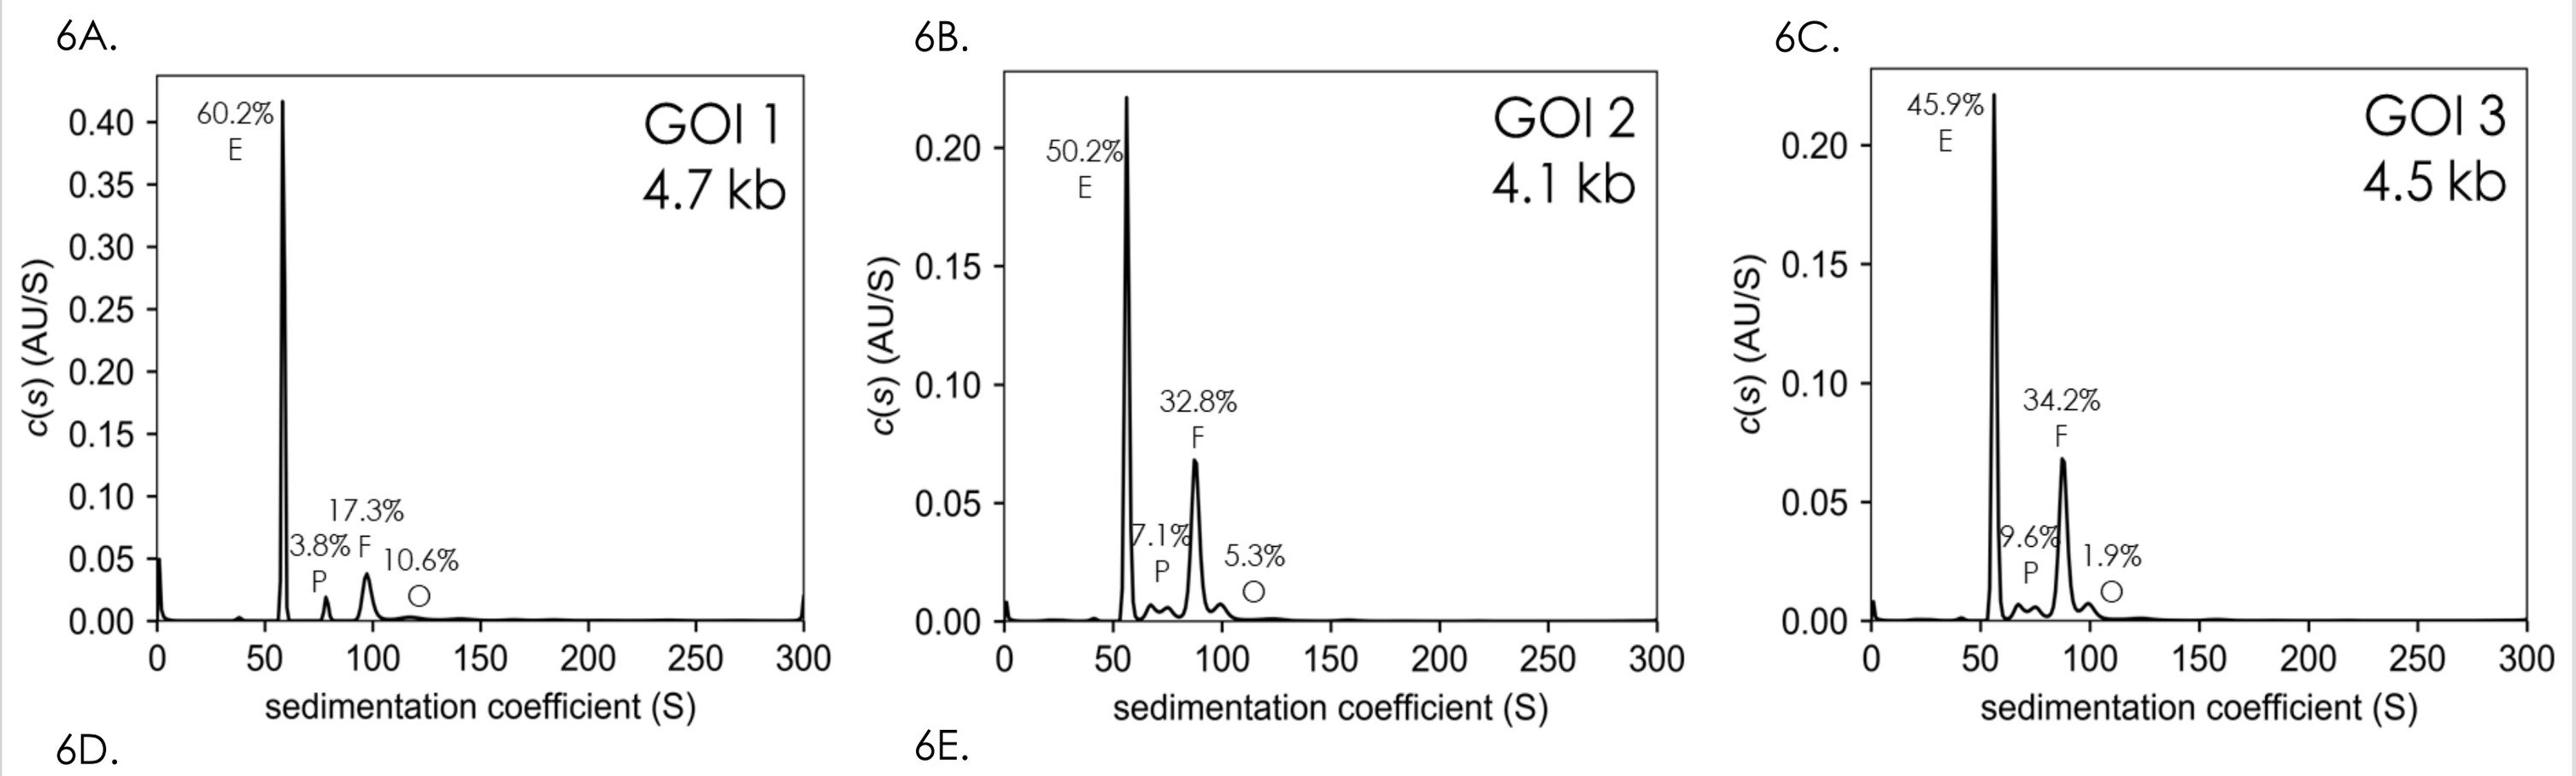
6A.
GOI 1
4.7 kb
60.2%
E
17.3%
F
3.8%
P
10.6%
6B.
GOI 2
4.1 kb
50.2%
E
32.8%
F
7.1%
P
5.3%
6C.
GOI 3
4.5 kb
45.9%
E
34.2%
F
9.6%
P
1.9%
c(s) (AU/S)
sedimentation coefficient (S)

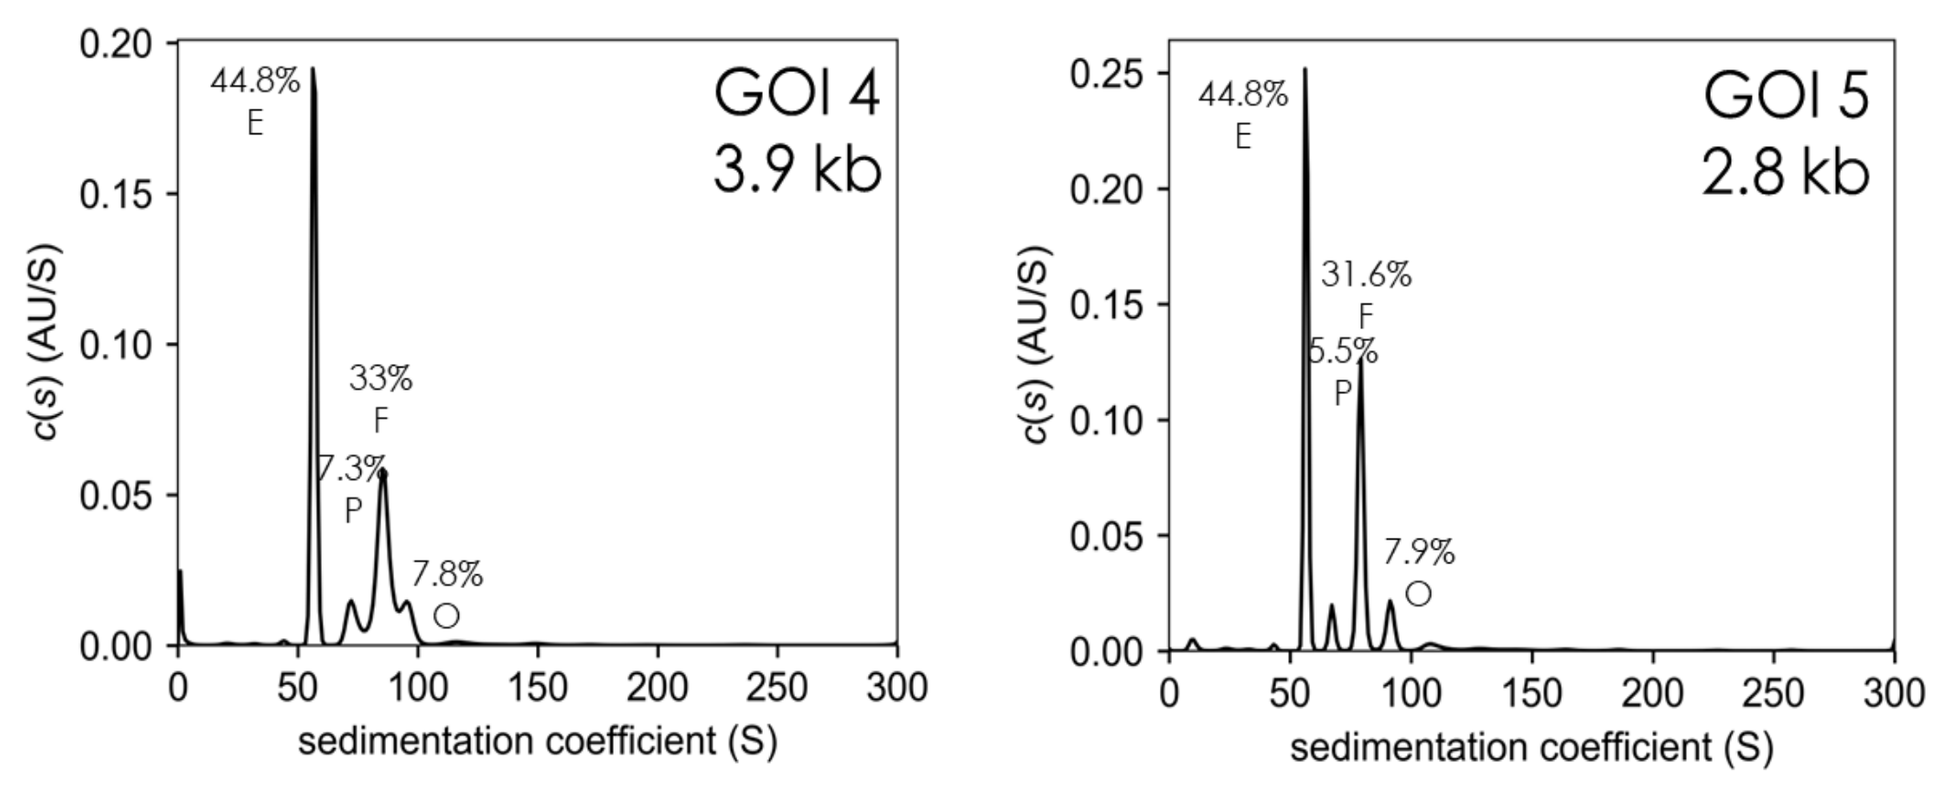
6D.
GOI 4
3.9 kb
44.8%
E
33%
F
7.3%
P
7.8%
6E.
GOI 5
2.8 kb
44.8%
E
31.6%
F
5.5%
P
7.9%
c(s) (AU/S)
sedimentation coefficient (S)

Figure 6. Capsid content determination by Analytical Ultracentrifugation (AUC) (Beckman Coulter). AUC sedimentation distribution plots for PD AAV6 material. (A-E) Percentage of empty (E) capsids, partial (P) capsids, full (F) capsids, and overpacked (O) capsids are shown for each construct.

3rd Generation Sequencing Shows Similar Read Alignment To Genome

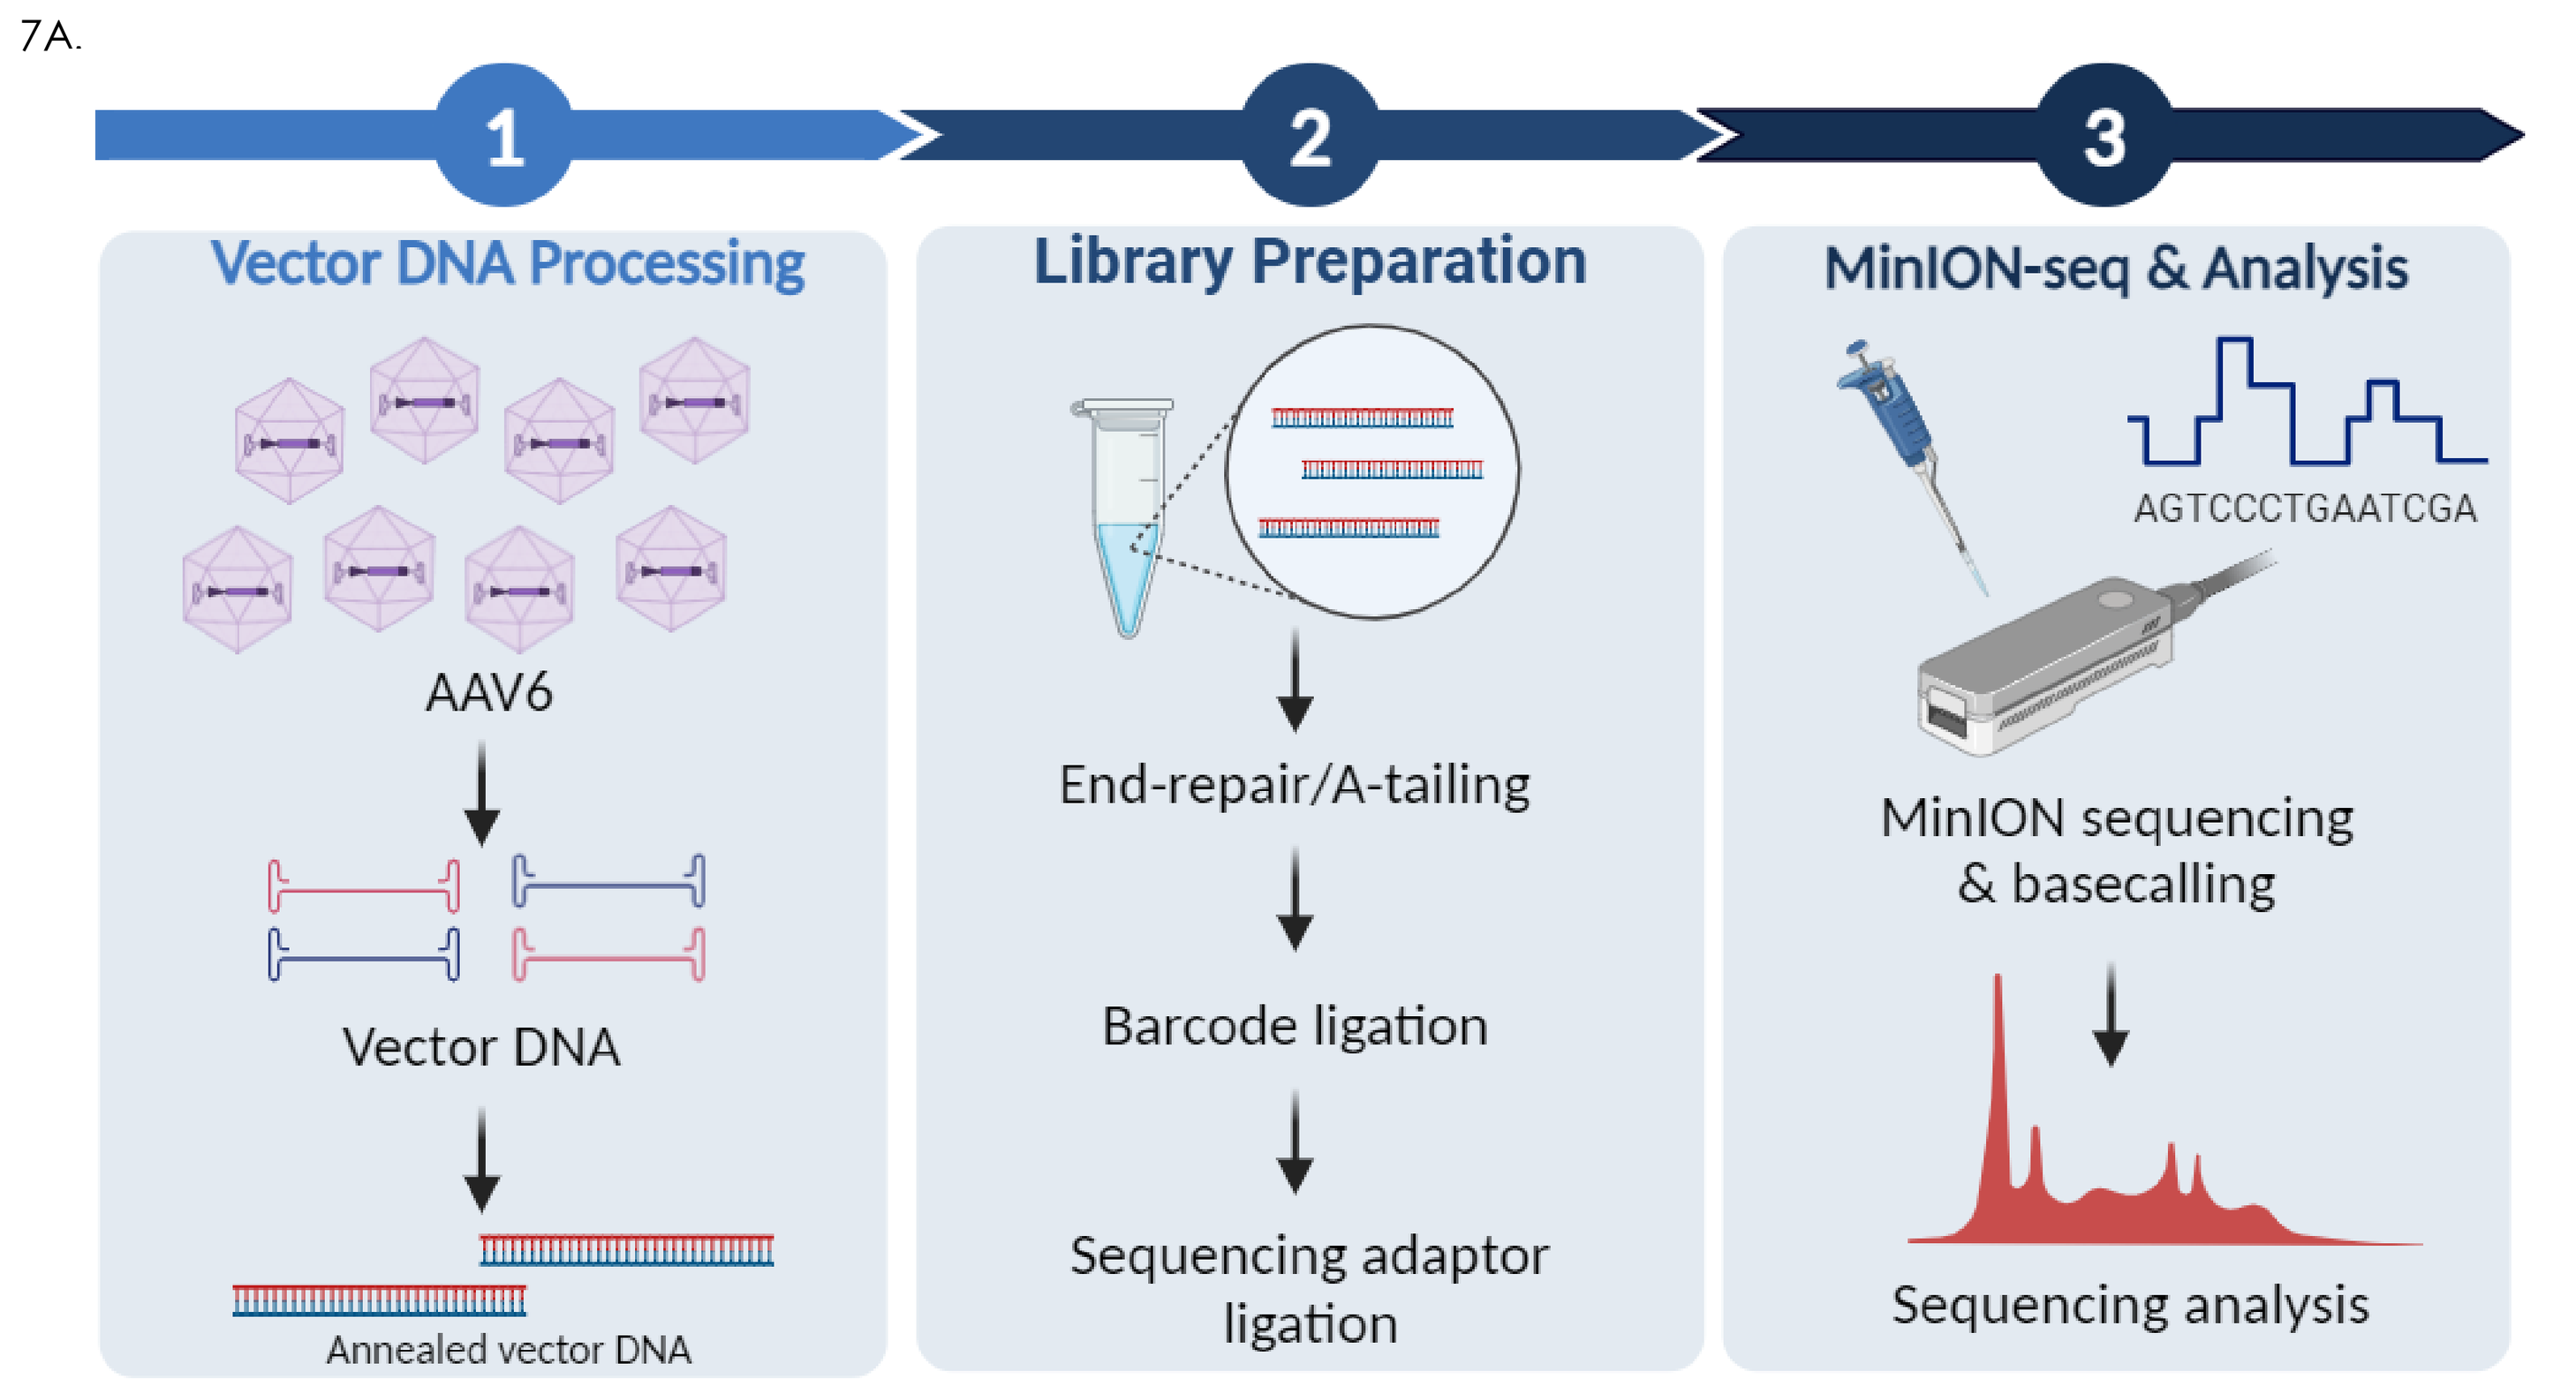
7A.
1
Vector DNA Processing
AAV6
Vector DNA
Annealed vector DNA
2
Library Preparation
End-repair/A-tailing
Barcode ligation
Sequencing adaptor ligation
3
MinION-seq & Analysis
AGTCCCTGAATCGA
MinION sequencing & basecalling
Sequencing analysis

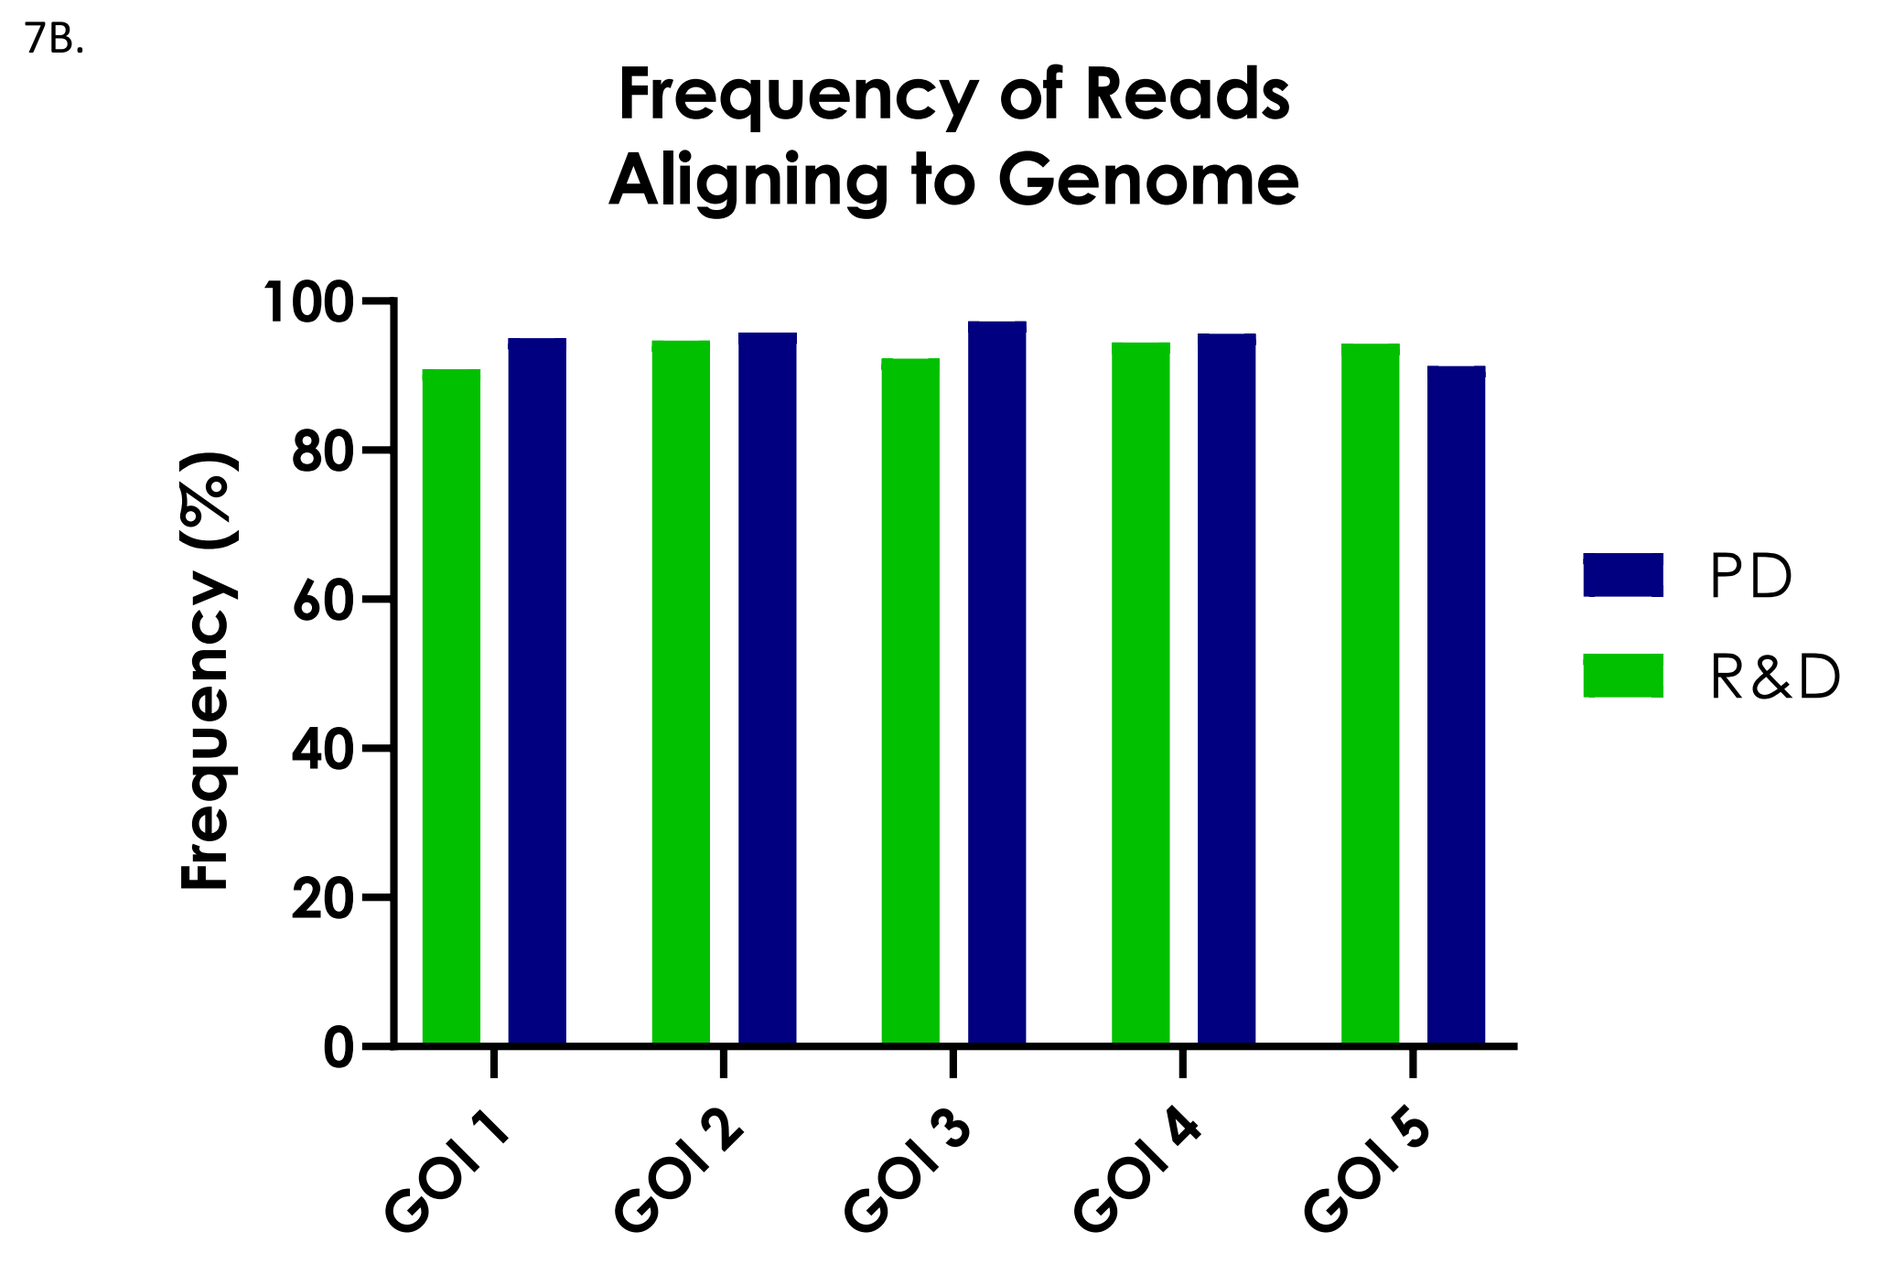
7B.
Frequency of Reads
Aligning to Genome
Frequency (%)
PD
R&D
GOI 1
GOI 2
GOI 3
GOI 4
GOI 5

Figure 7. Workflow and comparative analysis of ssAAV genome using Oxford nanopore technology (ONT). (A) Workflow for prepping ssAAV for 3rd generation sequencing analysis on the MinION-ONT. Image adapted from Oxford Nanopore, created using BioRender. (B) Frequency of reads generated by ONT that align to ssAAV genome sequence for both R&D and PD intermediate material. ssAAV genome analysis can include alignment that spans ITR-to-ITR genome, left partial, right partial, partial and vector + backbone.

Conclusion

- Analysis of R&D and PD AAV6 material showcased similar output across various assays regarding titering, packaging efficiency, viral capsid protein ratios, and AAV virion genomic DNA populations (summary of variance between R&D and PD material in Table 2).
- Results demonstrate upstream characterization of AAV6 candidates allow for earlier, and more efficient testing of multiple construct designs, and cost-efficient movement of manufacturable lead therapeutic candidates from the R&D to PD teams at ElevateBio.
- Next steps are to further optimize in-house 3rd generation sequencing, comparable to PacBio, and to perform AUC on R&D material for comparison against PD AAV6 material.

	Mass Photometry	Maurice (VP Protein Ratio)	AUC	Nanopore Sequencing
GOI 1	<5%	<5%	NA	<5%
GOI 2	<5%	<10%	NA	<5%
GOI 3	<5%	<10%	NA	<5%
GOI 4	<5%	<10%	NA	<5%
GOI 5	<5%	<10%	NA	<5%

Table 2. Summary of variance between R&D material and PD material across various assays.

Contact Information

Tess Kitchener | ElevateBio | 914-471-1931 | tkitchener@elevate.bio